No A55

Potent, Dual cIAP1/XIAP Antagonists Induce Apoptosis in a Melanoma Stem Cell Population

Maria Ahn, George Ward, Gianni Chessari, Keisha Hearn, Christopher N. Johnson, Jon Lewis, Neil Thompson, Pamela Williams

Astex Pharmaceuticals, 436 Cambridge Science Park, Milton Road, Cambridge, CB4 0QA, United Kingdom

INTRODUCTION

Melanoma is a highly aggressive malignancy with an exceptional ability to develop resistance. Inhibitor of apoptosis proteins (IAP) play a key role in preventing cell death by apoptosis. IAPs are highly regulated by endogenous antagonists (e.g. SMAC) but in melanoma, expression levels of IAPs are generally high and depleting IAPs tended to reduce cell viability, with XIAP reduction having the greatest effect [1]. Cancer Stem Cell (CSC) populations are more apoptosis resistant than the bulk cell population and they have been associated with resistance to cancer therapy, relapse and disease progression. Blockade of the apoptotic pathway by up-regulation of anti-apoptotic factors has been implicated in conferring resistance in CSC fractions and increased XIAP expression has also been reported in these cells [2,3]. Here we have set out to determine whether our more potent dual cIAP/XIAP antagonists have enhanced apoptosis-inducing capacity on melanoma CSC-like fractions.

FIRST GENERATION OF IAP ANTAGONISTS

- All candidate molecules in development have been derived via peptidomimetic approach based on SMAC peptide sequence (AVPI), which contains an alanine warhead and has a selectivity for cIAP1 over XIAP

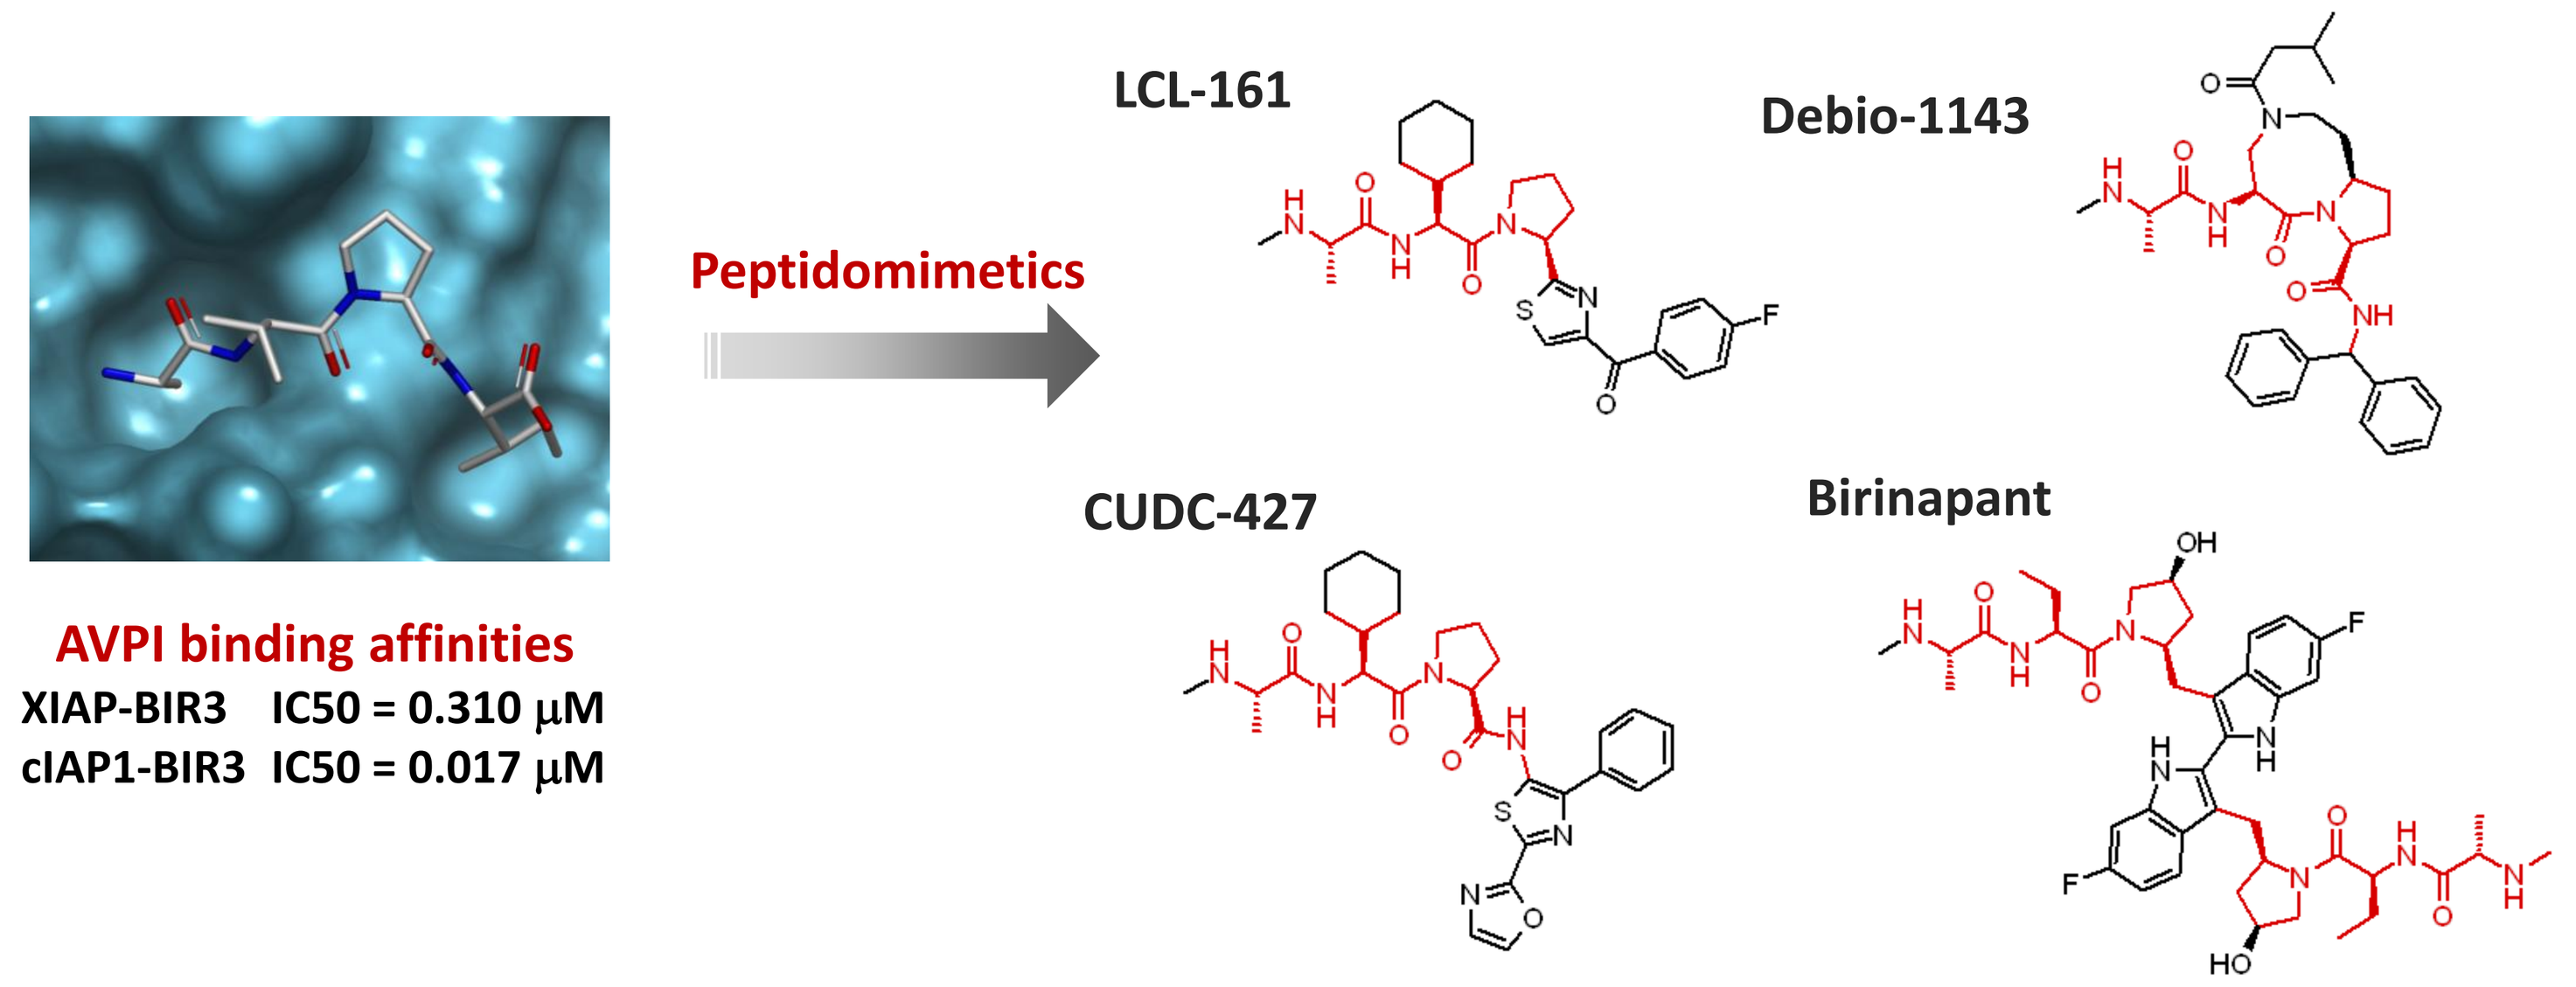
Peptidomimetics
LCL-161
Debio-1143
CUDC-427
Birinapant
AVPI binding affinities
XIAP-BIR3 IC50 = 0.310 µM
cIAP1-BIR3 IC50 = 0.017 µM

IAP ANTAGONISTS DERIVED FROM FRAGMENT-BASED DRUG DISCOVERY

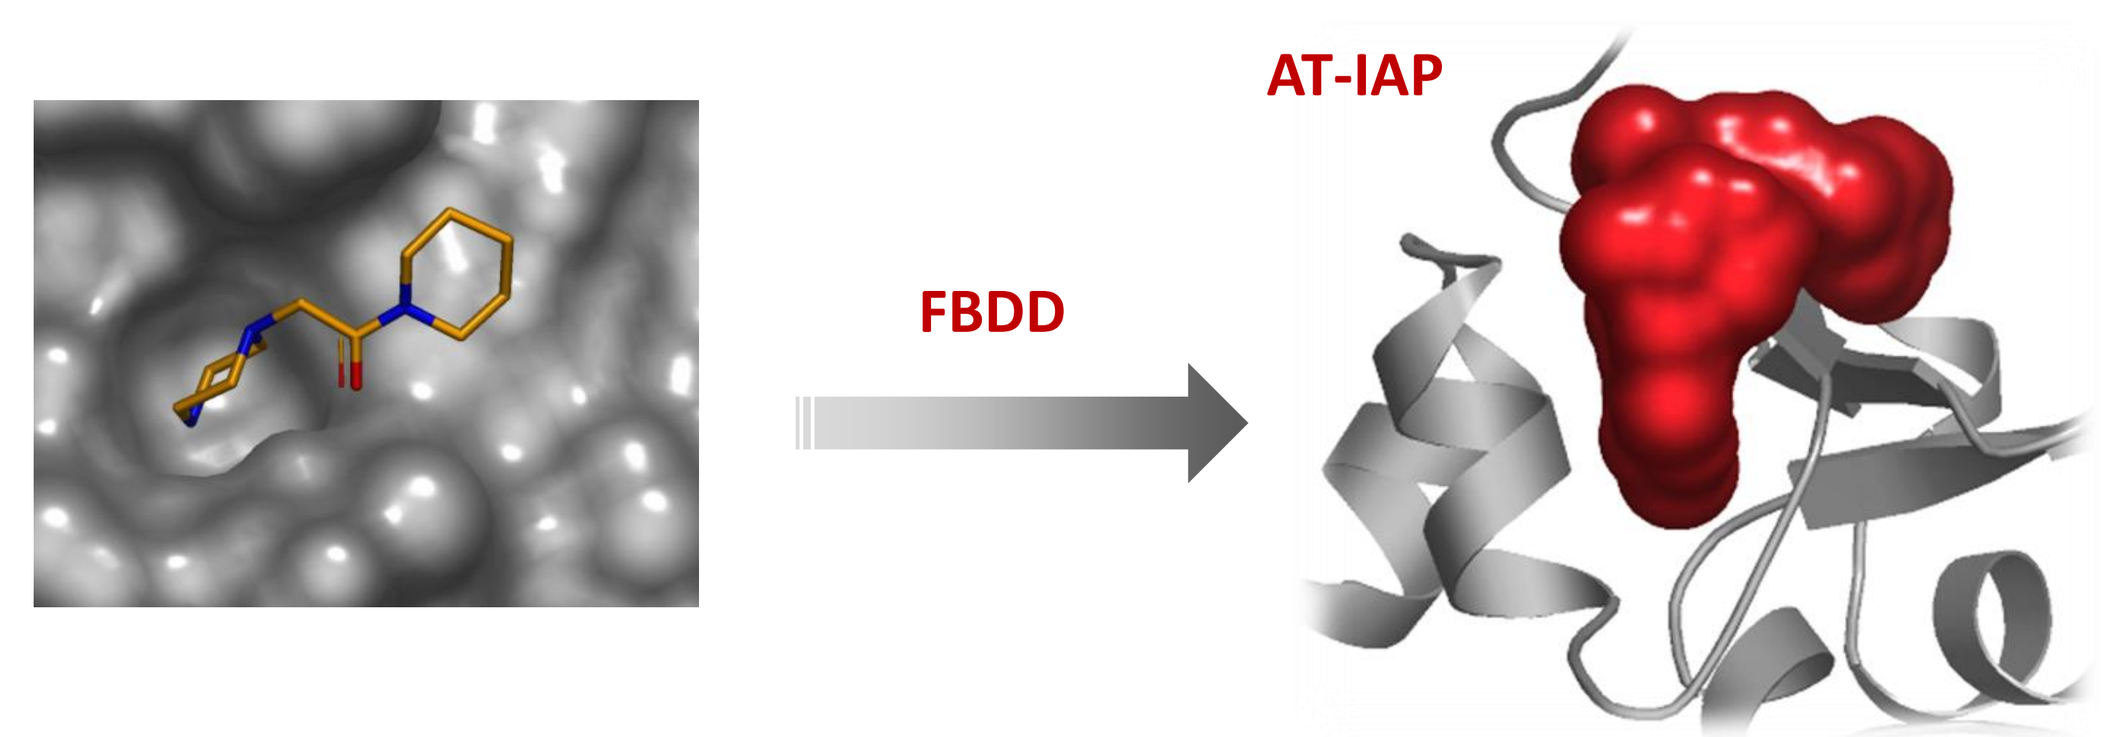
FBDD
AT-IAP

- Balanced cIAP1/XIAP profile
- Non peptidomimetics
- Non alanine wahead
- Oral

	Assay (IC_{50} in nM)	AT-IAP	Cmpd 1	LCL-161	Debio-1143	Birinapant
XIAP cellular activity	HEK293-XIAP-Caspase9 (I.P.)	5.1	3.9	35	35	23
cIAP1 cellular activity	MDA-MB-231 (cIAP1 degradation)	0.32	0.28	0.40	0.99	0.23
Proliferation	MDA-MB-231	4.2	1.8	7.8	18	0.98
	HCT-116 (control)	inactive	inactive	inactive	inactive	inactive

NON-ALANINE WARHEAD IMPORTANT FOR BALANCED cIAP1/XIAP PROFILE

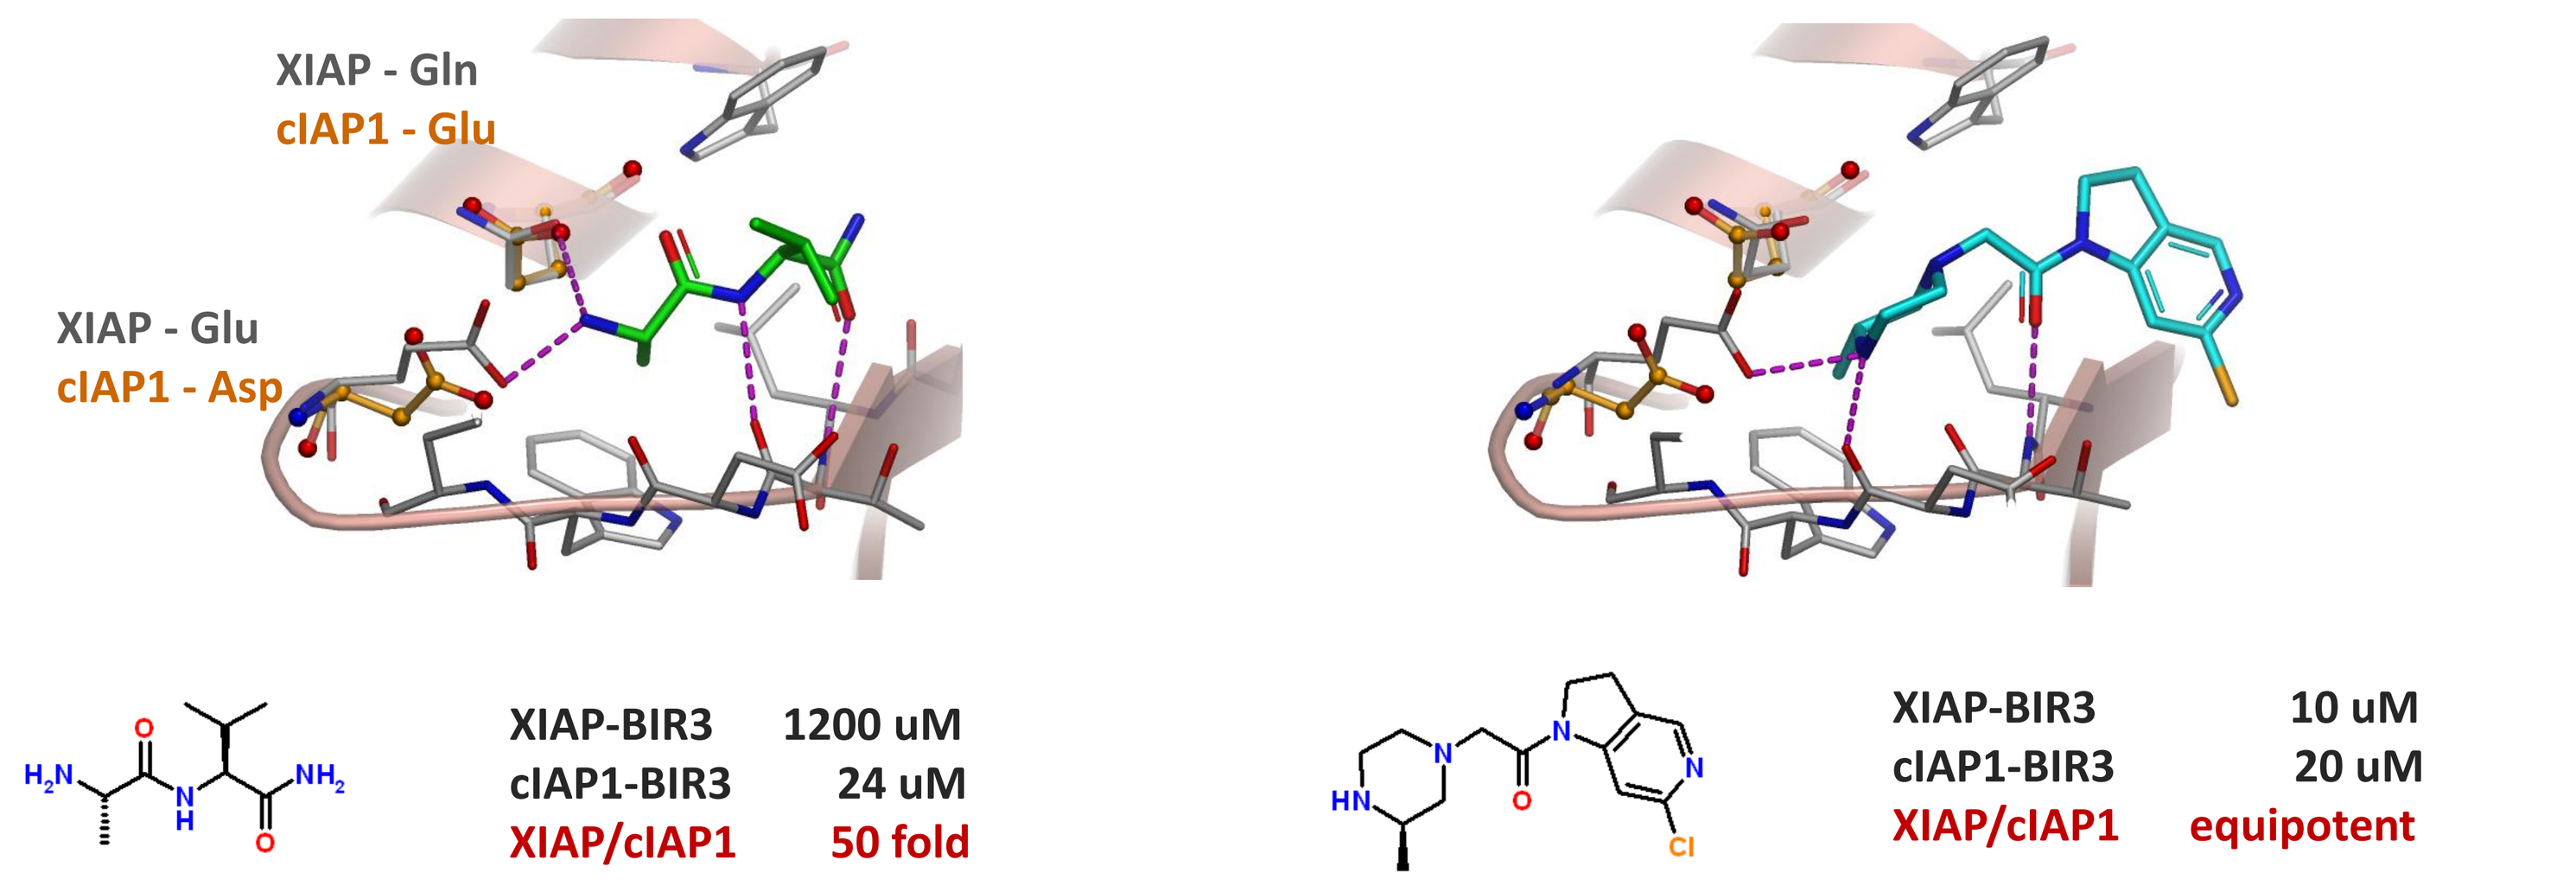
XIAP - Gln
cIAP1 - Glu
XIAP - Glu
cIAP1 - Asp
XIAP-BIR3 1200 uM
cIAP1-BIR3 24 uM
XIAP/cIAP1 50 fold
XIAP-BIR3 10 uM
cIAP1-BIR3 20 uM
XIAP/cIAP1 equipotent

- X-ray crystal structures of alanine fragment (green) and optimised piperazine fragment (cyan) in XIAP-BIR3 overlaid with key amino acid residues from the cIAP1-BIR3 protein crystal structure (orange), with H-bonds highlighted (purple)
- The alanine warhead forms charge-charge interactions with two acidic residues in cIAP1, but with only one acidic residue in XIAP
- The piperazine warhead forms charge-charge interactions with only one acidic residue in both cIAP1 and XIAP, together with H-bonds to the conserved backbone, hence it has an inherently more balanced profile

AT-IAP ACTIVITY IN MELANOMA CELL LINES AND PRIMARY TUMORS

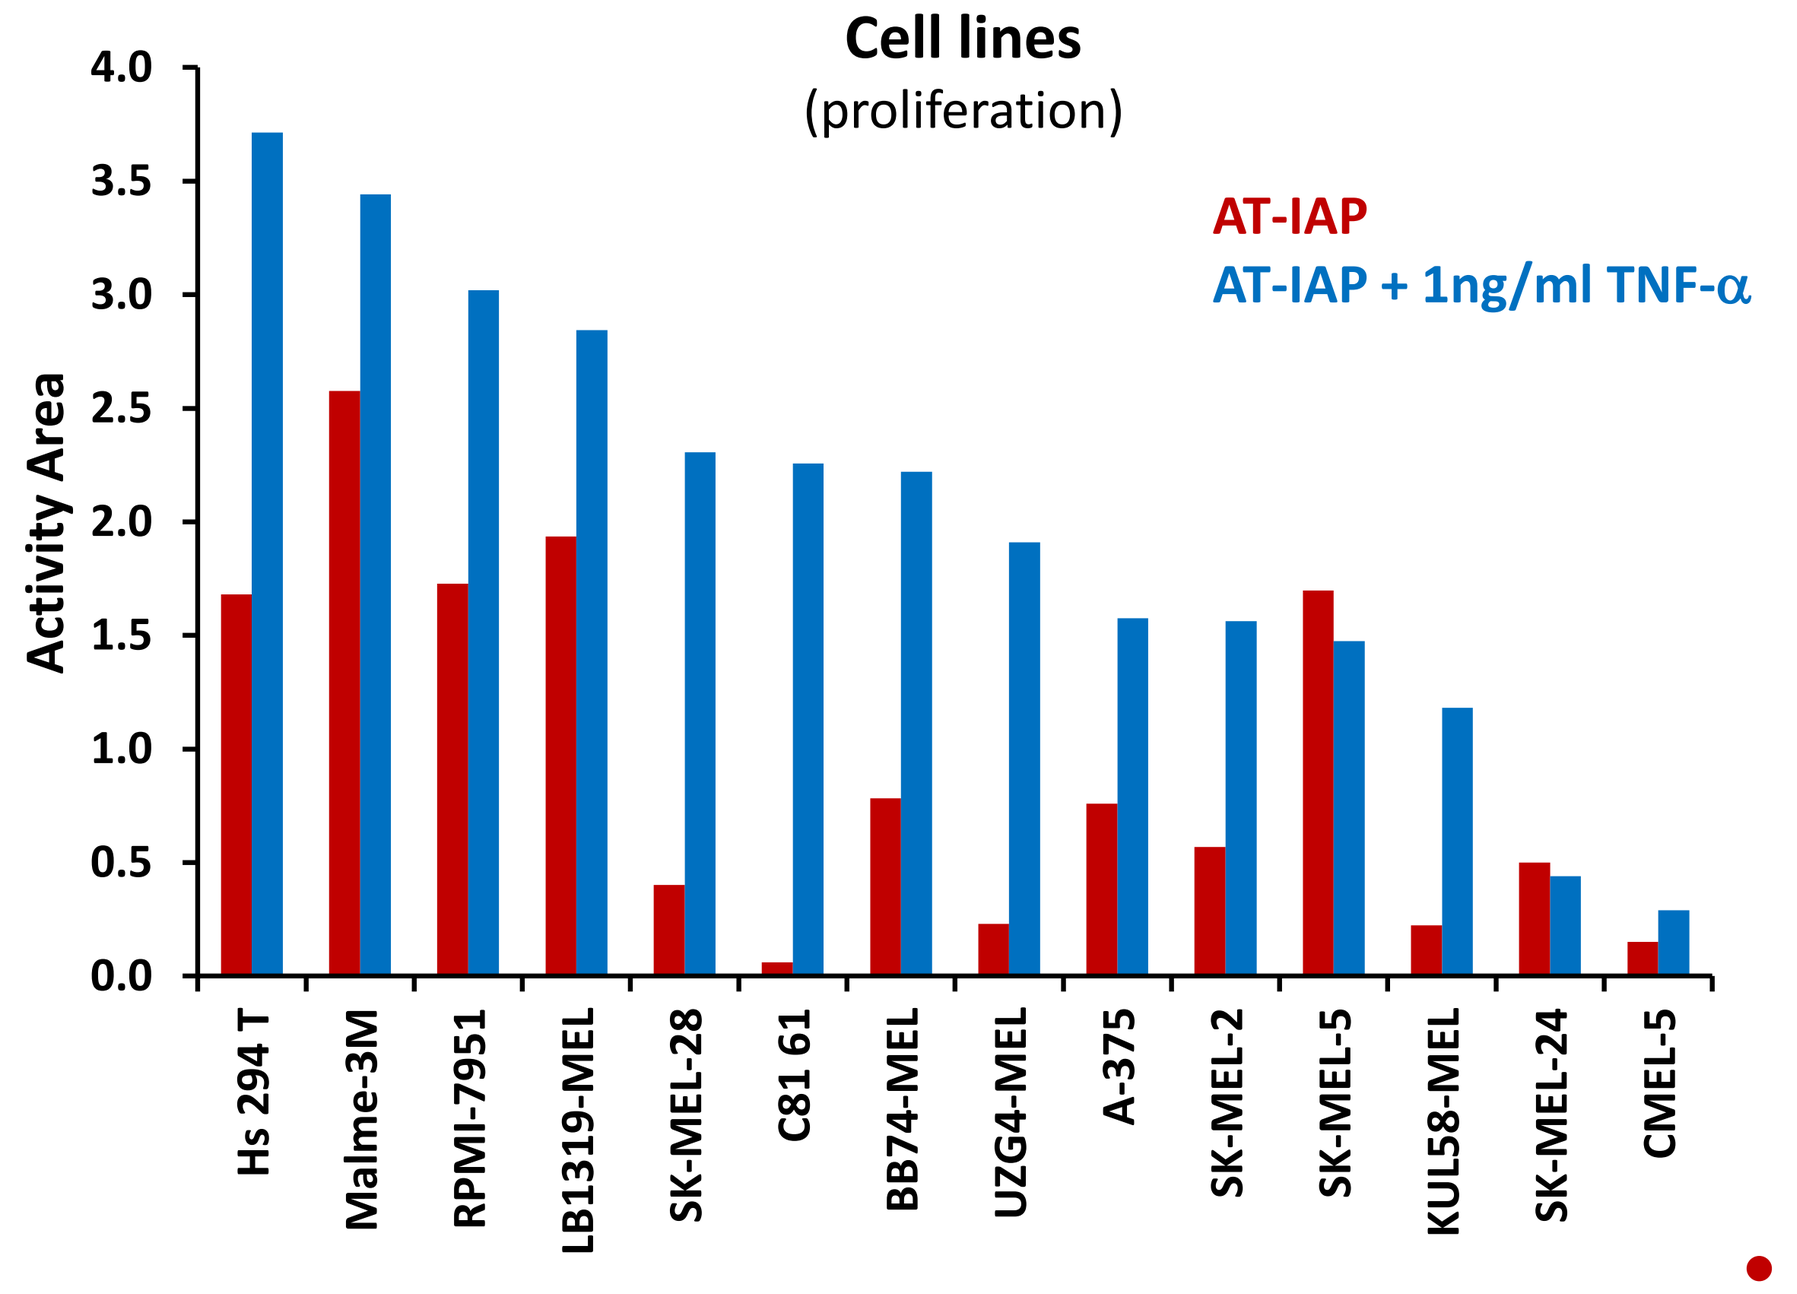
Cell lines
(proliferation)
AT-IAP
AT-IAP + 1ng/ml TNF-α
Activity Area
Hs 294 T
Malme-3M
RPMI-7951
LB1319-MEL
SK-MEL-28
C81 61
BB74-MEL
UZG4-MEL
A-375
SK-MEL-2
SK-MEL-5
KUL58-MEL
SK-MEL-24
CMEL-5

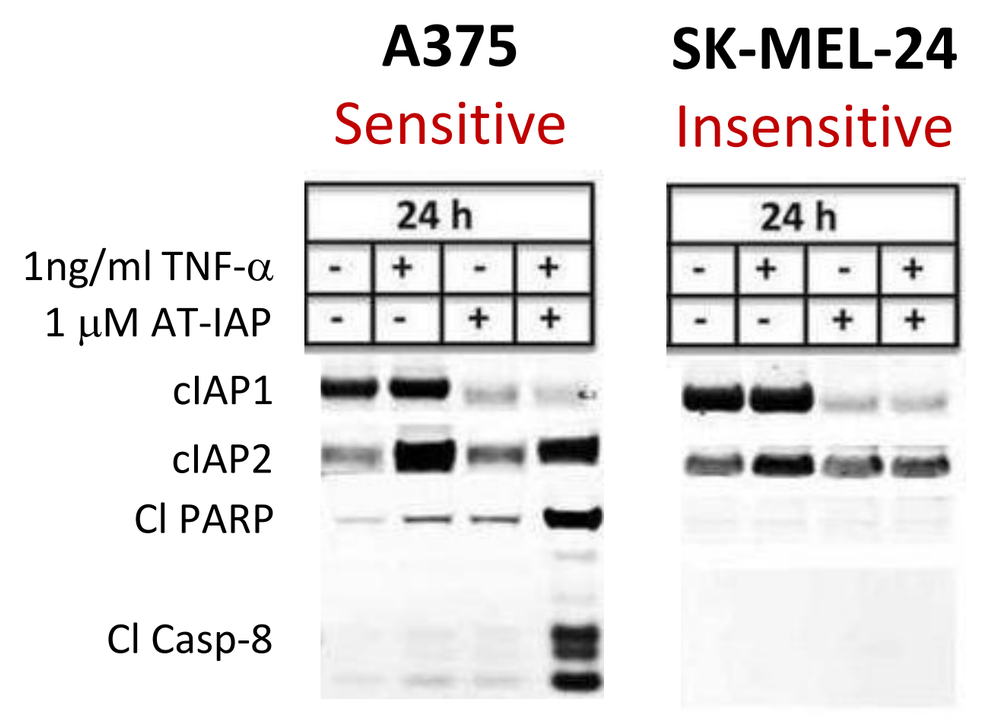
A375
Sensitive
SK-MEL-24
Insensitive
24 h
1ng/ml TNF-α
1 µM AT-IAP
cIAP1
cIAP2
Cl PARP
Cl Casp-8

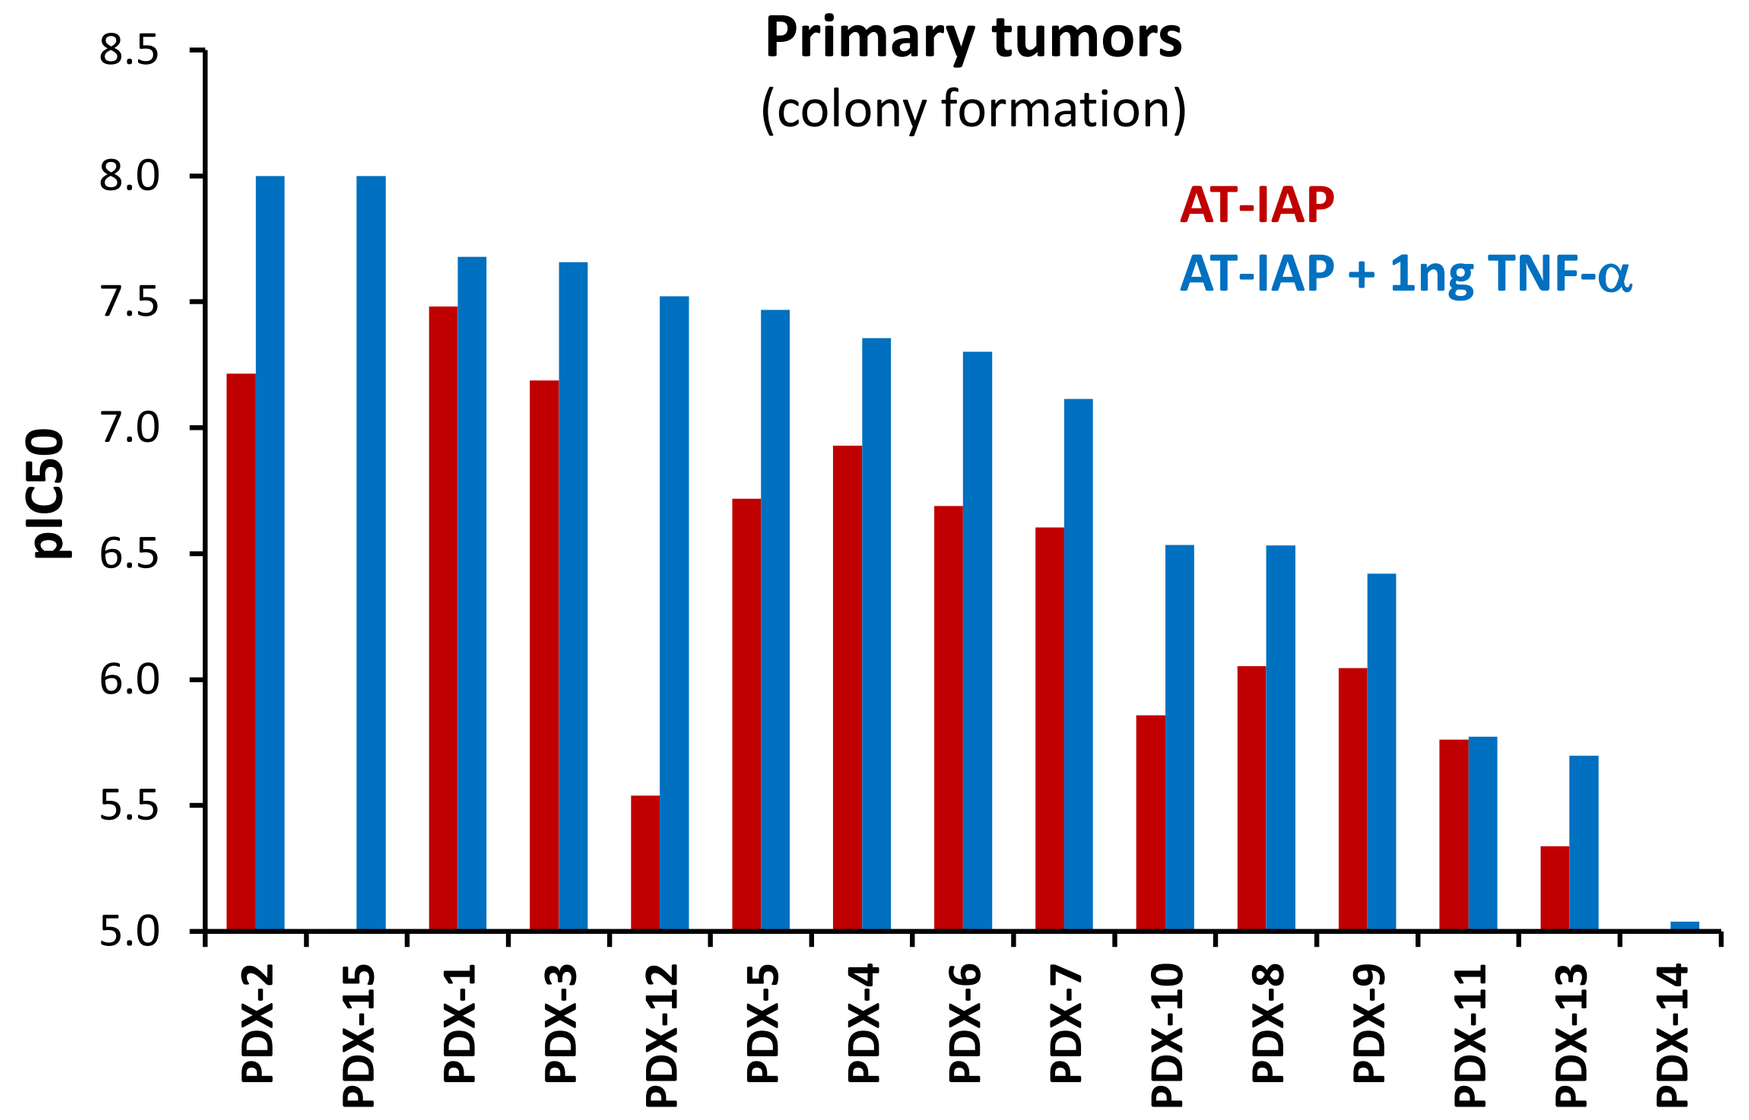
Primary tumors
(colony formation)
AT-IAP
AT-IAP + 1ng TNF-α
pIC50
PDX-2
PDX-15
PDX-1
PDX-3
PDX-12
PDX-5
PDX-4
PDX-6
PDX-7
PDX-10
PDX-8
PDX-9
PDX-11
PDX-13
PDX-14

- cIAP1 degradation observed in all cell lines – sensitive or insensitive
- Sensitivity in melanoma cell lines is driven by the ability of AT-IAP to switch TNF-α signalling from pro-survival to pro-apoptotic.
- TNF-α can be endogenously produced by the cell line
- TNF-α can also be produced by inflammatory cells present in tumour micro-environment

CANCER STEM CELL POPULATIONS IN MELANOMA CELL LINES

- $CD133^{+}$ cells in melanoma: show stem-like characteristics (i.e. able to self-renew and differentiate, and display enhanced tumourigenicity)

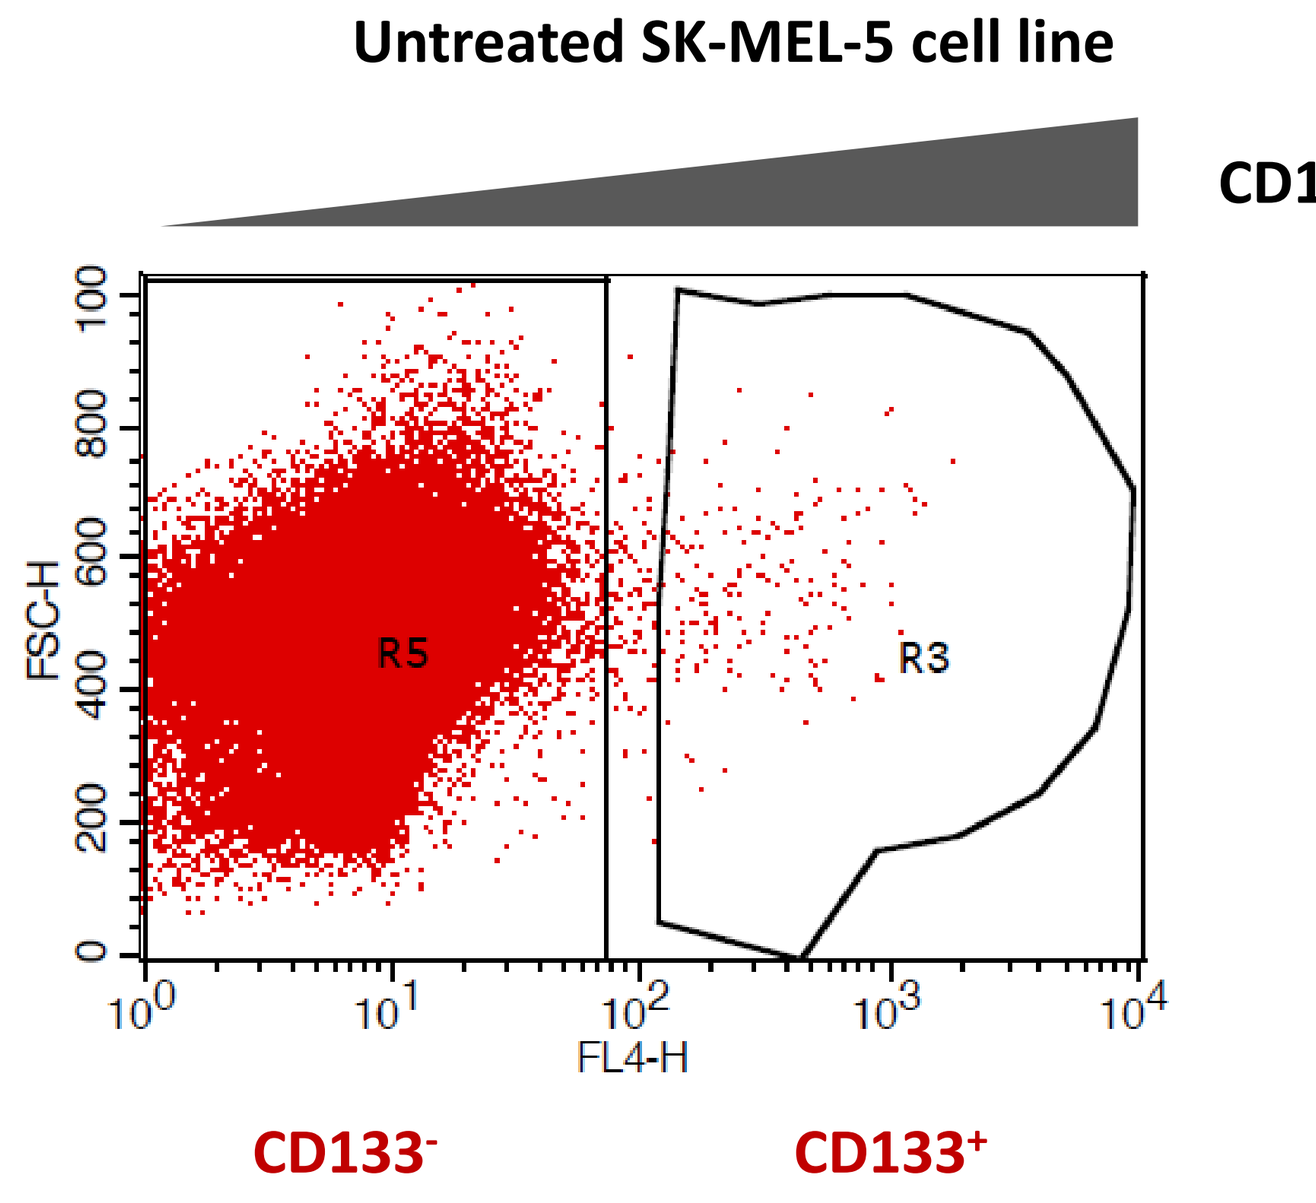
Untreated SK-MEL-5 cell line
CD133
FSC-H
FL4-H
R5
R3
CD133-
CD133+

Melanoma cell lines stained with anti CD133-APC from Miltenyi Biotech

Cell line	CD133+ cells
SK-MEL-2	1-2 %
SK-MEL-5	0.5-1 %
SK-MEL-28	2-5 %
A375	none

- Small sub-population of CD133+ CSCs were found in SK-MEL-5, SK-MEL-2 and SK-MEL-28 (but not A375) cell lines

DUAL cIAP1/XIAP ANTAGONIST TARGETS STEM CELL POPULATIONS

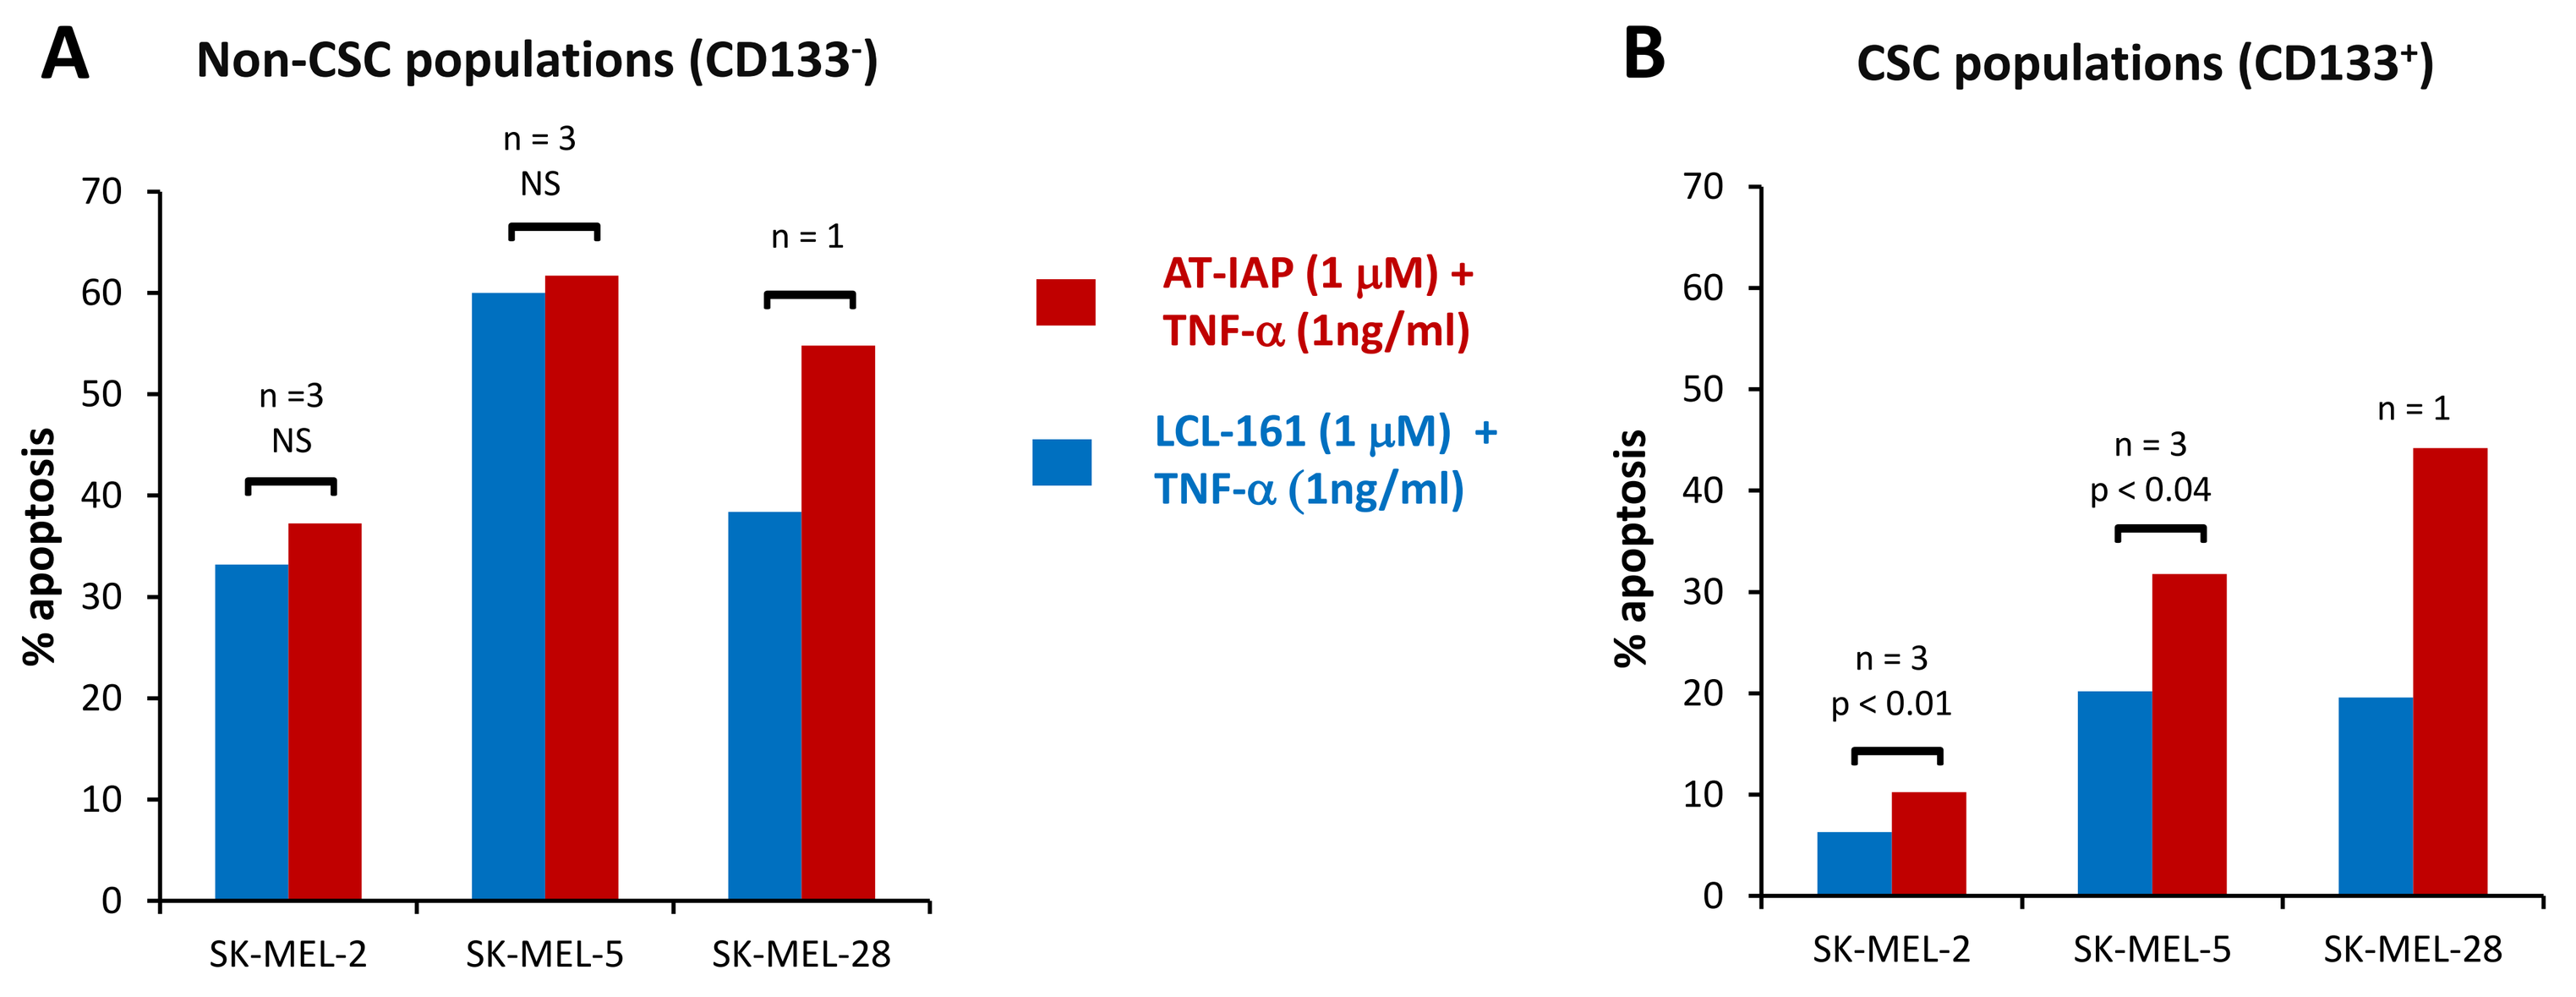
A
Non-CSC populations (CD133-)
B
CSC populations (CD133+)
% apoptosis
n =3
NS
n = 3
NS
n = 1
n = 3
p < 0.01
n = 3
p < 0.04
n = 1
AT-IAP (1 µM) + TNF-α (1ng/ml)
LCL-161 (1 µM) + TNF-α (1ng/ml)
SK-MEL-2
SK-MEL-5
SK-MEL-28

- Both **LCL-161** and **AT-IAP** are equivalent in inducing apoptosis in non-CSC populations (**A**)
- CSC are more resistant to apoptosis induced by IAP antagonists (**B**)
- The apoptosis induced by **AT-IAP** in CSC is significantly higher that the apoptosis induced by **LCL-161** (**B**, **D** and **E**)
- The dual cIAP1/XIAP antagonist **AT-IAP** induces apoptosis in CSC to levels approaching the apoptosis in normal cells (**B**)

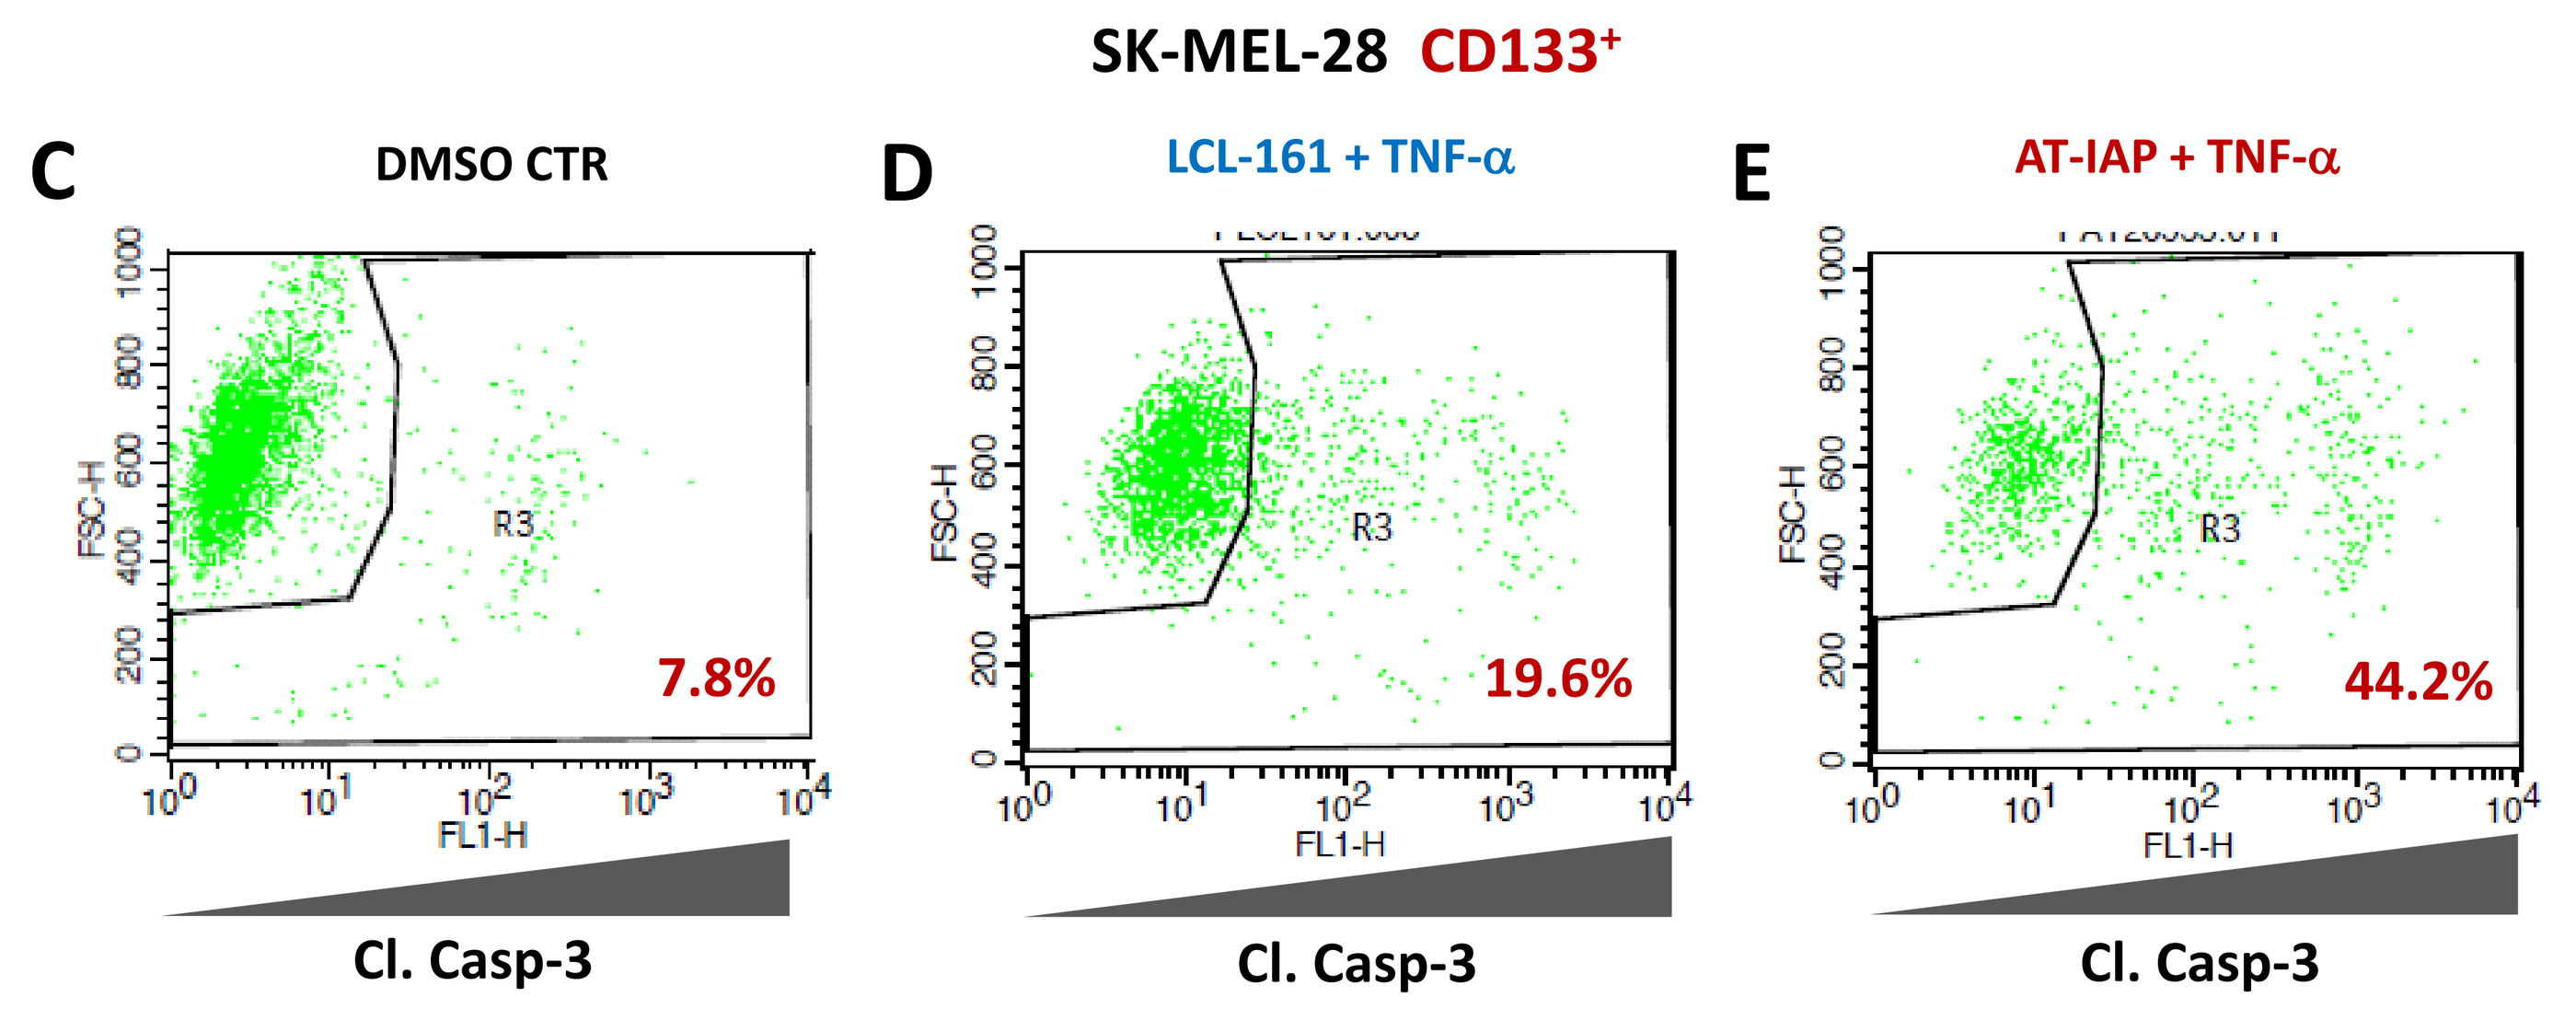
SK-MEL-28 CD133+
C
DMSO CTR
7.8%
D
LCL-161 + TNF-α
19.6%
E
AT-IAP + TNF-α
44.2%
R3
FSC-H
FL1-H
Cl. Casp-3

DUAL ANTAGONIST ENHANCES SENSITIVITY OF CSC TO VEMURAFENIB

- SK-MEL-28 is a BRAF V600E mutant cell line
- Enhanced apoptosis in SK-MEL-28, both total and CSC-fraction, when the dual antagonist **Compound 1** is combined with vemurafenib

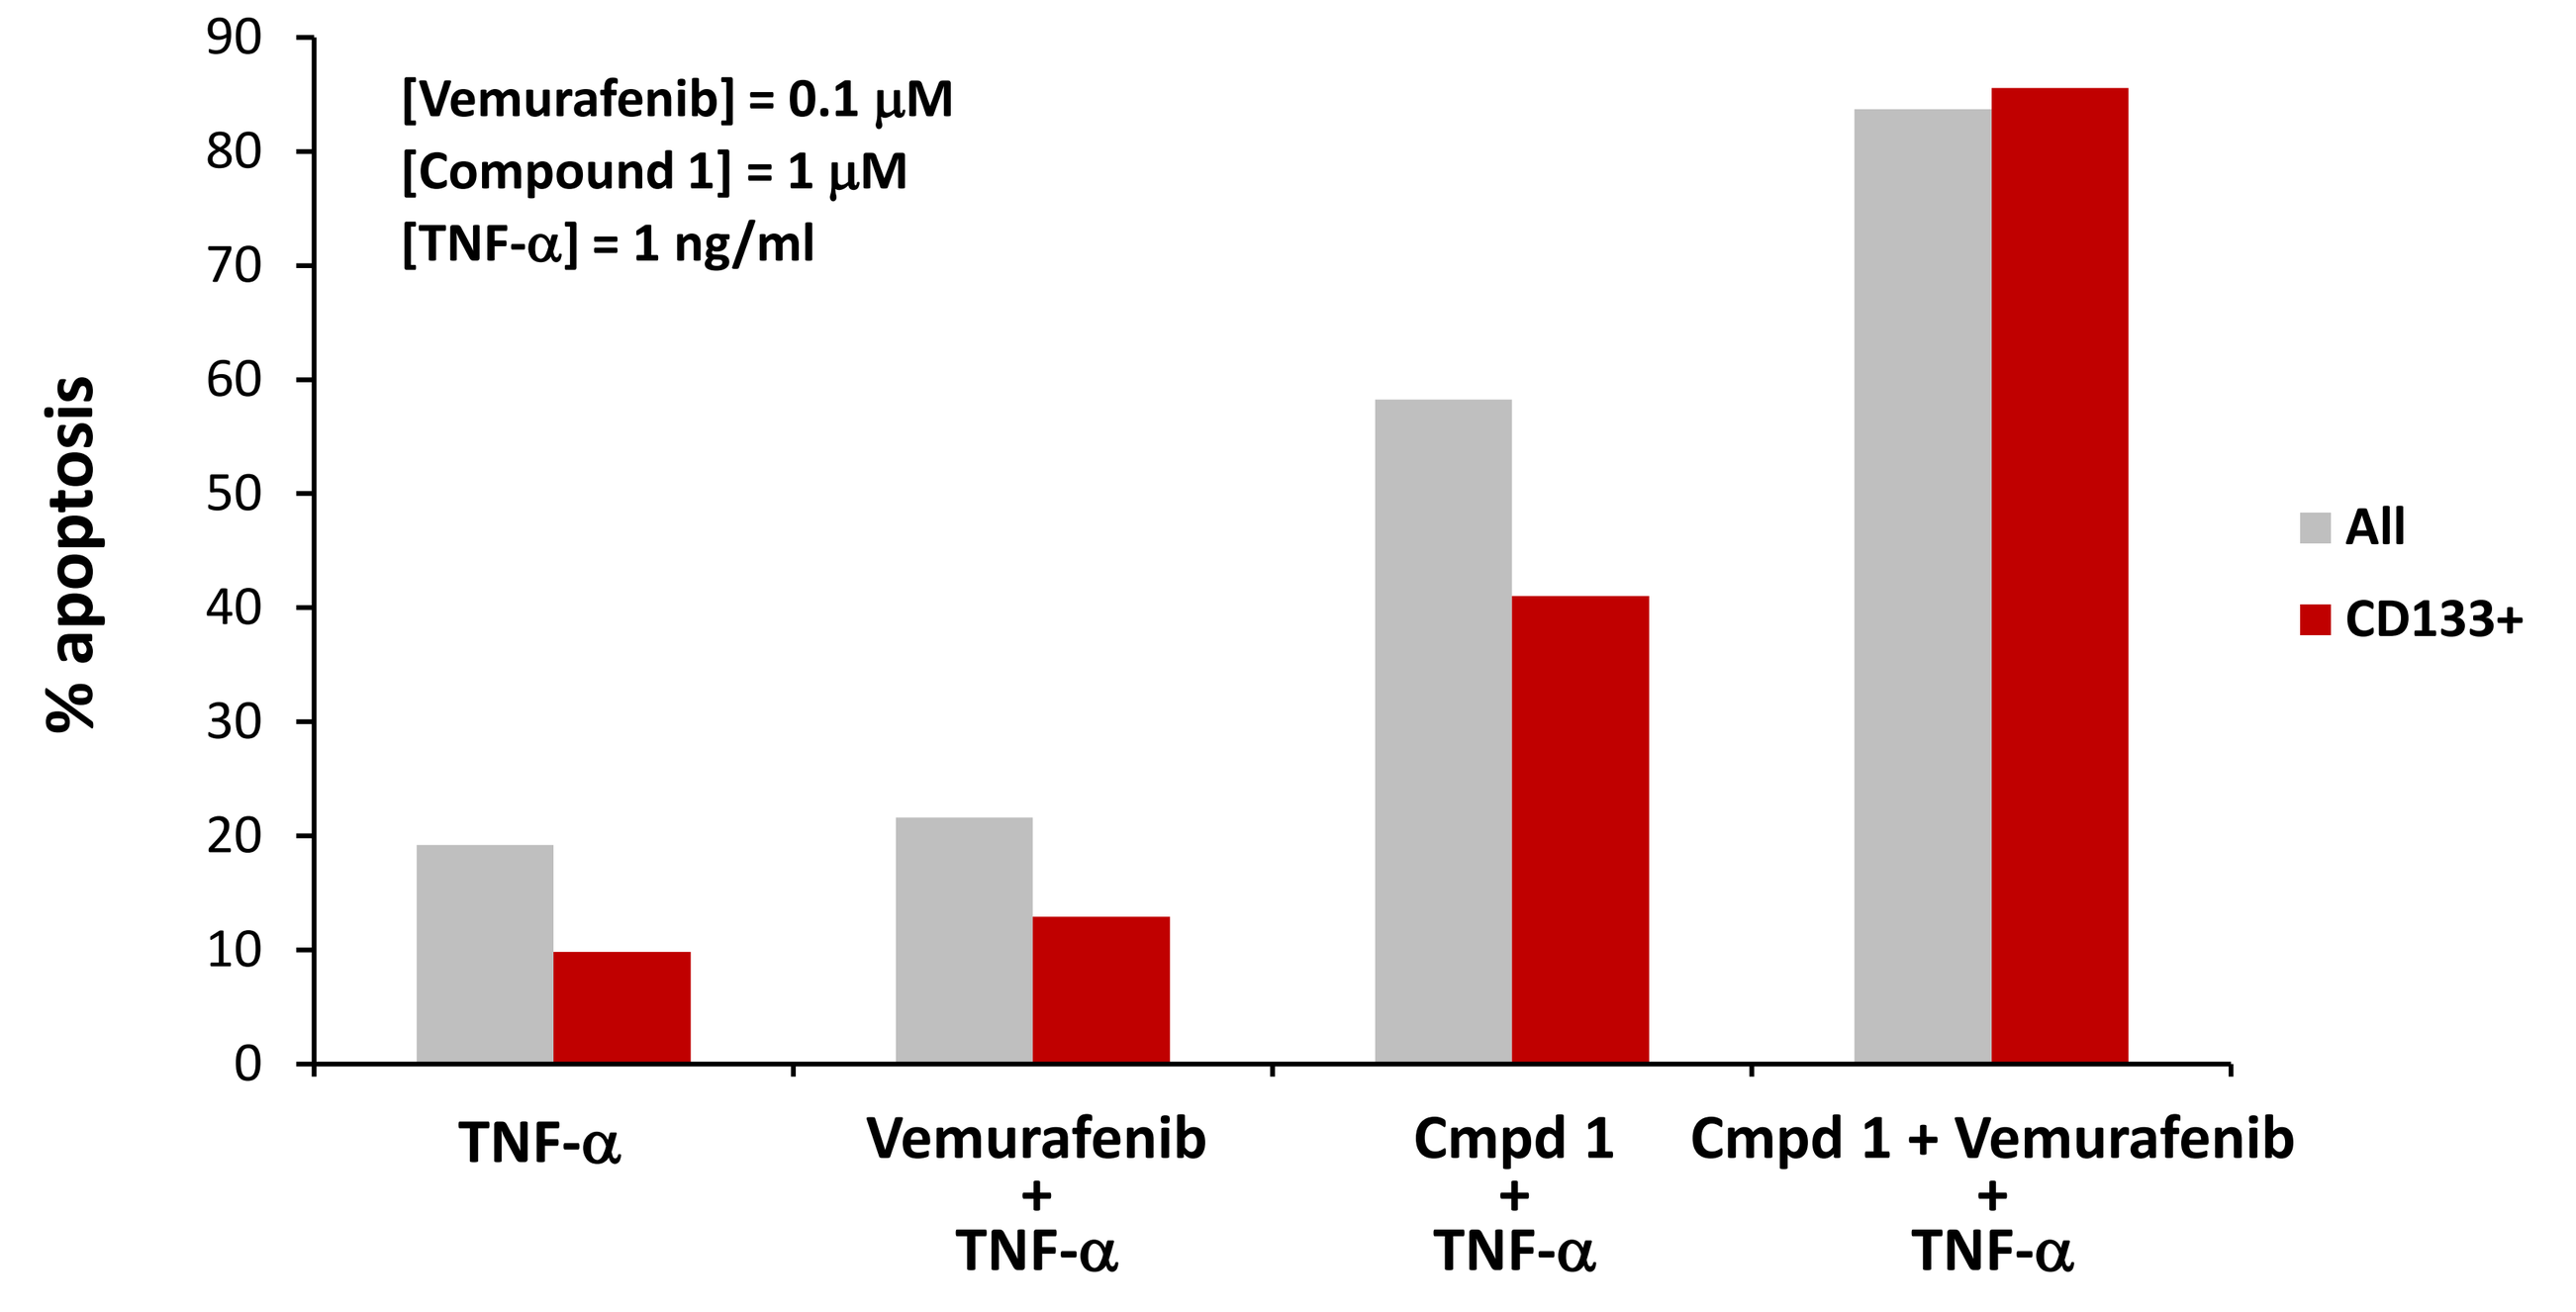
[Vemurafenib] = 0.1 µM
[Compound 1] = 1 µM
[TNF-α] = 1 ng/ml
% apoptosis
All
CD133+
TNF-α
Vemurafenib + TNF-α
Cmpd 1 + TNF-α
Cmpd 1 + Vemurafenib + TNF-α

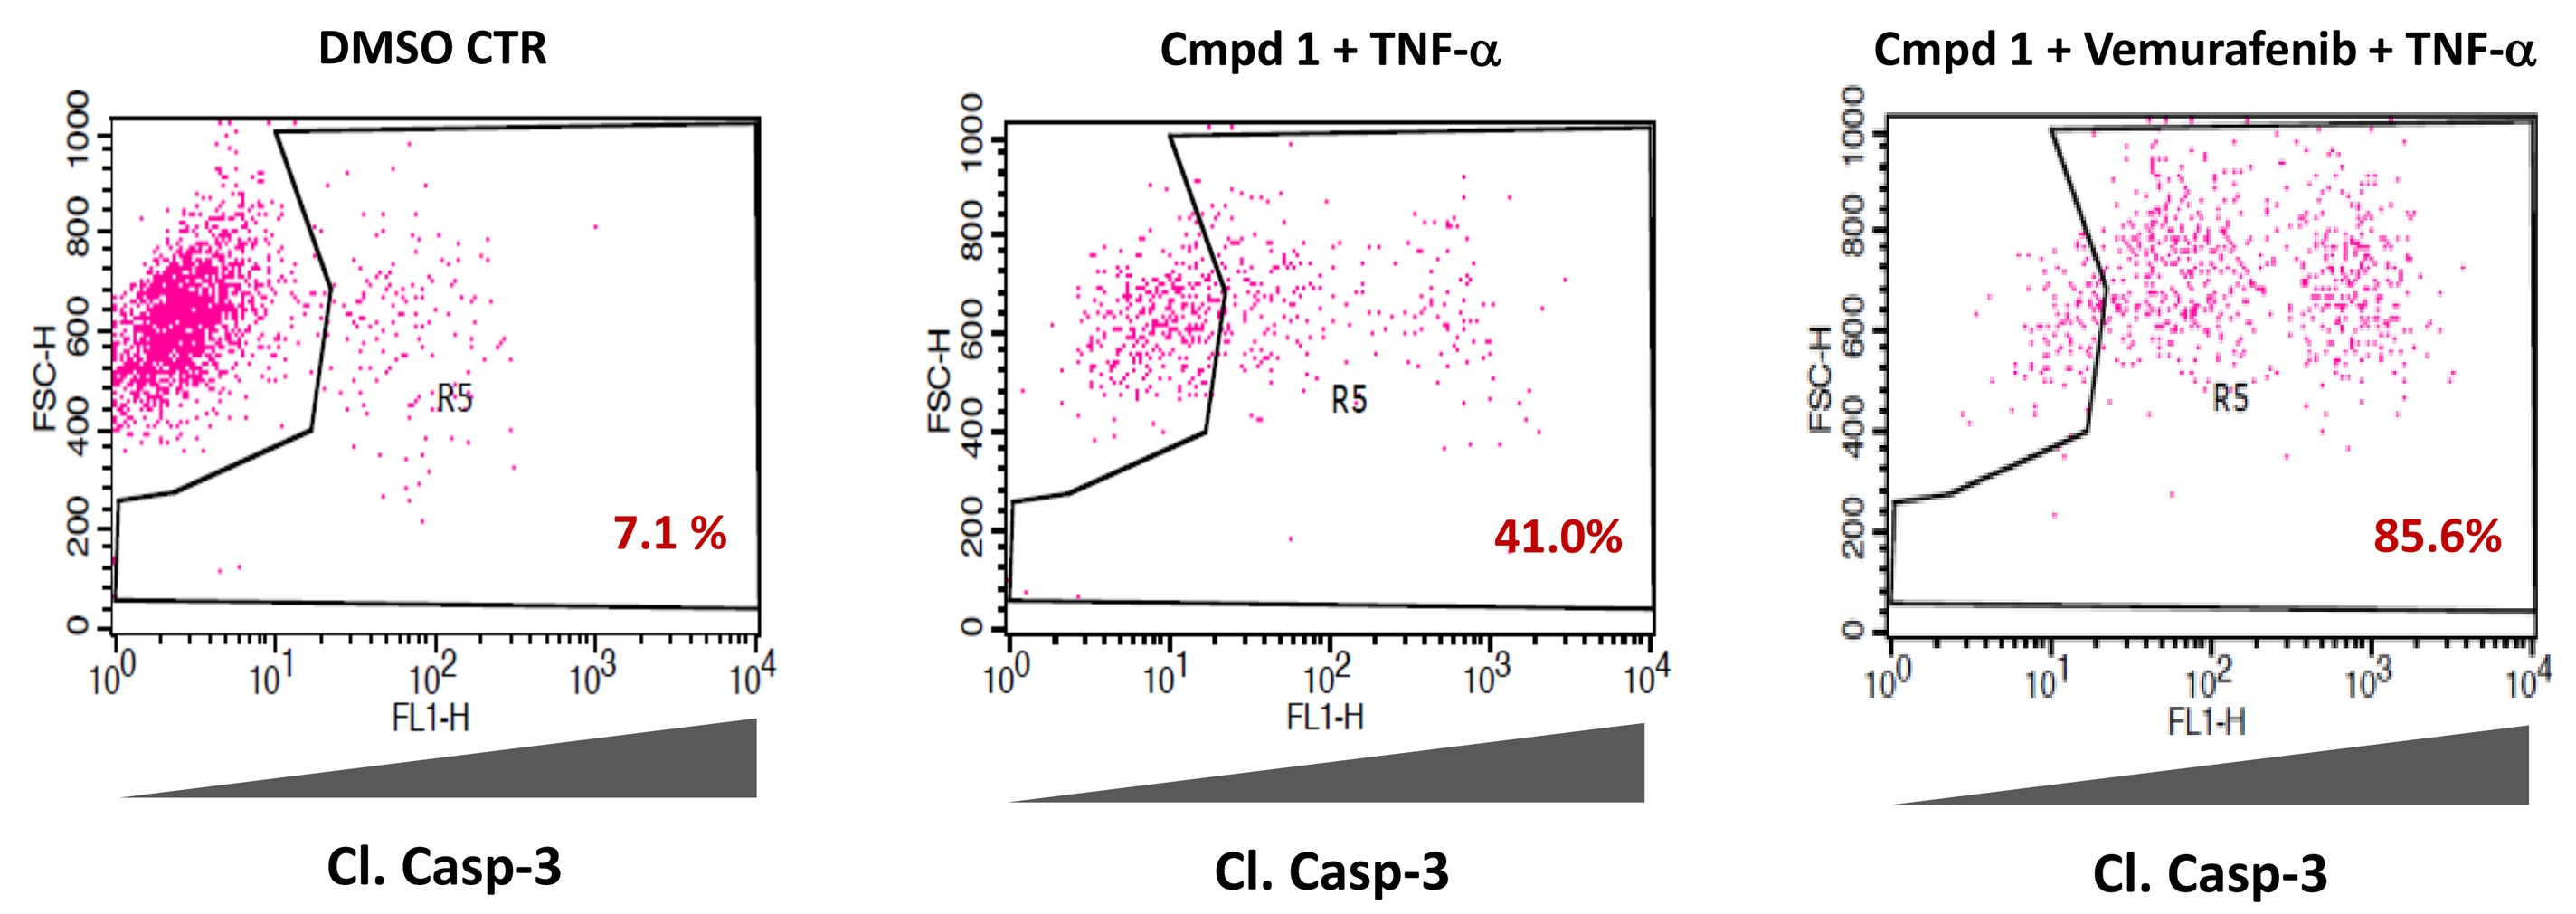
DMSO CTR
7.1 %
Cmpd 1 + TNF-α
41.0%
Cmpd 1 + Vemurafenib + TNF-α
85.6%
R5
FSC-H
FL1-H
Cl. Casp-3

CONCLUSIONS

- **AT-IAP** and **Compound 1** represent a novel class of IAP antagonists with a potent dual cIAP1 and XIAP antagonist profile
- In vitro cell line testing suggested significant activity against a panel of melanoma primary tumors and cell lines, which was enhanced on addition of exogenous TNF-α
- Our preliminary studies have shown that **AT-IAP** has enhanced apoptosis-inducing capacity in melanoma CSC-fractions
- Combination of IAP antagonism with vemurafenib enhances apoptosis in both total and CSC fractions

REFERENCES

[1] Engesaeter et al., Cancer Biology & Therapy, 2011, 12 (1), 47
[2] Bergmann A., Steller H., Apoptosis, Stem Cells, and Tissue Regeneration Science Signaling, 2010, 3, 145
[3] Fulda S. and Pervaiz S., Apoptosis signaling in cancer stem cells. International Journal of Biochemistry and Cell Biology, 2010, 42, 31

We thank Dr. Brent Graham for helpful scientific discussions

© Astex Pharmaceuticals Inc.

Poster presented at AACR-NCI-EORTC Molecular Targets and Cancer Therapeutics Conference, October 19 – 23, 2013 • Boston (USA)

Poster can be downloaded from www.astx.com