Systems Pharmacology Modeling of Hypomethylating agents Decitabine and SGI-110 for Evaluation of AML treatment by targeting the S-phase with prolonged Pharmacokinetic exposures

A. Oganesian[1], O. Demin Jr. [2], A. Nikitich [2], O. Demin[2] and M. Azab[1]

[1] Astex Pharmaceuticals Inc., Dublin, CA ; [2] Institute for Systems Biology, Moscow, Russia

BACKGROUND

Acute myeloid leukemia (AML) is a tumor associated with myeloid line of blood cells, characterized by the abnormal rapid proliferation of white blood cells in the bone marrow. The most common approach for AML treatment is the reduction of abnormal cell proliferation rate which can be achieved by targeting the key stages of the cell cycle. Decitabine is a well characterized hypomethylating agent (HMA), which is incorporated into DNA during the S-phase of cell cycle, inhibits methylation of antitumor genes and induces G2/M arrest. However, it has a very short half-life (15-35 min) after IV infusion due to rapid degradation by cytidine deaminase. SGI-110, a 2nd generation HMA was designed to increase the in vivo exposure/potential efficacy of its active metabolite decitabine. The aim of this effort was to explore how changes in exposure window of decitabine affect DNA demethylation and tumor cell proliferation in AML patients.

MATERIALS and METHOD

A systems pharmacology model was developed describing myeloblasts cell cycle; PK of decitabine after IV infusion and after dosing with SQ SGI-110; LINE-1 demethylation changes following treatment and progression of AML. The model is a system of differential equations characterizing myeloblasts transition between cell cycle phases and proliferation of healthy neutrophils. Parameters of the model were calculated on the basis of the literature data or fitted against available and published experimental data. 95% confidential intervals were calculated for fitted parameters.

Table 1. Comparison of number of experimental points and model parameters for fitting and validation

Sub-model	# of parameters fitted in this sub-model	# of experimental points for fitting	# of points for validation
PK sub-model	10	74	NA#
Cell cycle sub-model without decitabine	8	24	25
Cell cycle sub-model with decitabine	3	40	30
LINE1 PD sub-model (invivo)	16	61	31
LINE1 PD sub-model for healthy subject	1	3	n.a.*
Hematopoietic sub-model for AML Patients	9	62	n.a.*
TOTAL:	47	264	86

#NA - Not Applicable *n.a. - No Available Data

Figure 1. Block-Scheme of Systems Pharmacology Model of Acute Myelogenous Leukemia

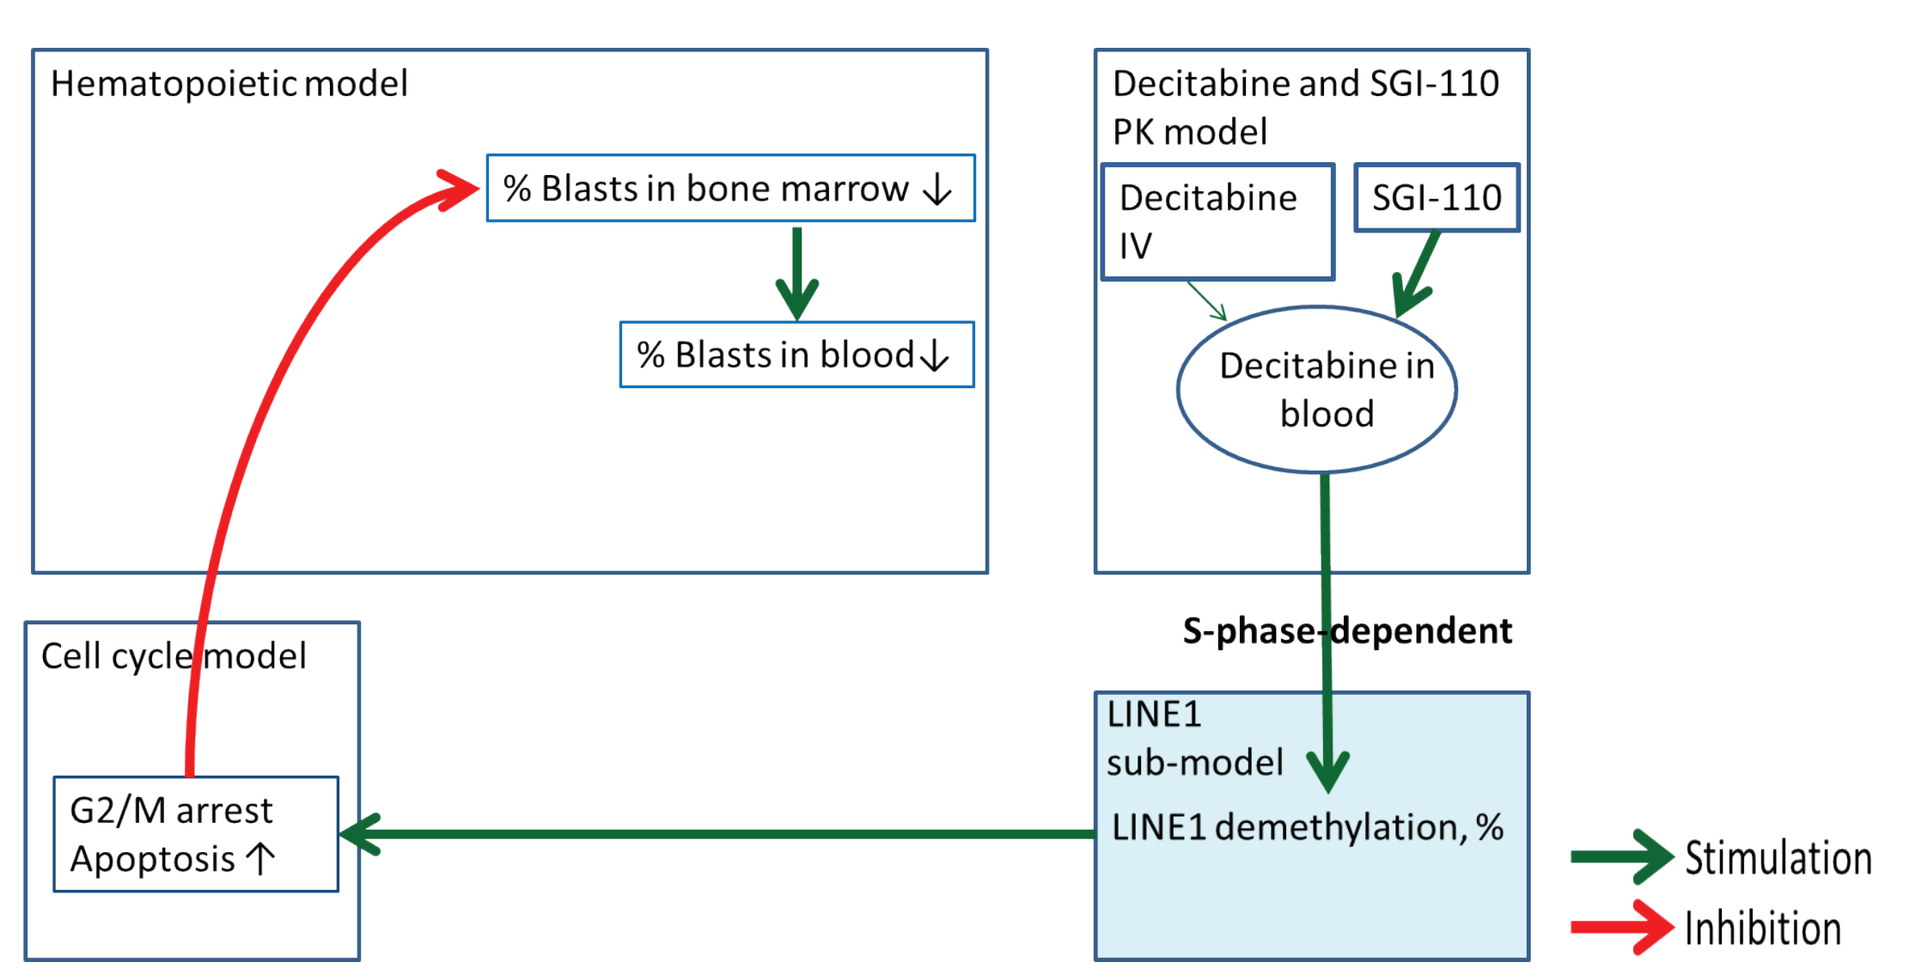
Hematopoietic model
% Blasts in bone marrow ↓
% Blasts in blood↓
Decitabine and SGI-110 PK model
Decitabine IV
SGI-110
Decitabine in blood
S-phase-dependent
Cell cycle model
G2/M arrest
Apoptosis ↑
LINE1 sub-model
LINE1 demethylation, %
Stimulation
Inhibition

RESULTS

Figure 2. Fitting against: A) Leukemic KG-1 cell proliferation data (dots represent experimental data, lines – simulation) B) distribution of cell cycle phases for KG-1 without decitabine C) distribution of cell cycle phases for KG-1 after treatment with 0.1 μM decitabine

A

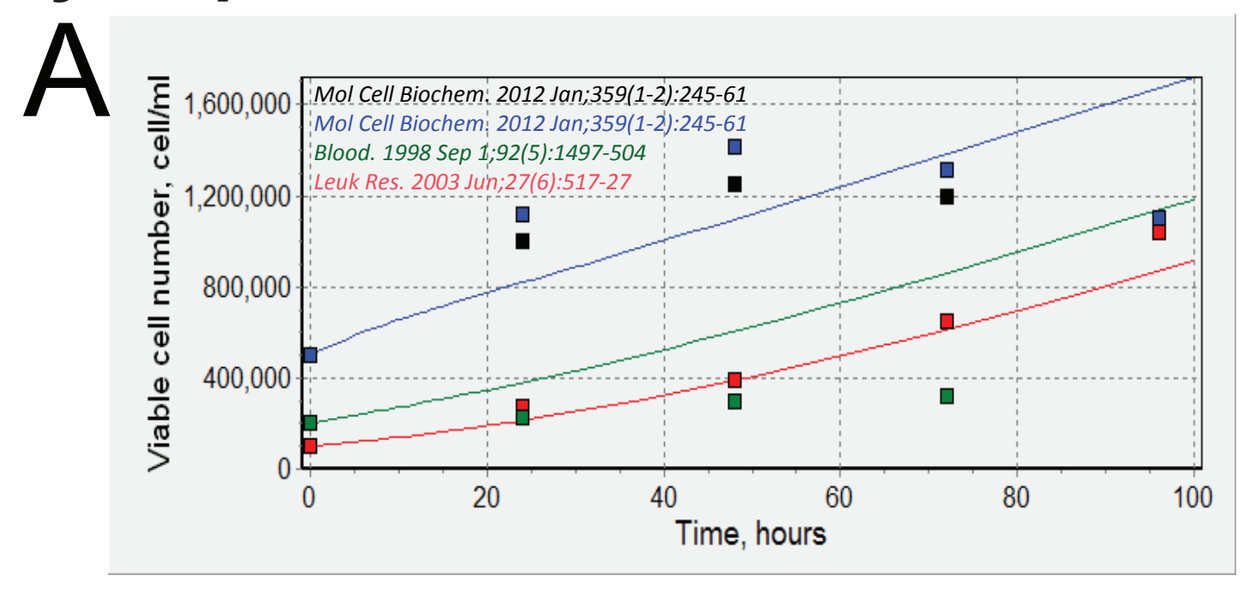
Mol Cell Biochem. 2012 Jan;359(1-2):245-61
Mol Cell Biochem. 2012 Jan;359(1-2):245-61
Blood. 1998 Sep 1;92(5):1497-504
Leuk Res. 2003 Jun;27(6):517-27
Viable cell number, cell/ml
Time, hours

B

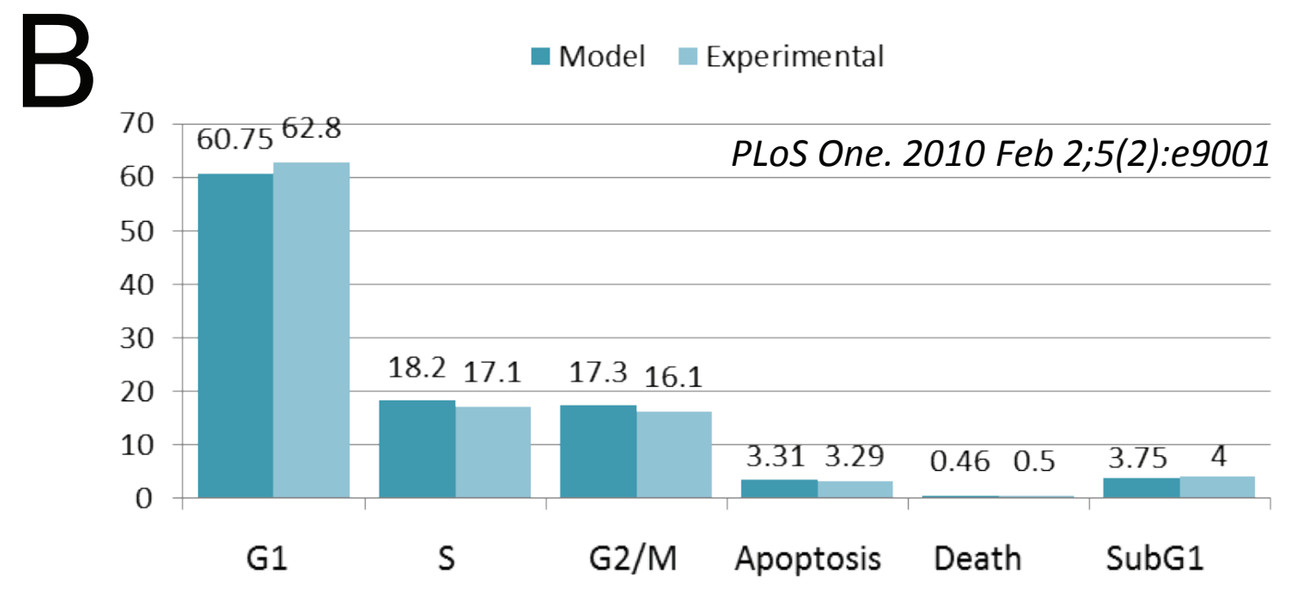
Model
Experimental
PLoS One. 2010 Feb 2;5(2):e9001
G1
S
G2/M
Apoptosis
Death
SubG1

C

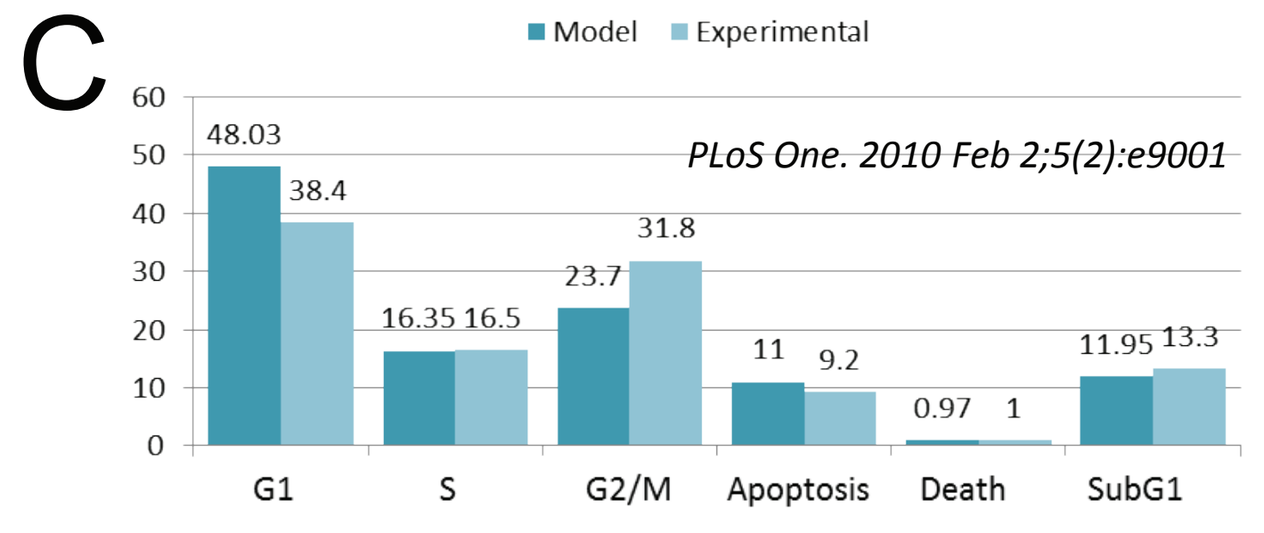
Model
Experimental
PLoS One. 2010 Feb 2;5(2):e9001
G1
S
G2/M
Apoptosis
Death
SubG1

The model satisfactorily reproduces following data: In vitro data from experiments with myeloblast cell lines showing proliferation and distribution between cell cycle phases with or without decitabine treatment

Figure 3. Fitting against:
A) PK of decitabine after 1h IV infusion at 20 mg/m² for 5 days
B) PK of decitabine after 60, 90 and 125 mg/m² SGI-110 SQ injection

A

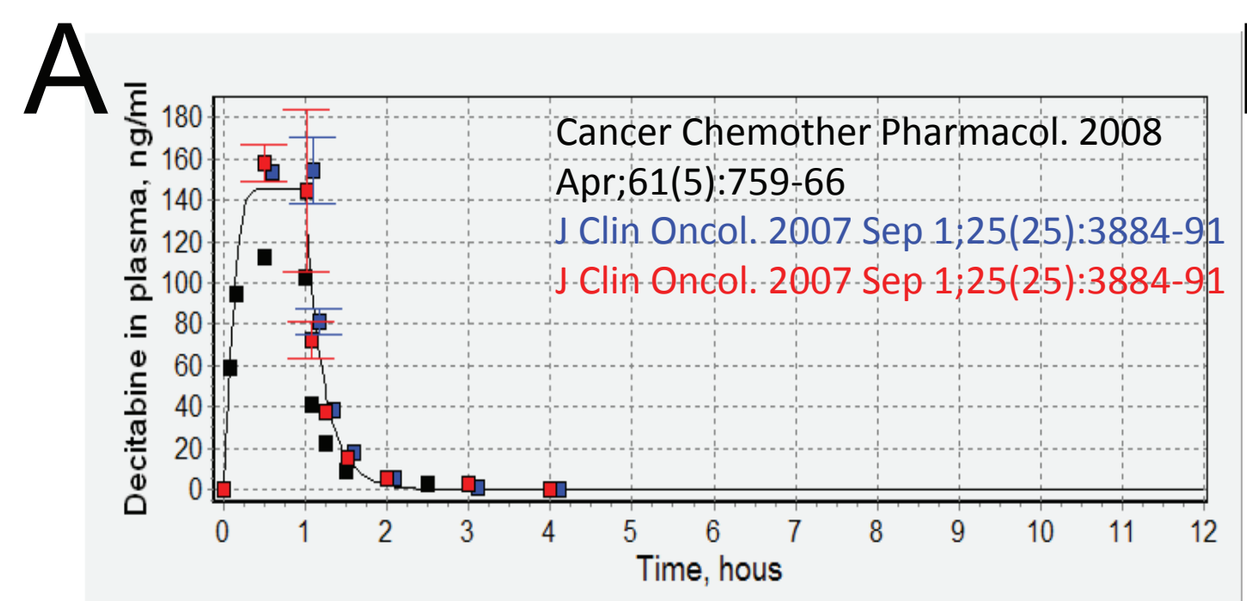
Cancer Chemother Pharmacol. 2008 Apr;61(5):759-66
J Clin Oncol. 2007 Sep 1;25(25):3884-91
J Clin Oncol. 2007 Sep 1;25(25):3884-91
Decitabine in plasma, ng/ml
Time, hous

B

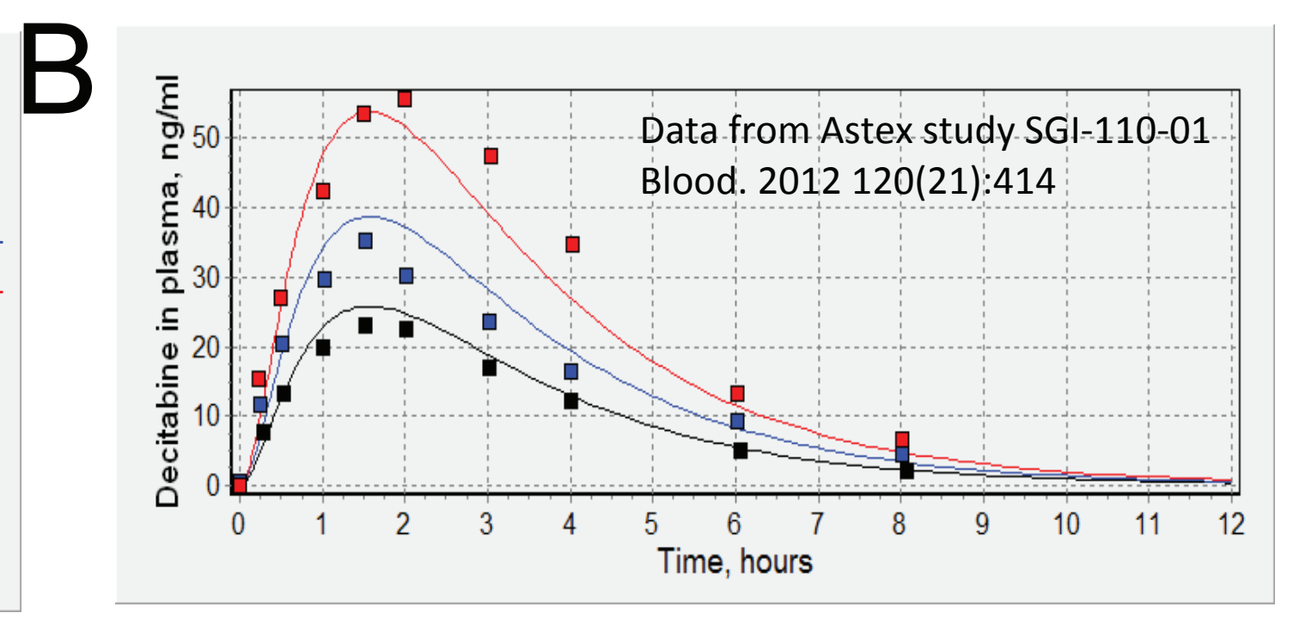
Data from Astex study SGI-110-01
Blood. 2012 120(21):414
Decitabine in plasma, ng/ml
Time, hours

- Decitabine pharmacokinetics in plasma after 1-hr IV decitabine infusion at 20 mg/m² and SQ SGI-110 injection at different dose levels.
- Longer exposure window for decitabine is observed after SGI-110 SQ administration.

Figure 4. LINE-1 demethylation after IV decitabine or SQ SGI-110

A

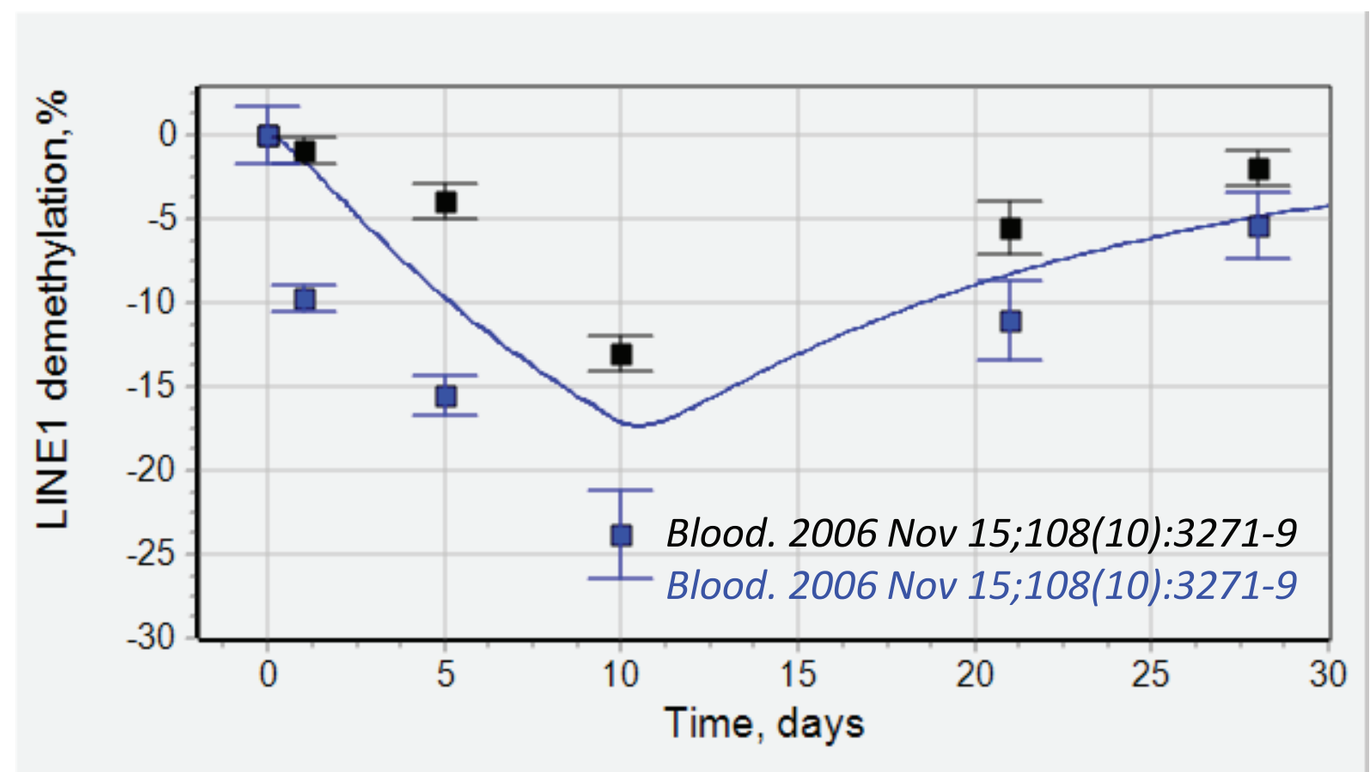
LINE1 demethylation, %
Blood. 2006 Nov 15;108(10):3271-9
Blood. 2006 Nov 15;108(10):3271-9
Time, days

B

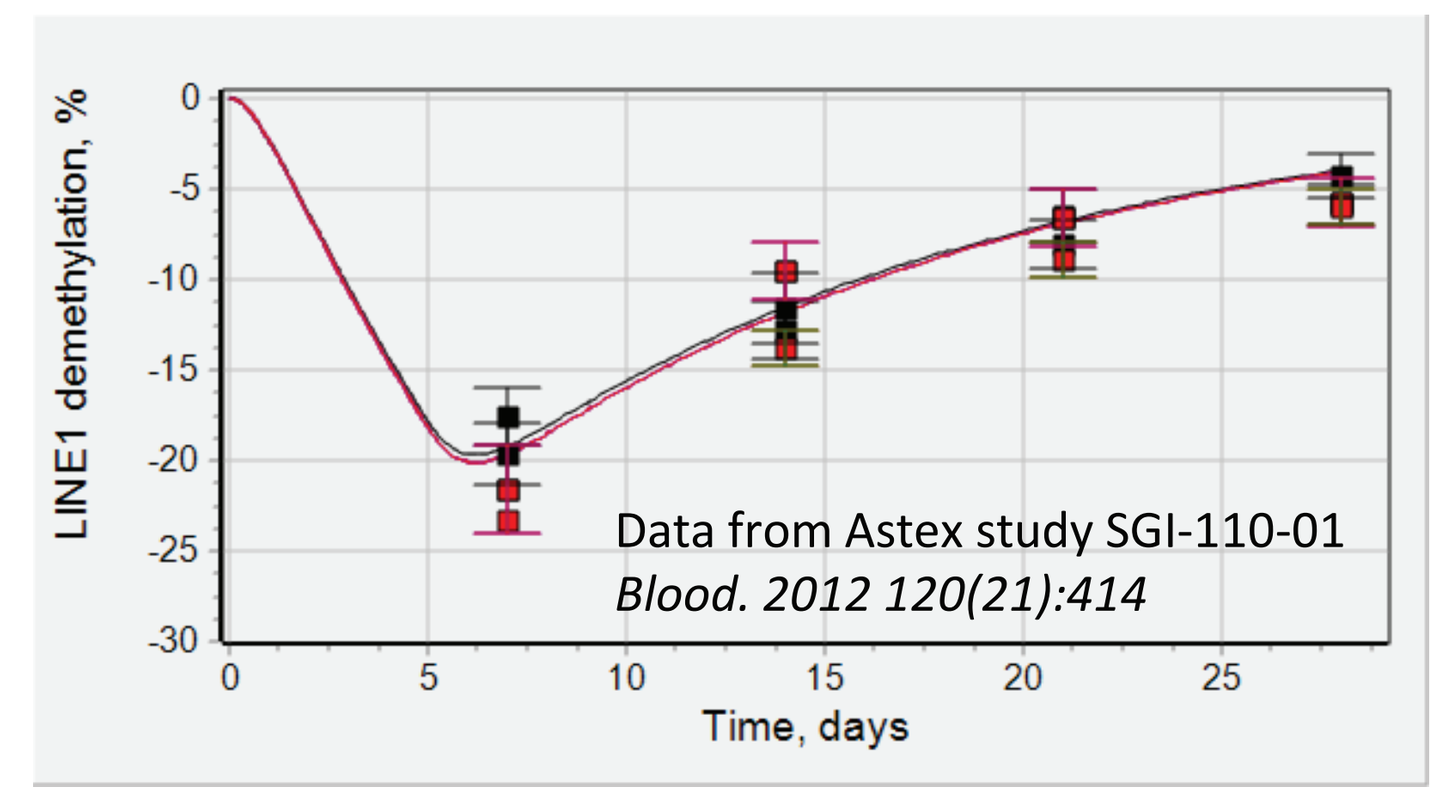
LINE1 demethylation, %
Data from Astex study SGI-110-01
Blood. 2012 120(21):414
Time, days

A) LINE-1 demethylation after 1-hr decitabine IV infusion at 15 mg/m² daily X10 (responders and nonresponders)
B) LINE-1 demethylation after 60 (black) or 90 mg/m² (red) SGI-110 SQ administration daily X5 in patients with r/r AML

Model shows that administration of SGI-110 at 60 mg/m² daily for 5 days of a 28-day treatment cycle induces LINE-1 demethylation at a higher level due to wider exposure window of the active metabolite decitabine, compared to decitabine 1-hr IV infusion at 20 mg/m² daily for 5 days.

Figure 5. Simulations of Decitabine in plasma (A), in cell (B) and DNA (C)

We compared model simulations of decitabine concentrations in plasma, in the cell (decitabine-triphosphate) and incorporated within DNA after 20 mg/m² Decitabine IV (1-hr, DailyX5) or 60 mg/m² SGI-110 Daily X5

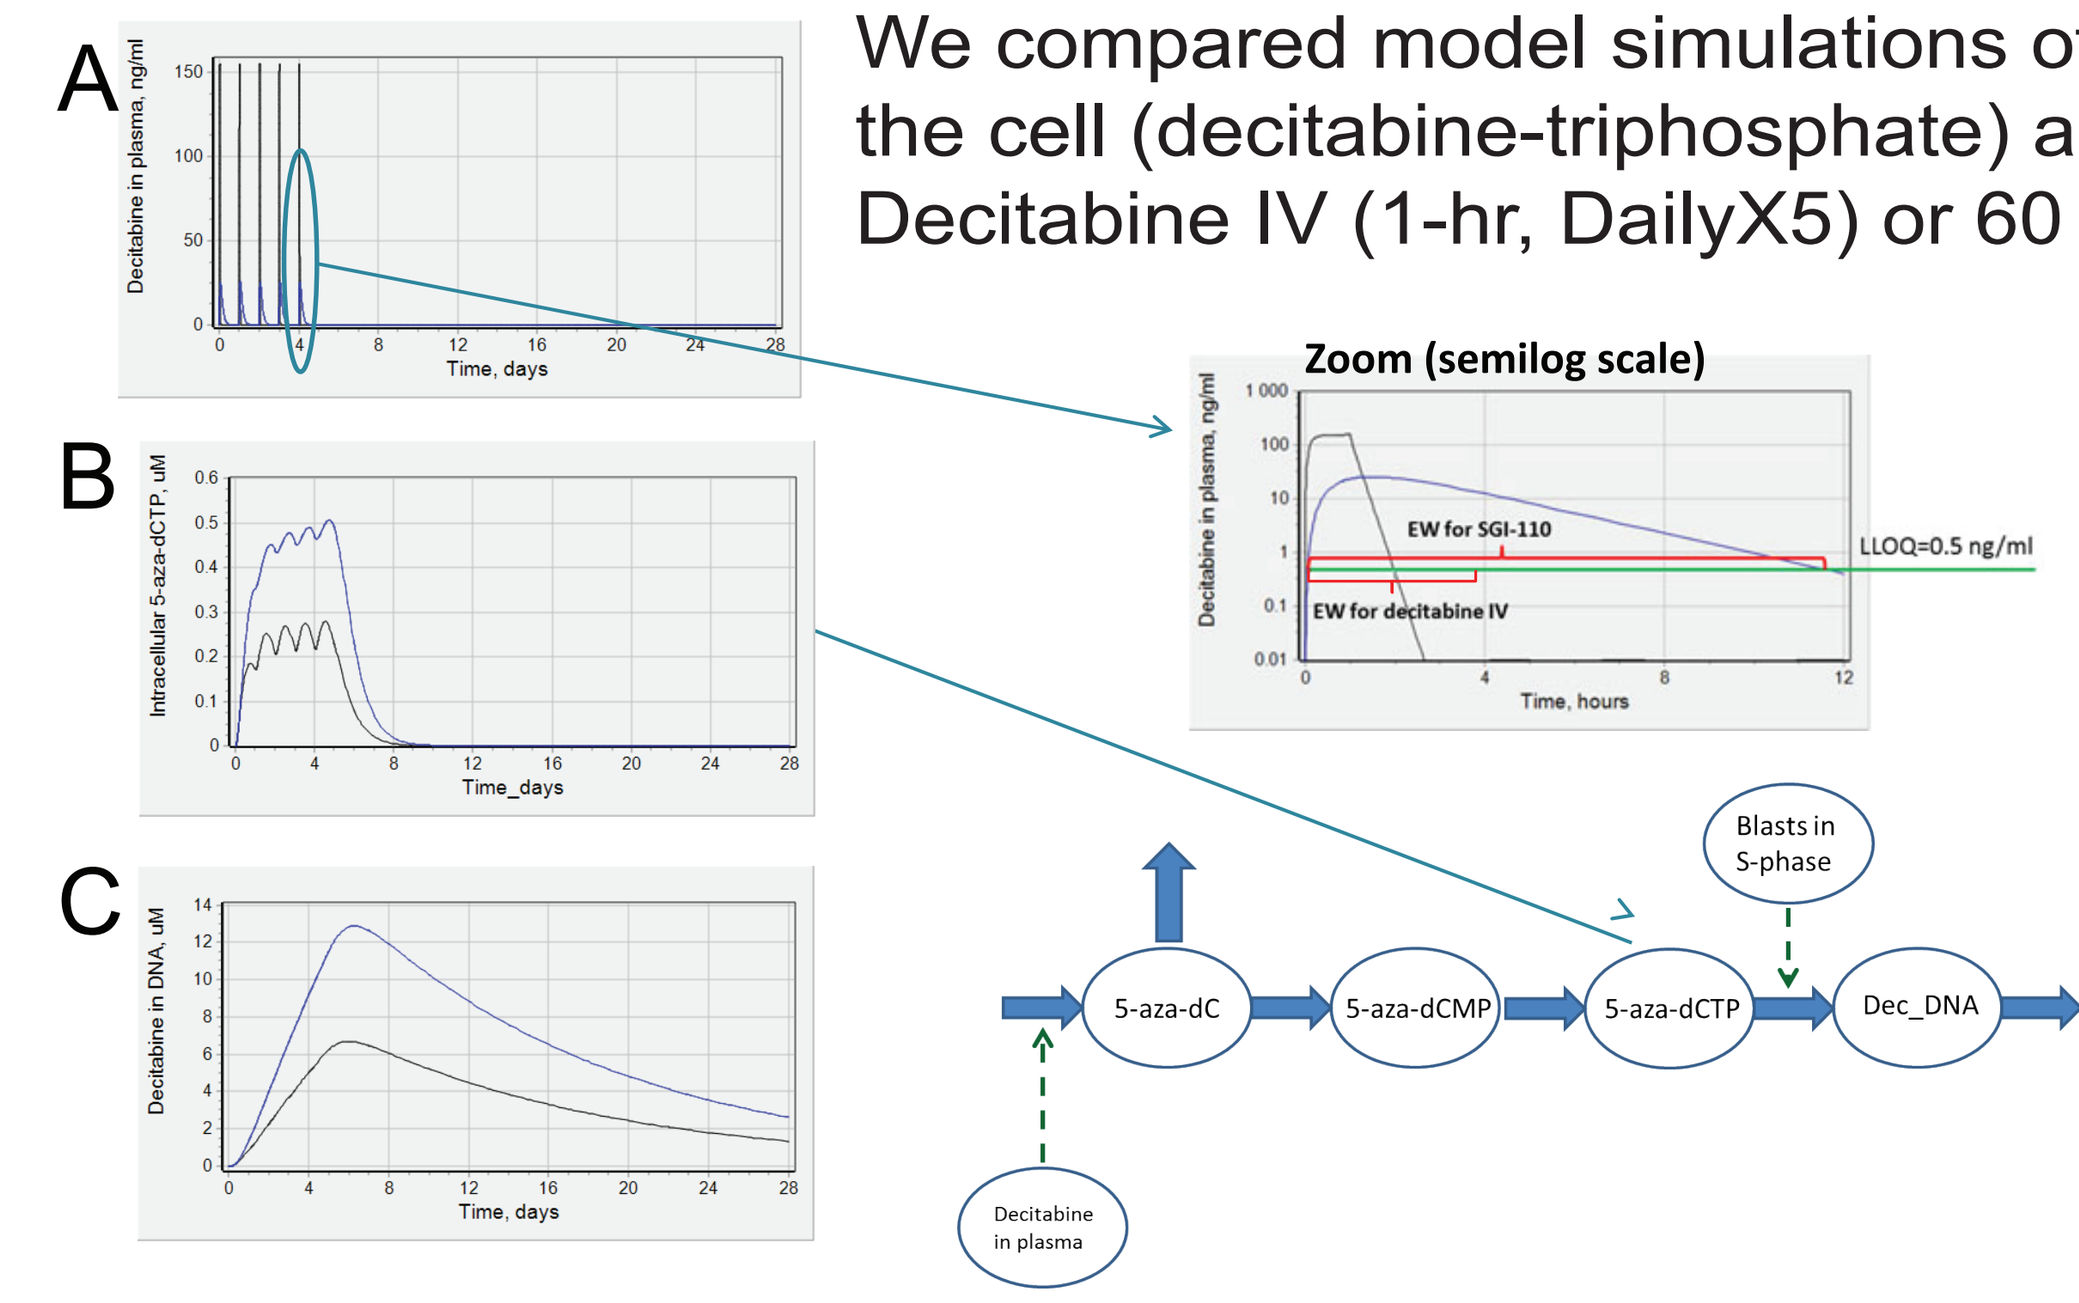
A
B
C
Decitabine in plasma, ng/ml
Intracellular 5-aza-dCTP, uM
Decitabine in DNA, uM
Time, days
Zoom (semilog scale)
EW for SGI-110
EW for decitabine IV
LLOQ=0.5 ng/ml
Time, hours
5-aza-dC
5-aza-dCMP
5-aza-dCTP
Dec_DNA
Blasts in S-phase
Decitabine in plasma

Black curve – decitabine in plasma after decitabine IV infusion with 20 mg/m²;
Blue curve – decitabine in plasma after SGI-110 SQ injection at 60 mg/m²

Figure 6. Changes in % blasts in blood (black) and bone marrow (blue) during AML progression for high (A) or low (B) proliferative virtual patients

A

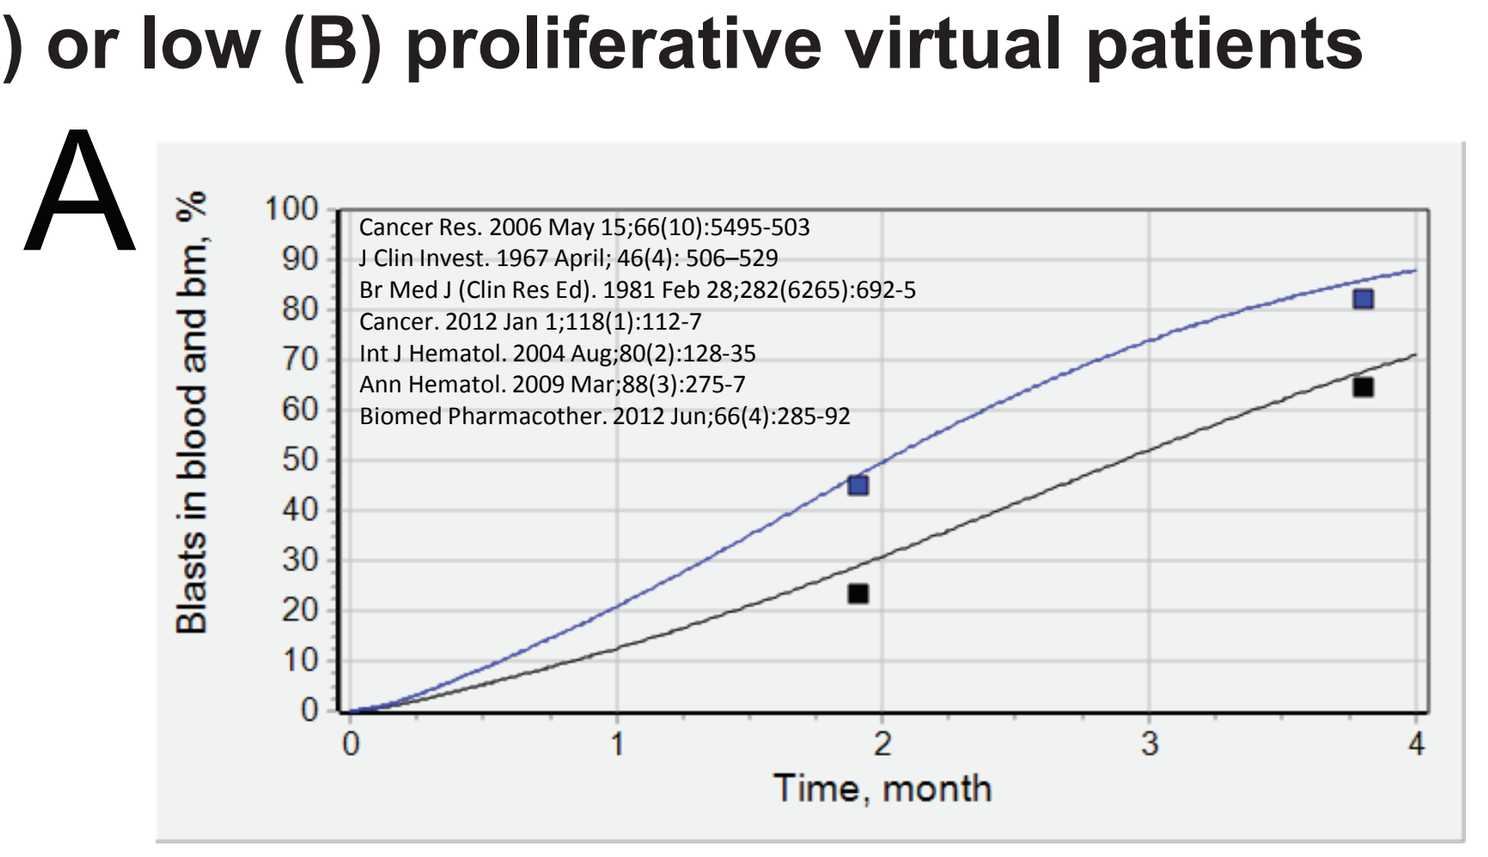
Cancer Res. 2006 May 15;66(10):5495-503
J Clin Invest. 1967 April; 46(4): 506–529
Br Med J (Clin Res Ed). 1981 Feb 28;282(6265):692-5
Cancer. 2012 Jan 1;118(1):112-7
Int J Hematol. 2004 Aug;80(2):128-35
Ann Hematol. 2009 Mar;88(3):275-7
Biomed Pharmacother. 2012 Jun;66(4):285-92
Blasts in blood and bm, %
Time, month

B

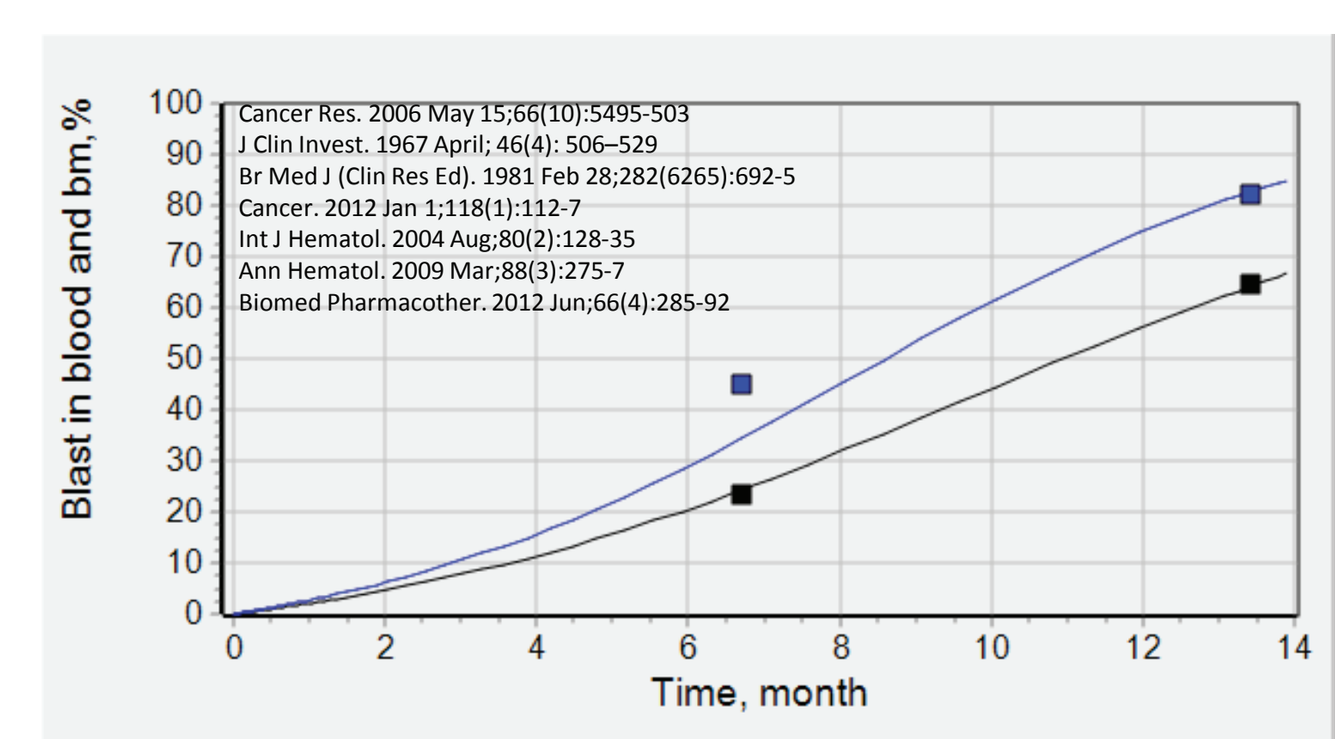
Cancer Res. 2006 May 15;66(10):5495-503
J Clin Invest. 1967 April; 46(4): 506–529
Br Med J (Clin Res Ed). 1981 Feb 28;282(6265):692-5
Cancer. 2012 Jan 1;118(1):112-7
Int J Hematol. 2004 Aug;80(2):128-35
Ann Hematol. 2009 Mar;88(3):275-7
Biomed Pharmacother. 2012 Jun;66(4):285-92
Blast in blood and bm, %
Time, month

Peripheral blast levels in blood during AML progression of patients with or without treatment by HMAs. Model describes two types of virtual patients with high (A) or low (B) rate of proliferation of blasts.

Figure 7A. Changes in bone marrow blasts for high (A) and low (B) proliferative virtual AML patients with or without treatment for 6 cycles

A

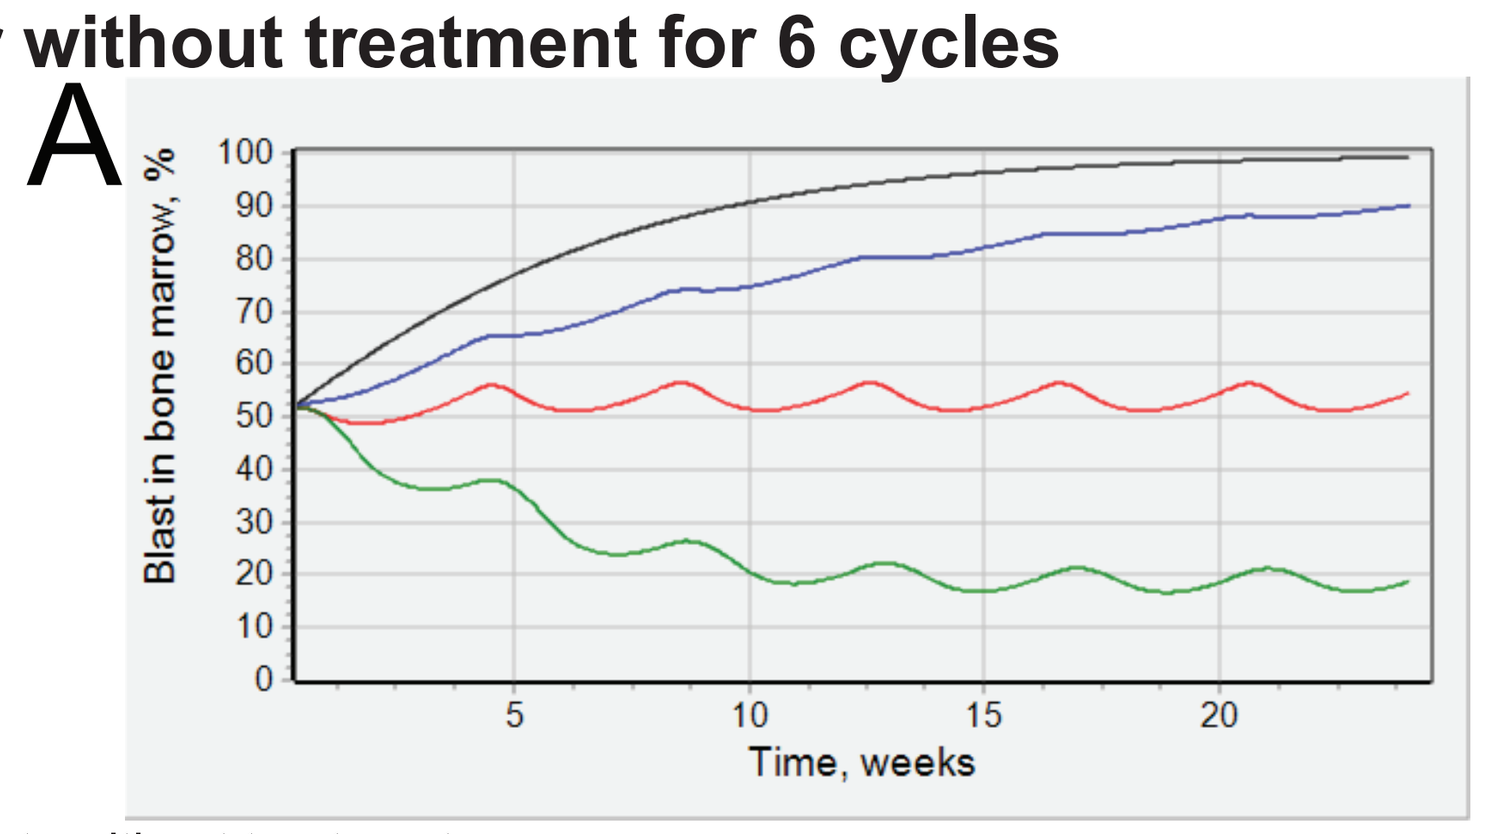
Blast in bone marrow, %
Time, weeks

B

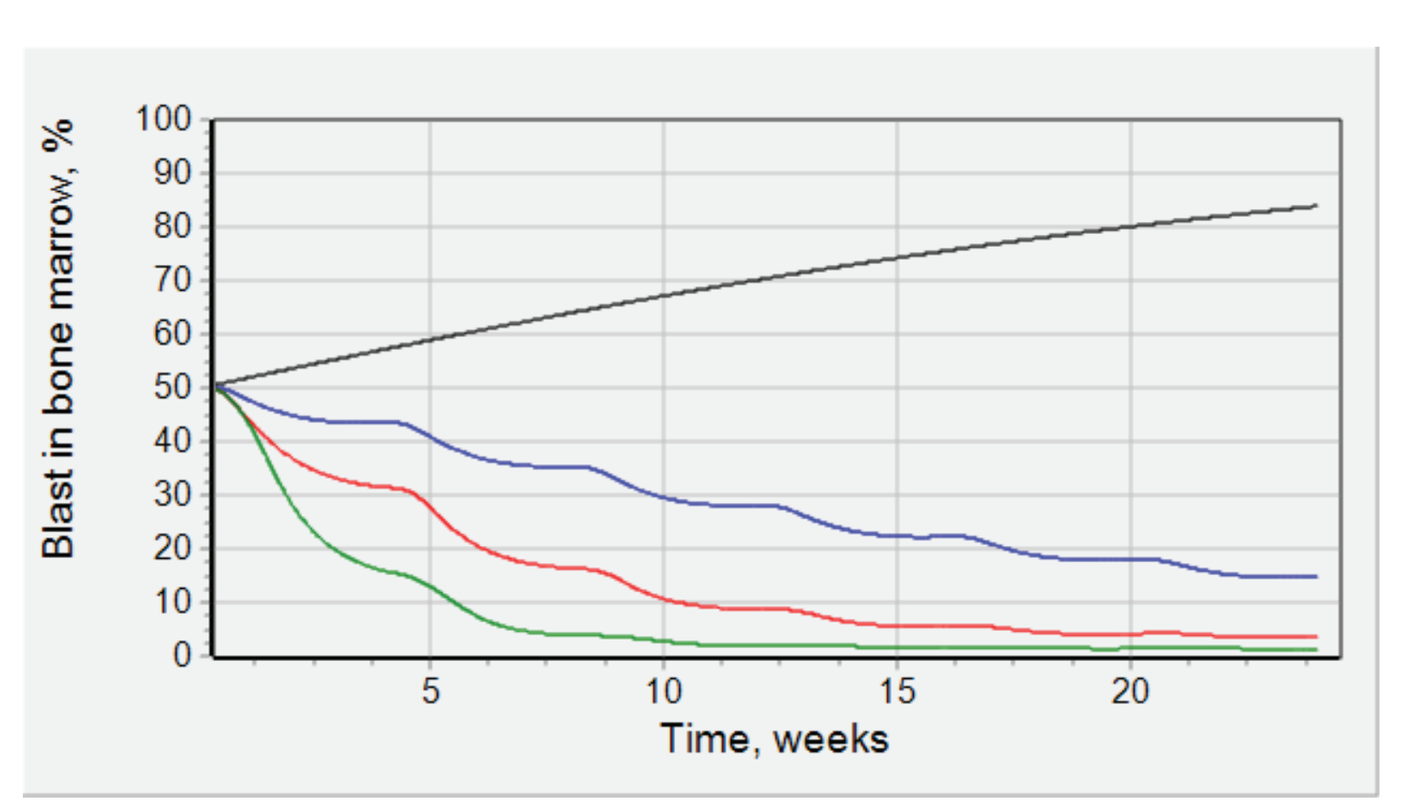
Blast in bone marrow, %
Time, weeks

Black - blasts without treatment
Blue – treatment with decitabine 1-hr IV infusion at 20 mg/m² for 5 days (of a 28 day cycle)
Red – treatment with SGI-110, 5 days at 60 mg/m² (of a 28 day cycle)
Green - treatment with SGI-110, 10 days at 60 mg/m² SGI-110 (of a 28 day cycle)

Simulation of 6 treatment cycles in high (A) and low (B) proliferative virtual AML patients showed that SGI-110 performed better than IV decitabine in affecting proliferation of myeloblasts in bone marrow and peripheral blood.

Figure 7B. Changes in peripheral blood blasts for high (A) and low (B) proliferative virtual AML patients with or without treatment for 6 cycles

A

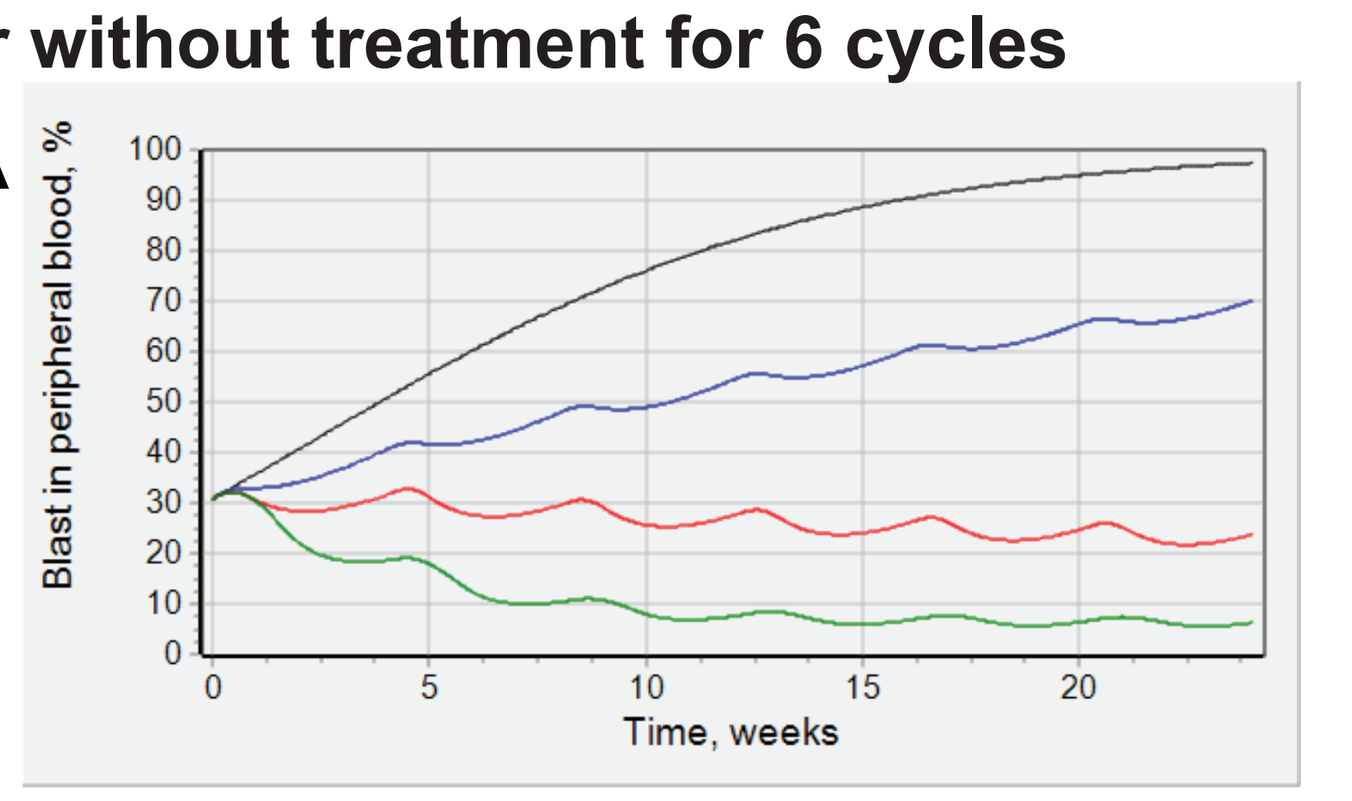
Blast in peripheral blood, %
Time, weeks

B

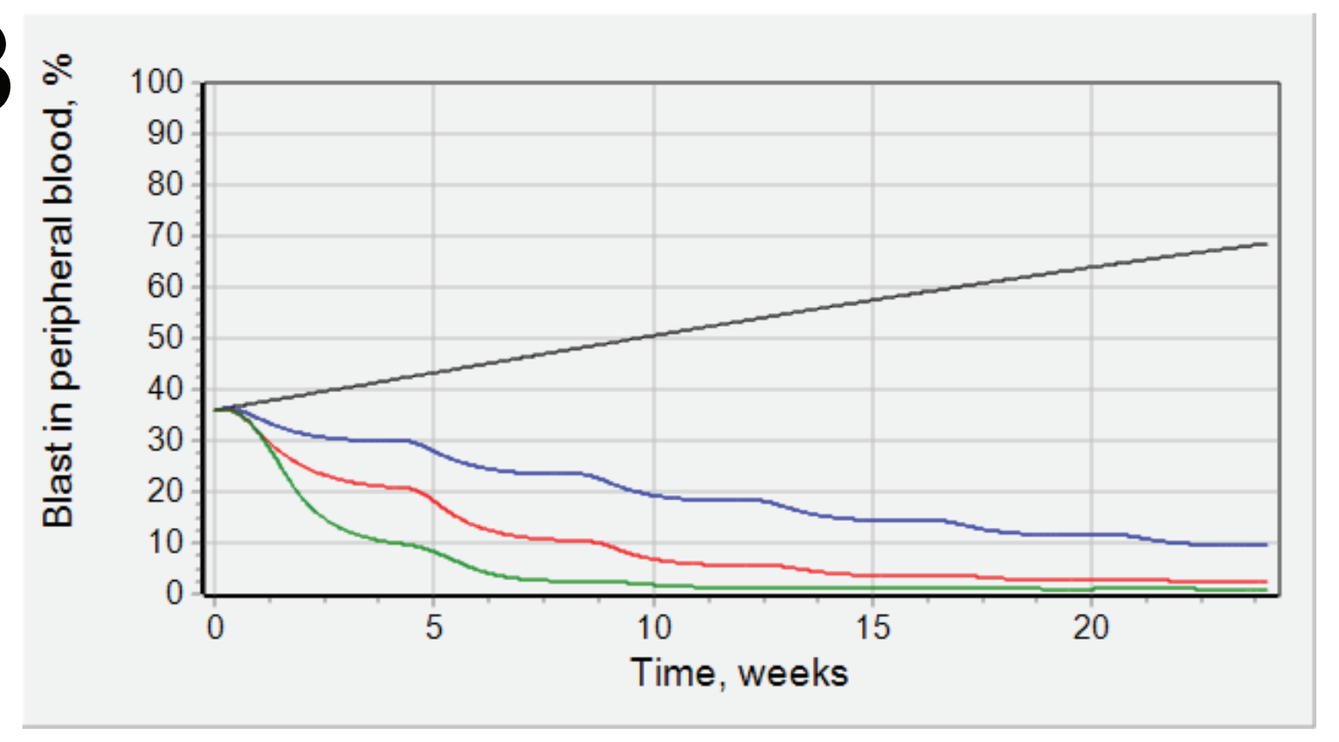
Blast in peripheral blood, %
Time, weeks

Black - blasts without treatment
Blue – treatment with decitabine 1-hr IV infusion at 20 mg/m² for 5 days (of a 28 day cycle)
Red – treatment with SGI-110, 5 days at 60 mg/m² (of a 28 day cycle)
Green - treatment with SGI-110, 10 days at 60 mg/m²SGI-110 (of a 28 day cycle)

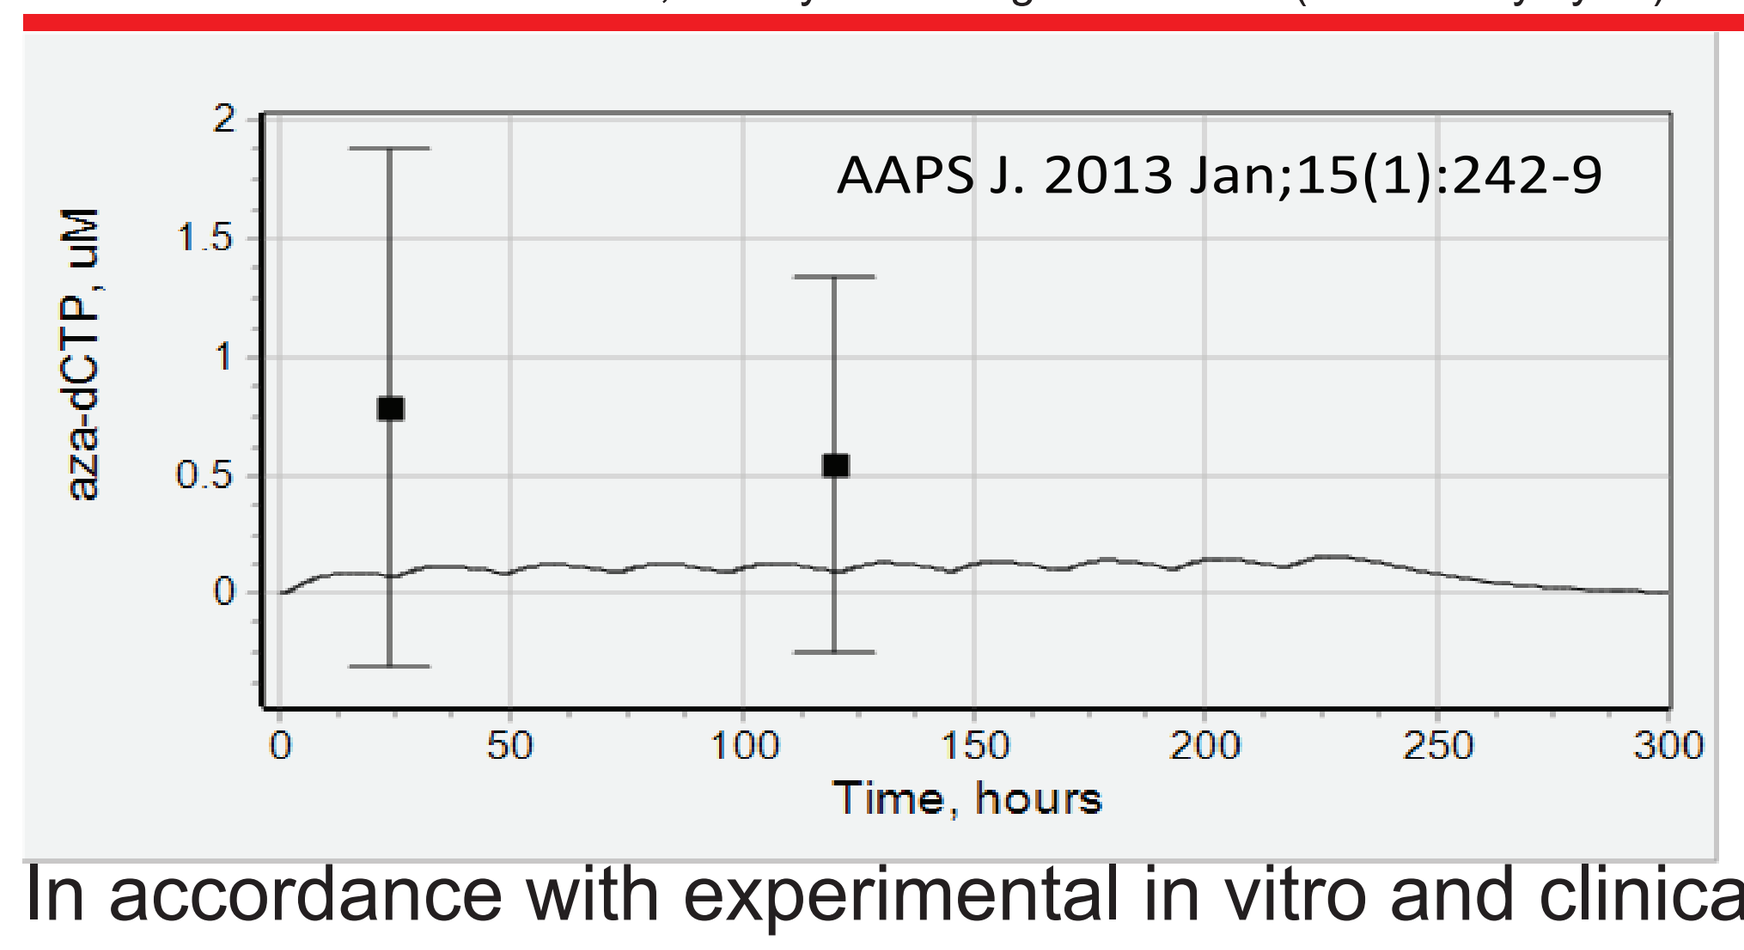
AAPS J. 2013 Jan;15(1):242-9
aza-dCTP, uM
Time, hours

Figure 8. Intracellular levels of tri-phosphorylated form of decitabine after decitabine IV 1-hr infusion, daily X10

In accordance with experimental in vitro and clinical data [Cancer Chemother Pharmacol. 2012 Jun;69(6):1457-66; AAPS J. 2013 Jan;15(1):242-9], model shows that intracellular tri-phosphorylated form of decitabine accumulates during treatment period. Model predicts that SGI-110 SQ results in higher accumulation of d-TP than Decitabine IV due to more favorable exposure window (figure 5b).

Figure 9. Blast dynamics in bone marrow (blue) and blood (red) during treatment with SGI-110 daily X5

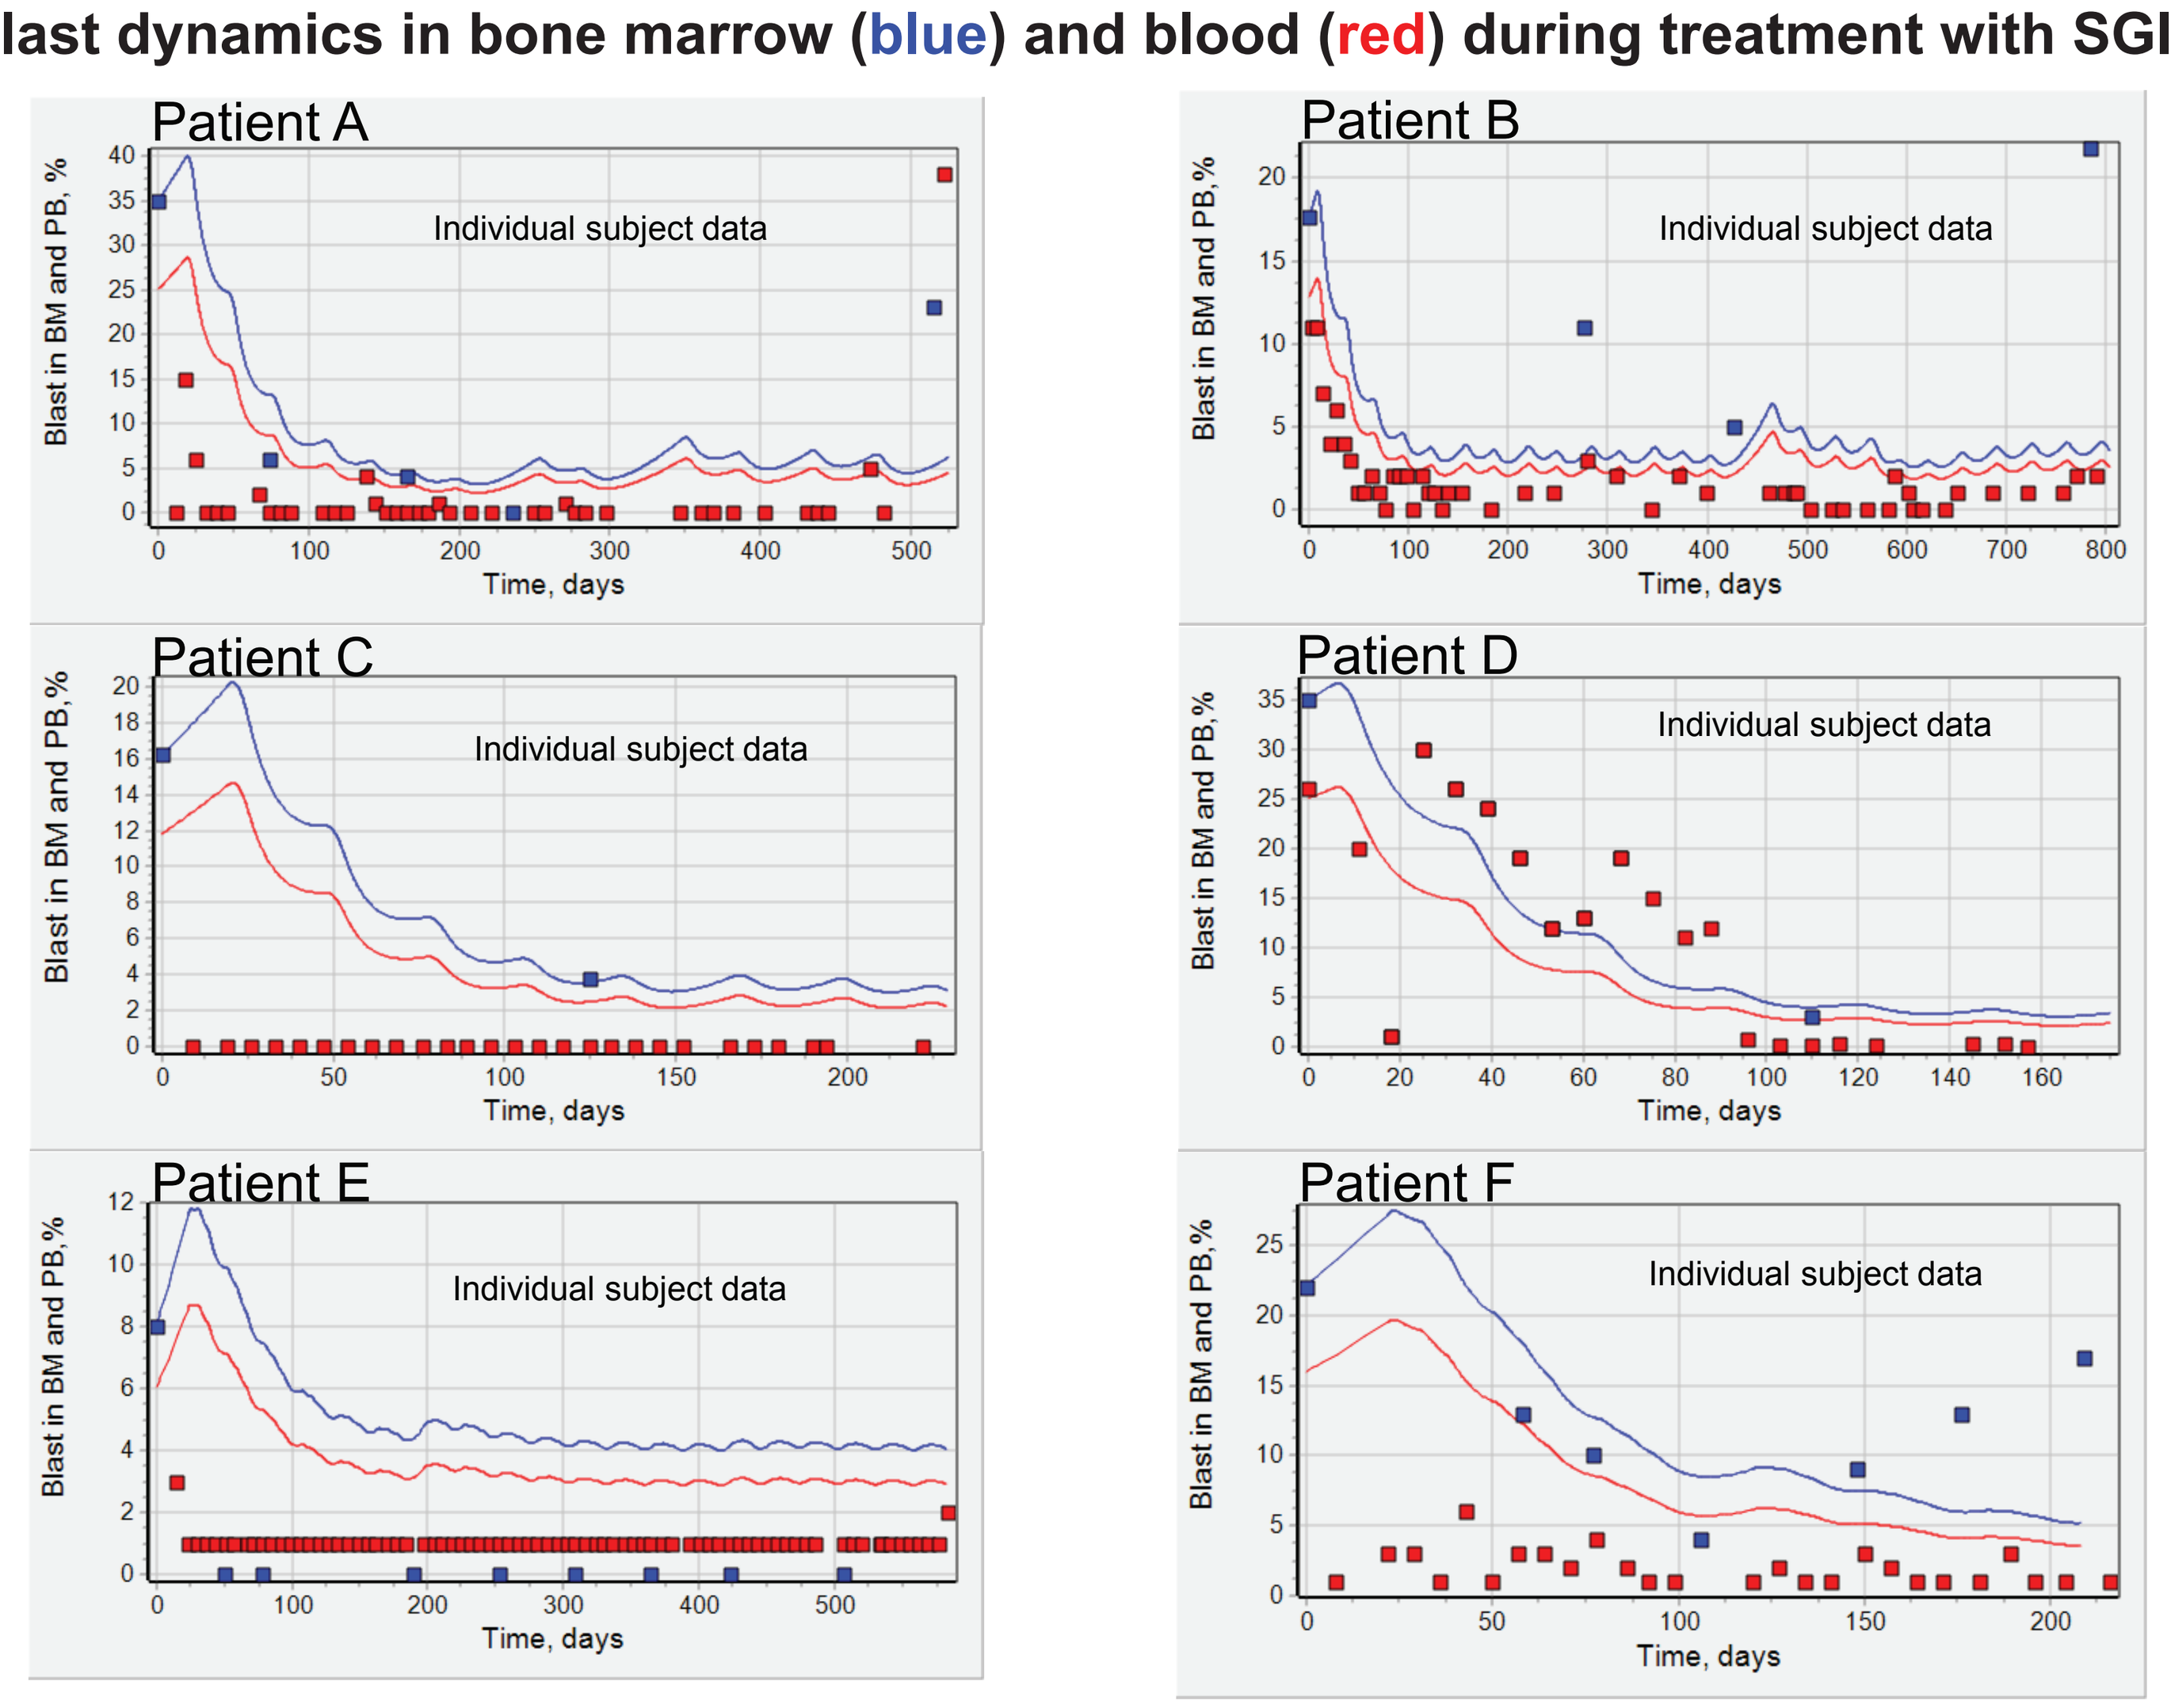
Patient A
Patient B
Patient C
Patient D
Patient E
Patient F
Individual subject data
Blast in BM and PB, %
Time, days

Model satisfactorily predicts bone marrow and peripheral blast dynamics in responders to SGI-110 treatment.

CONCLUSIONS

- Prolonged exposure window of active metabolite of SGI-110, decitabine, leads to a more pronounced effect on LINE-1 demethylation.
- Simulations in virtual AML patients showed that SGI-110 performed better than IV decitabine in affecting proliferation of myeloblasts in bone marrow and peripheral blood.
- Model successfully predicts blast dynamics in responders to SGI-110 treatment.

© Astex Pharmaceuticals, Inc.

Poster presented at: ASCPT Annual Meeting, New Orleans, LA March 3 - 7, 2015

Poster can be downloaded from www.astx.com