No 4076

SGI-110, a Novel Subcutaneous (SQ) Second Generation DNA Hypomethylating Agent Achieves Improved Pharmacodynamics (PD), Safety and Pharmacokinetics (PK) in comparison to IV Decitabine (DAC) in a non-human primate in vivo study

Pietro Taverna, Jason Scholl, Chongtie Shi, Aram Oganesian, Sanjeev Redkar, and Mohammad Azab

Astex Pharmaceuticals Inc., Dublin, CA, USA

Introduction

➢SGI-110 is a dinucleotide of decitabine (DAC) and deoxyguanosine designed to be more stable than decitabine to deamination by cytidine deaminase, thus offering a promising alternative to current hypomethylating agents approved in MDS.

➢We report here the results of a preclinical study in which safety, PK, and PD of different dosing regimens of SGI-110 SQ were compared to the clinical dose and regimen of DAC IV in 4 groups of male cynomolgus monkeys (n=3).

Study Design

➢The treatment groups consisted of:

1. Control group of DAC IV 1-h infusion at a dose equivalent to the clinically approved regimen of 20 mg/m² x 5 (1.7 mg/kg daily x 5);
2. 1.7 mg/kg SGI-110 SQ daily x 5 (molar equivalent to 42% of the clinical DAC dose);
3. 3.0 mg/kg SGI-110 SQ daily x 5 (molar equivalent to 75% of the clinical DAC dose);
4. 3.0 mg/kg of SGI-110 SQ once weekly x3 (molar equivalent to 44% of the total clinical DAC dose).

➢DAC and SGI-110 plasma levels were measured and monkeys were monitored for 28 days for hematological changes, and global DNA methylation (LINE-1).

Myelosuppression

➢Reversible hematological changes included a reduction in leukocytes, red blood cells (RBCs), and neutrophil counts with the nadir counts generally occurring between D8 and D14 and recovery occurring D21 to D28.

➢The DAC-treated group showed the greatest reduction and slowest recovery compared to all SGI-110 treated groups suggesting better safety for SGI-110.

Erythrocytes

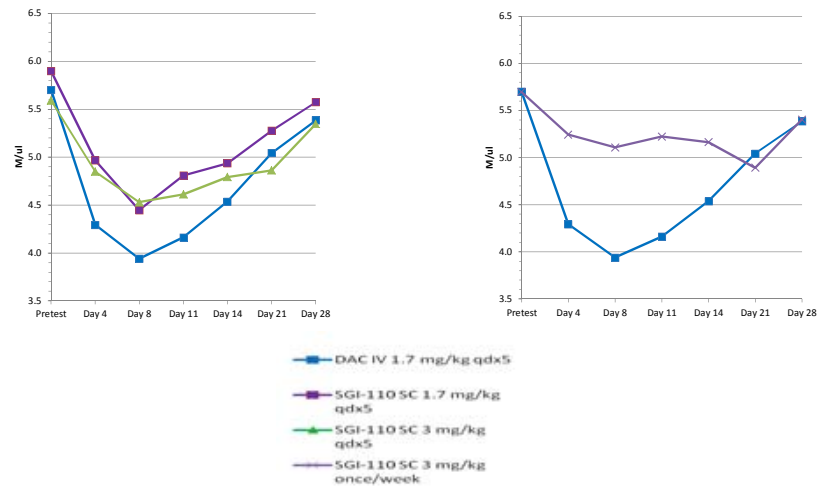
M/ul
Pretest
Day 4
Day 8
Day 11
Day 14
Day 21
Day 28
DAC IV 1.7 mg/kg qdx5
SGI-110 SC 1.7 mg/kg qdx5
SGI-110 SC 3 mg/kg qdx5
SGI-110 SC 3 mg/kg once/week

- Erythrocytes decrease in 5-day dosing groups, with smaller changes observed in the once weekly SGI-110 group
- Animals receiving 1.7 mg/kg DAC appear to have the greatest changes in RBC levels

Neutrophils

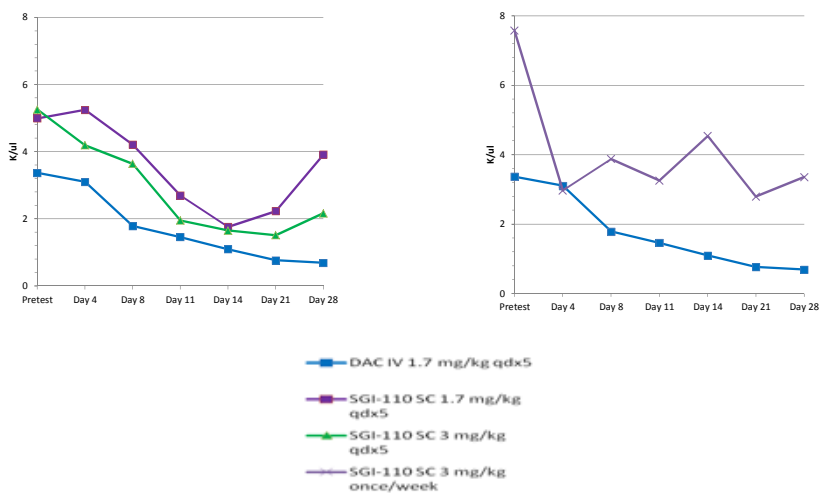
K/ul
Pretest
Day 4
Day 8
Day 11
Day 14
Day 21
Day 28
DAC IV 1.7 mg/kg qdx5
SGI-110 SC 1.7 mg/kg qdx5
SGI-110 SC 3 mg/kg qdx5
SGI-110 SC 3 mg/kg once/week

- Neutrophil decrease in all groups but do not recover in the 1.7 mg/kg DAC group

Pharmacodynamics of Hypomethylation

➢Changes in methylation patterns of LINE-1 and microRNA 29b (miRNA29b) were evaluated in DNA extracted from monkey blood as PD markers of biological efficacy after treatment with SGI-110 or DAC.

➢All treatment groups achieved a decrease in LINE-1 methylation.

➢SGI-110 3 mg/kg SQ (75% of the clinical DAC dose) or 1.7 mg/kg SQ (42% of the clinical DAC dose) on a daily x 5 regimen achieved slightly more or the same hypomethylation than DAC IV between day 8 and 11.

➢SGI-110 3.0 mg/kg SQ once weekly x 3 (44% of the total clinical DAC dose) achieved approximately 10% demethylation from baseline at day 8 and this effect was maintained until day 15.

➢Similar results, in terms of extent and duration of hypomethylation, were observed when DNA methylation was assessed on the sequence of miR29b.

➢Overall, the magnitude and duration of the decrease in LINE1 methylation at lower molar equivalent doses of SQ daily SGI-110 were similar to or better than DAC IV.

LINE1 Demethylation

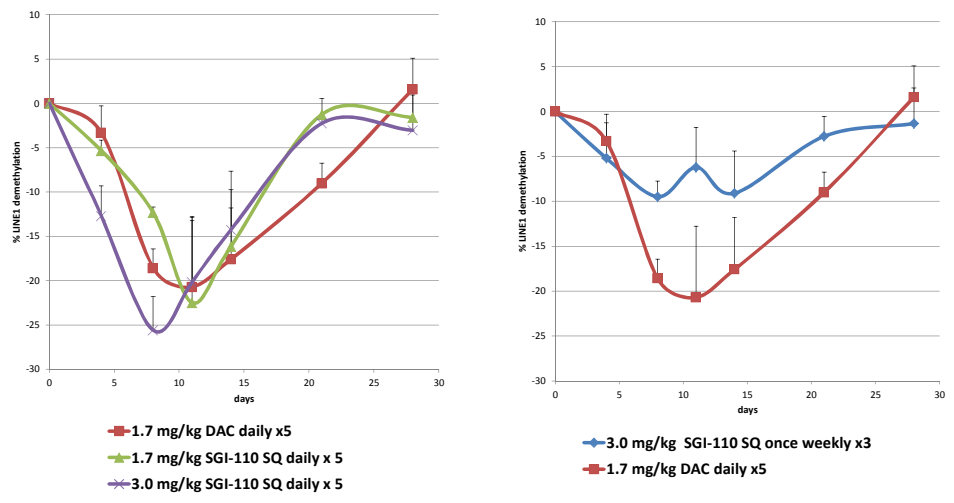
% LINE1 demethylation
days
1.7 mg/kg DAC daily x5
1.7 mg/kg SGI-110 SQ daily x 5
3.0 mg/kg SGI-110 SQ daily x 5
3.0 mg/kg SGI-110 SQ once weekly x3
1.7 mg/kg DAC daily x5

miR29b Demethylation

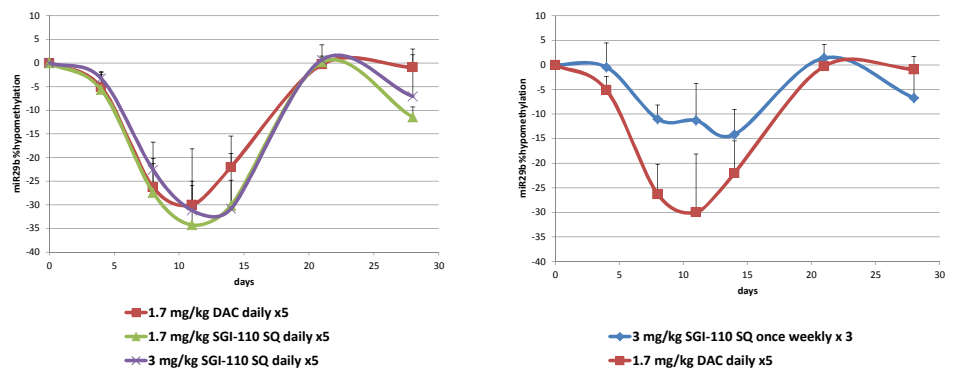
miR29b %hypomethylation
days
1.7 mg/kg DAC daily x5
1.7 mg/kg SGI-110 SQ daily x5
3 mg/kg SGI-110 SQ daily x5
3 mg/kg SGI-110 SQ once weekly x 3
1.7 mg/kg DAC daily x5

Pharmacokinetics

➢SGI-110 appeared to convert to DAC resulting in similar exposure window compared to IV DAC.

➢The dose-adjusted plasma DAC exposures, on molar equivalence basis, were somewhat lower compared to the DAC group.

➢For SGI-110 SQ groups, C_{max} ranged 157-200 (1.7 mg/kg) or 190-378 ng/mL (3 mg/kg) for SGI-110; and 52-82 (1.7 mg/kg) or 60-163 ng/mL for decitabine. C_{max} for IV DAC was 215-525 ng/mL

➢For SGI-110 SQ groups, SGI-110 AUC ranged 90-101 (1.7 mg/kg) or 123-324 (3 mg/kg) ng*hr/mL; while decitabine AUCs were 37-68 (1.7mg/kg) or 69-155 (3mg/kg). AUC for IV DAC was 99-221.

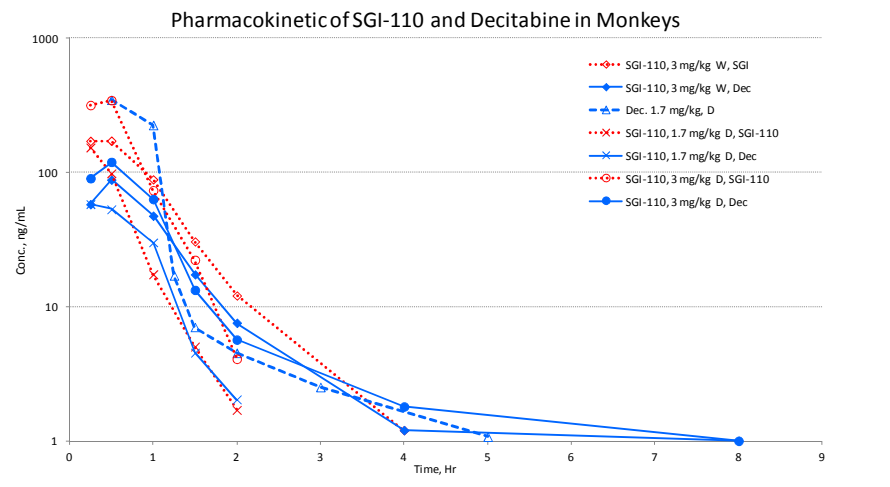
Pharmacokinetic of SGI-110 and Decitabine in Monkeys
Conc., ng/mL
Time, Hr
SGI-110, 3 mg/kg W, SGI
SGI-110, 3 mg/kg W, Dec
Dec. 1.7 mg/kg, D
SGI-110, 1.7 mg/kg D, SGI-110
SGI-110, 1.7 mg/kg D, Dec
SGI-110, 3 mg/kg D, SGI-110
SGI-110, 3 mg/kg D, Dec

- SQ SGI-110 delivers decitabine with a lower C_{max} over 5 h
- 1h IV infusion of DAC yields a higher C_{max}

Conclusions

➢SQ SGI-110 may represent a more convenient and tolerable option for delivering decitabine and achieving DNA hypomethylating effects at lower doses that were similar to or better than those seen with decitabine IV and with less myelosuppressive effects.

➢A randomized Phase 1-2 FIH PK/PD-guided, dose-escalation study is being conducted in subjects with relapsed/refractory intermediate or high-risk MDS or AML. ClinicalTrials.gov identifier: NCT01261312

➢Subjects are randomized to one of two SQ regimens (daily x 5 or once weekly x 3, both given in 28-day courses).

© Astex Pharmaceuticals Inc.

Poster presented at AACR Annual Meeting, Chicago, IL March 31 – April 4, 2012.

SGI-110-PB(1203)