abstract
A256

Epigenetic priming with novel hypomethylating agent SGI-110 improved antitumor activity of CTLA-4 blockade in a syngenic mouse model

Alessia Covre[1], Carolina Fazio[1], Hugues JMG Nicolay[1], Giulia Parisi[1], Pietro Taverna[2], Mohammad Azab[2], Sandra Coral[1] and Michele Maio[1]

[1]*Division of Medical Oncology and Immunotherapy, Department of Oncology, University Hospital of Siena, Istituto Toscano Tumori, Siena, Italy;* [2]*Astex Pharmaceuticals Inc. Dublin, California USA*

Abstract

Background. The second generation DNA hypomethylating agent SGI-110, a dinucleotide of decitabine (DAC) and deoxyguanosine formulated as a small volume SQ injection allowing extended DAC exposure compared to DAC i.v., has shown promising anti-cancer clinical activity. Our recent pre-clinical in vitro and in vivo data provides evidence that SGI-110 bears immunomodulating activities which improve the recognition of neoplastic cells by cytotoxic T lymphocytes. In addition, the hypomethylating activity of SGI-110 on cancer testis antigens (CTA)-specific promoters and the resulting induction/up-regulation of CTA expression have been extensively characterized in PBMCs from MDS or AML patients enrolled in a randomized Phase 1-2 First in Human PK/PD-guided, dose-escalation study. These well-defined immunomodulatory properties of SGI-110 prompt its potential use for the development of new immunotherapeutic approaches. Accordingly, in this study, we explored whether SGI-110 might improve the clinical efficacy of an anti-CTLA-4 monoclonal antibody (mAb).

Materials and Methods. Balb/c mice (6/group) were injected SQ in the flank region with murine mammary carcinoma cells TS/A (2×10^5). Mice bearing palpable tumor grafts (diameter ≥ 0.2 cm) were treated with 3mg/kg of reconstituted SGI-110 (daily x 5) from day 1, alone or in combination with 100μg/hamster anti-murine CTLA-4 mAb simultaneously with (days 2, 5 and 8) or sequentially to (days 8, 11 and 14) SGI-110. Control mice were injected with diluent for reconstitution. The effectiveness and tolerability of treatment was evaluated by tumor volume and body weight measurements, respectively.

Results. The best antitumor effect was achieved in mice treated with SGI-110 followed by anti-CTLA-4 mAb; in detail, a tumor mass significantly ($p<0.05$) smaller than that of control mice was observed, indicating, at day 26, a tumor growth inhibition of 84.4%. Moreover, a significant, but lower, reduction in tumor mass occurred in SGI-110-treated mice as compared to control mice, with a tumor growth inhibition of 62.9% at days 26. No difference was observed in tumor growth inhibition in mice treated with SGI-110 combined with simultaneous anti-CTLA-4 mAb treatment as compared to mice treated with SGI-110 alone. Noteworthy, no loss in body weight was observed in all mice investigated, demonstrating a good tolerability of therapeutic regimens tested.

Conclusion. Altogether, these data provide evidence for a scientific rationale to develop novel and more effective epigenetic-immunotherapeutic strategies for cancer patients. Accordingly, an exploratory phase I clinical trial to evaluate safety and immunobiologic activities of epigenetic priming with SGI-110 followed by CTLA-4 blockade is being planned.

Results

RT-PCR analysis of CTA expression in murine mammary carcinoma cell line TS/A treated with SGI-110

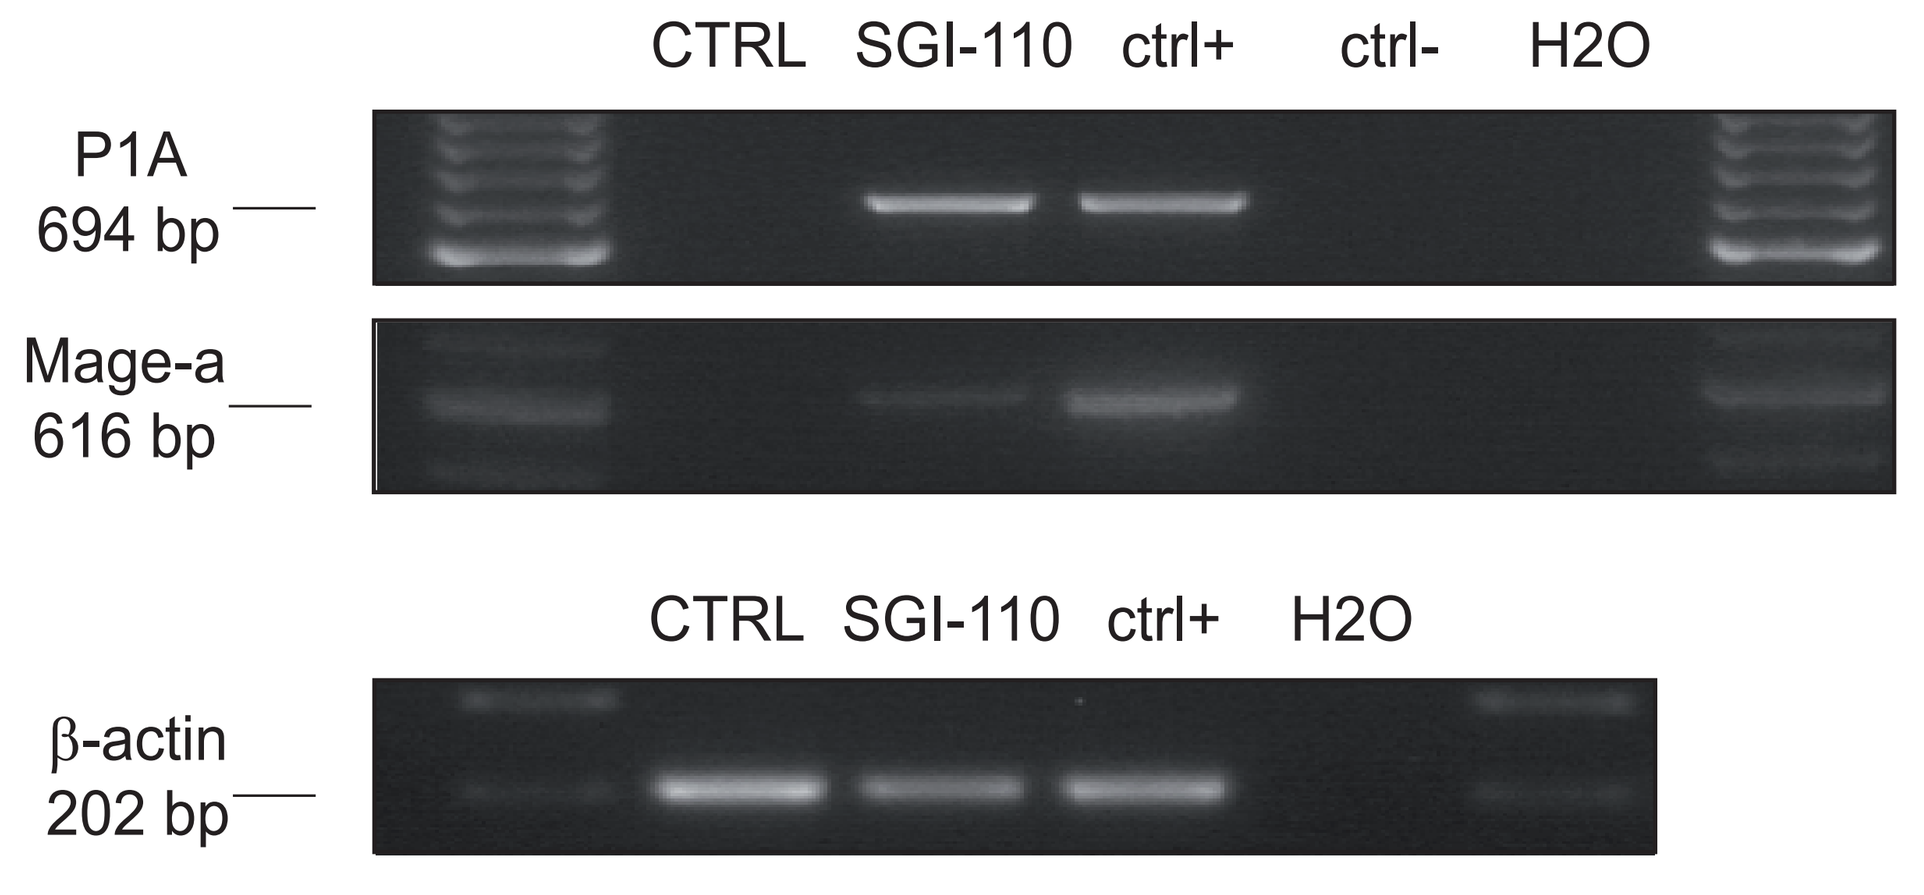
CTRL SGI-110 ctrl+ ctrl- H2O
P1A
694 bp
Mage-a
616 bp
CTRL SGI-110 ctrl+ H2O
β-actin
202 bp

Total RNA was extracted from the murine mammary carcinoma cell line TS/A, either untreated (CTRL) or treated with SGI-110 (1μM) every 12 hours for 2 days (4 pulses). RT-PCR analysis was performed using P1A-, or Mage-a-specific primer pairs. PCR products were then separated on a 2% agarose gel and visualized by ethidium bromide staining. RNA integrity and cDNA quality were confirmed by amplification of the house-keeping gene mouse β-actin.

Quantitative real-time RT-PCR analysis of P1A expression in murine mammary carcinoma cell line TS/A treated with SGI-110

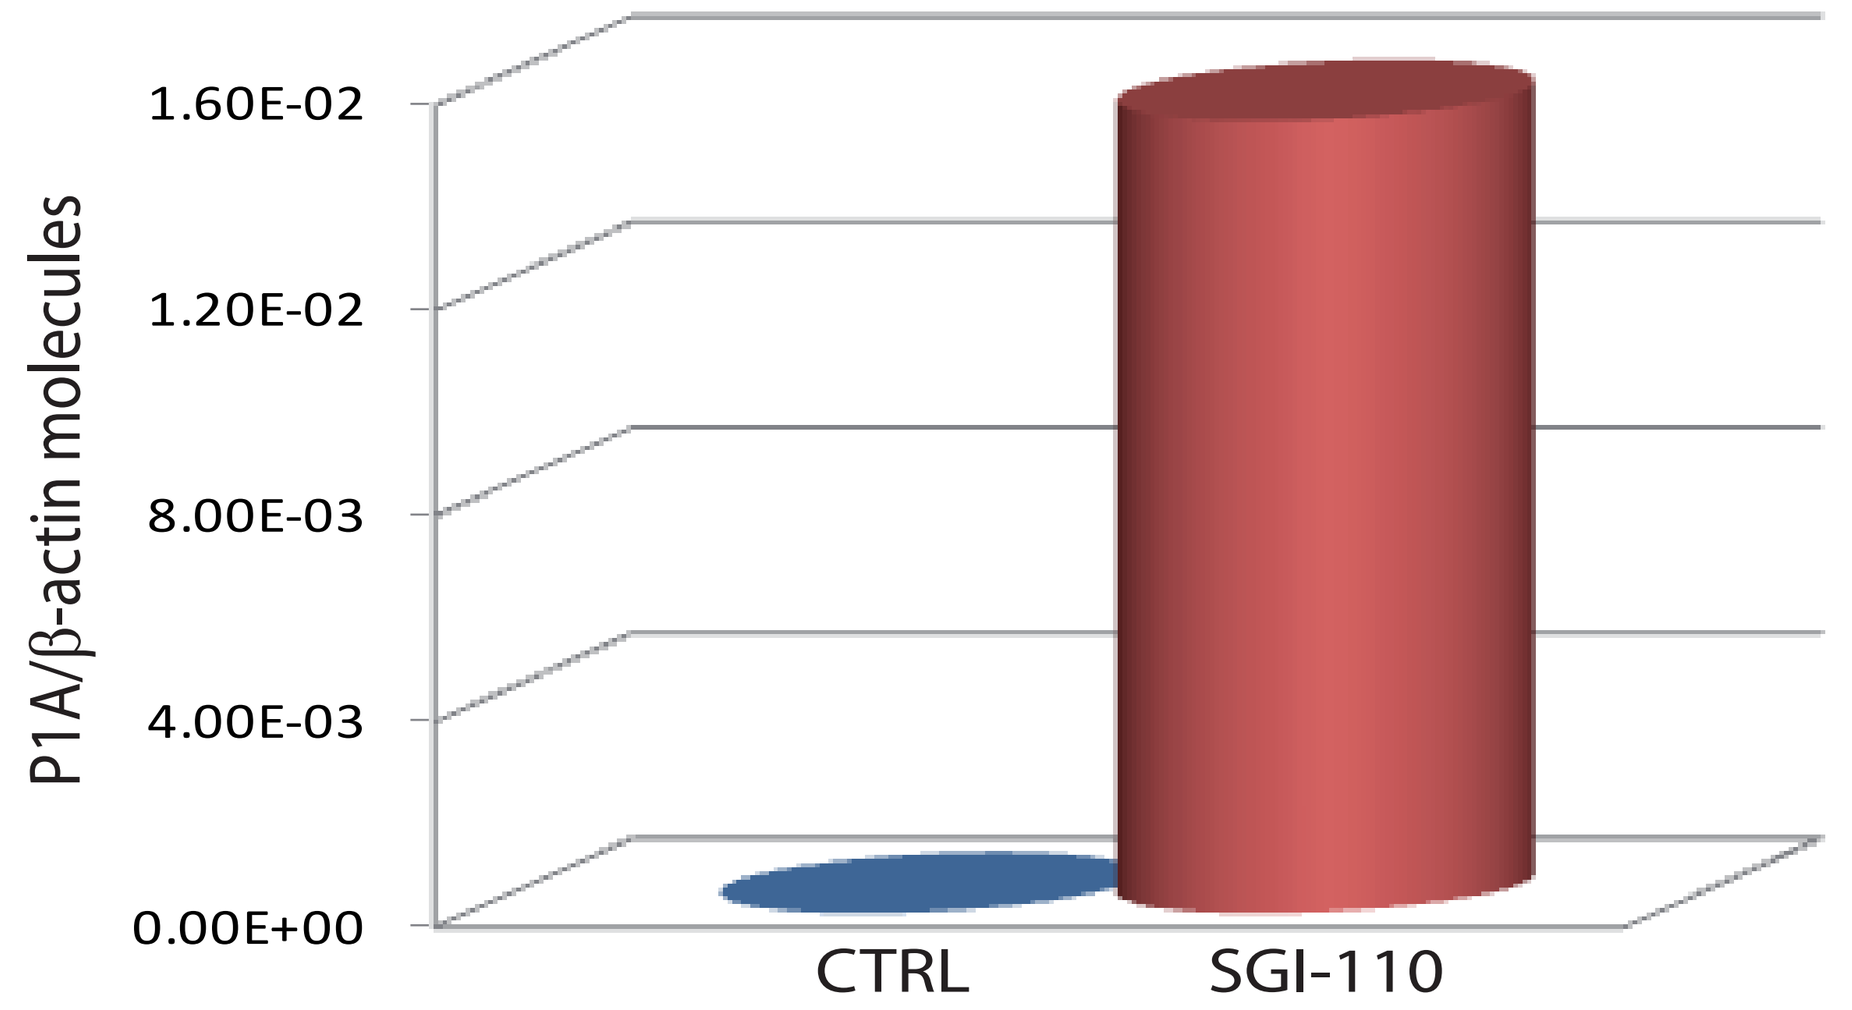
P1A/β-actin molecules
1.60E-02
1.20E-02
8.00E-03
4.00E-03
0.00E+00
CTRL
SGI-110

Total RNA was extracted from the murine mammary carcinoma cell line TS/A, either untreated (CTRL) or treated with SGI-110 (1μM) every 12 hours for 2 days (4 pulses). Retrotranscribed RNA was subjected to SYBR Green quantitative real-time RT-PCR analysis using P1A- and β-actin-specific primers. Data have been reported as P1A molecules/β-actin molecules. Columns, mean values of P1A molecules/β-actin molecules

Table 1. qdx5 days schedules of in vivo administration of SGI-110 and anti-CTLA-4 mAb

Group	Treatment[a]	Dose	Route[b]	Schedule
CTRL	Vehicle		SQ	Days 1-5 (5days)
SGI-110	SGI-110	3 mg/kg day	SQ	Days 1-5 (5days)
CTLA-4 simul[c]	Anti-CTLA-4 mAb	100 μg day	IP	Days 2, 5, 8
CTLA-4 seq[d]	Anti-CTLA-4 mAb	100 μg day	IP	Days 8, 11, 14
SGI + CTLA-4 simul	SGI-110 + Anti-CTLA-4 mAb	3 mg/kg day 100 μg day	SQ IP	Days 1-5 (5days) Days 2, 5, 8 (simultaneously)
SGI + CTLA-4 seq	SGI-110 + Anti-CTLA-4 mAb	3 mg/kg day 100 μg day	SQ IP	Days 1-5 (5days) Days 8, 11, 14 (sequentially)

[a] Mice were SQ injected with 0.1 ml of diluent for reconstitution (Vehicle) or reconstituted SGI-110 (SGI-110);
[b] SQ, subcutaneous; IP, intraperitoneum;
[c] simul, simultaneously;
[d] seq, sequentially;

Fig.1 Effectiveness of qdx5 days schedule of SGI-110 in combination with immunostimulatory anti-CTLA-4 mAb, in mouse cancer

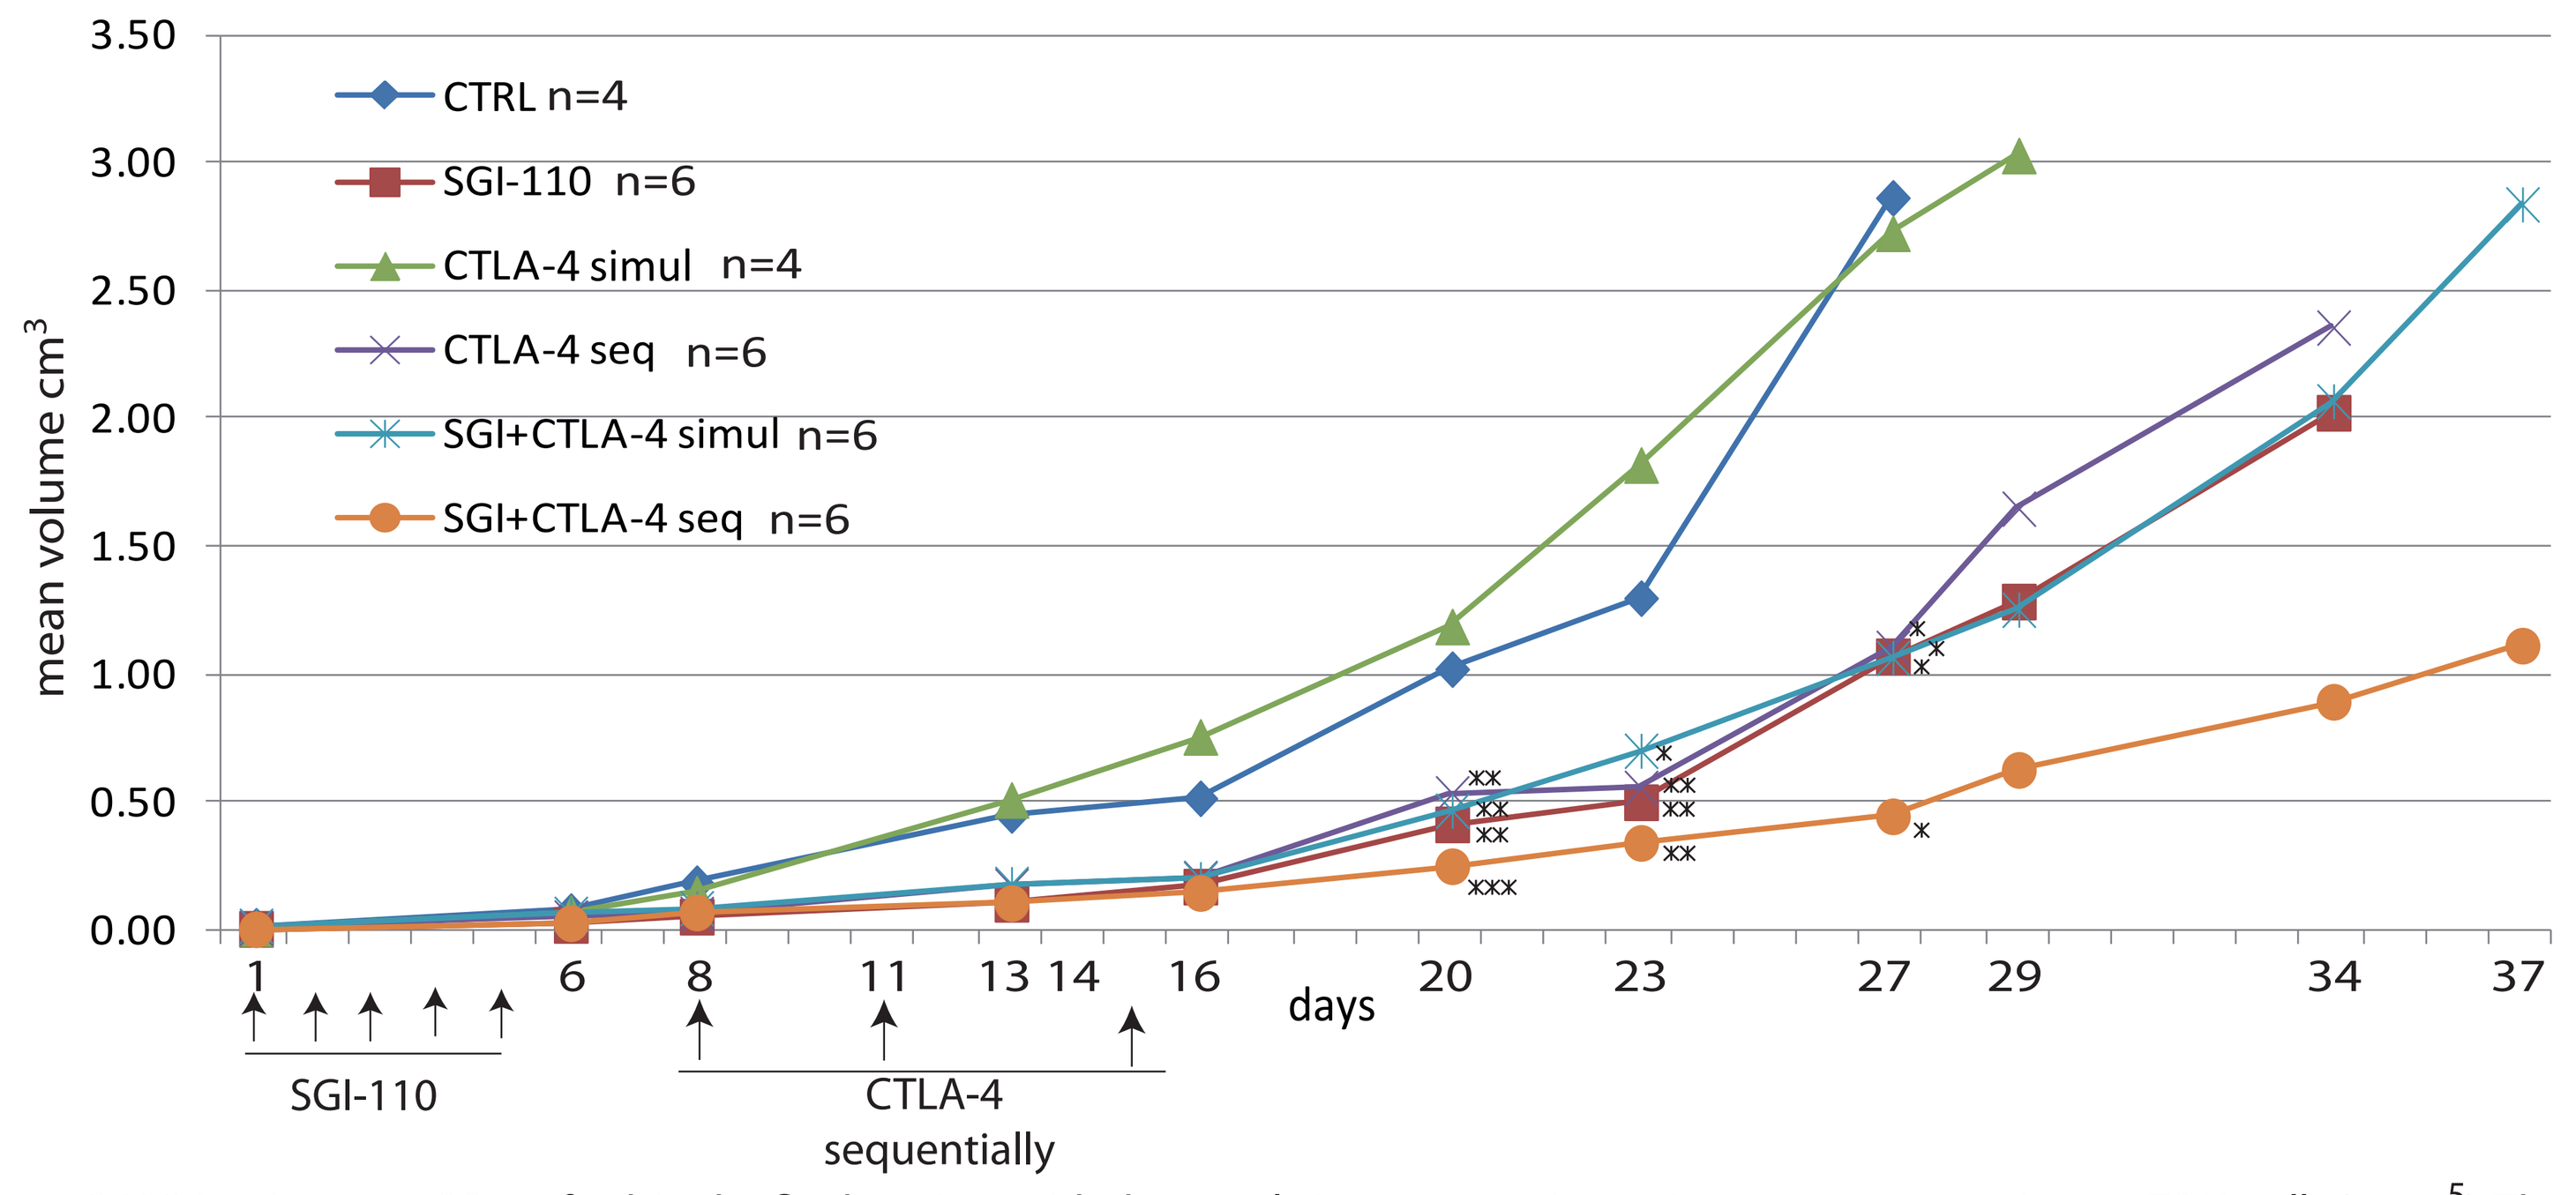
mean volume cm³
CTRL n=4
SGI-110 n=6
CTLA-4 simul n=4
CTLA-4 seq n=6
SGI+CTLA-4 simul n=6
SGI+CTLA-4 seq n=6
days
SGI-110
CTLA-4
sequentially

BALB/c mice were SQ grafted, in the flank region, with the poorly immunogenic murine mammary carcinoma TS/A cells (2×10^5). Then, groups of mice were injected with diluent solution for control, SGI-110, anti-CTLA-4 mAb, or with combined administration of SGI-110 and anti-CTLA-4 mAb, according to the reported qdx5 schedules of treatment (see Table 1). To evaluate the effectiveness of therapies, tumor volumes (TV) from mice were measured periodically, all along the treatment, by using a caliper and calculated as follows: TV=LD2/2 (in which L is the longest diameter and D the shortest one). Tumor mean values for each group are reported. Vertical arrows indicate days of different treatments. *, p≤0,05 ; **, p≤0,01; ***, p≤0,001 vs. tumor volumes of control group.

Fig.2 Tolerability of 5 days schedule of SGI-110 in combination with immunostimulatory anti-CTLA-4 mAb, in mouse cancer

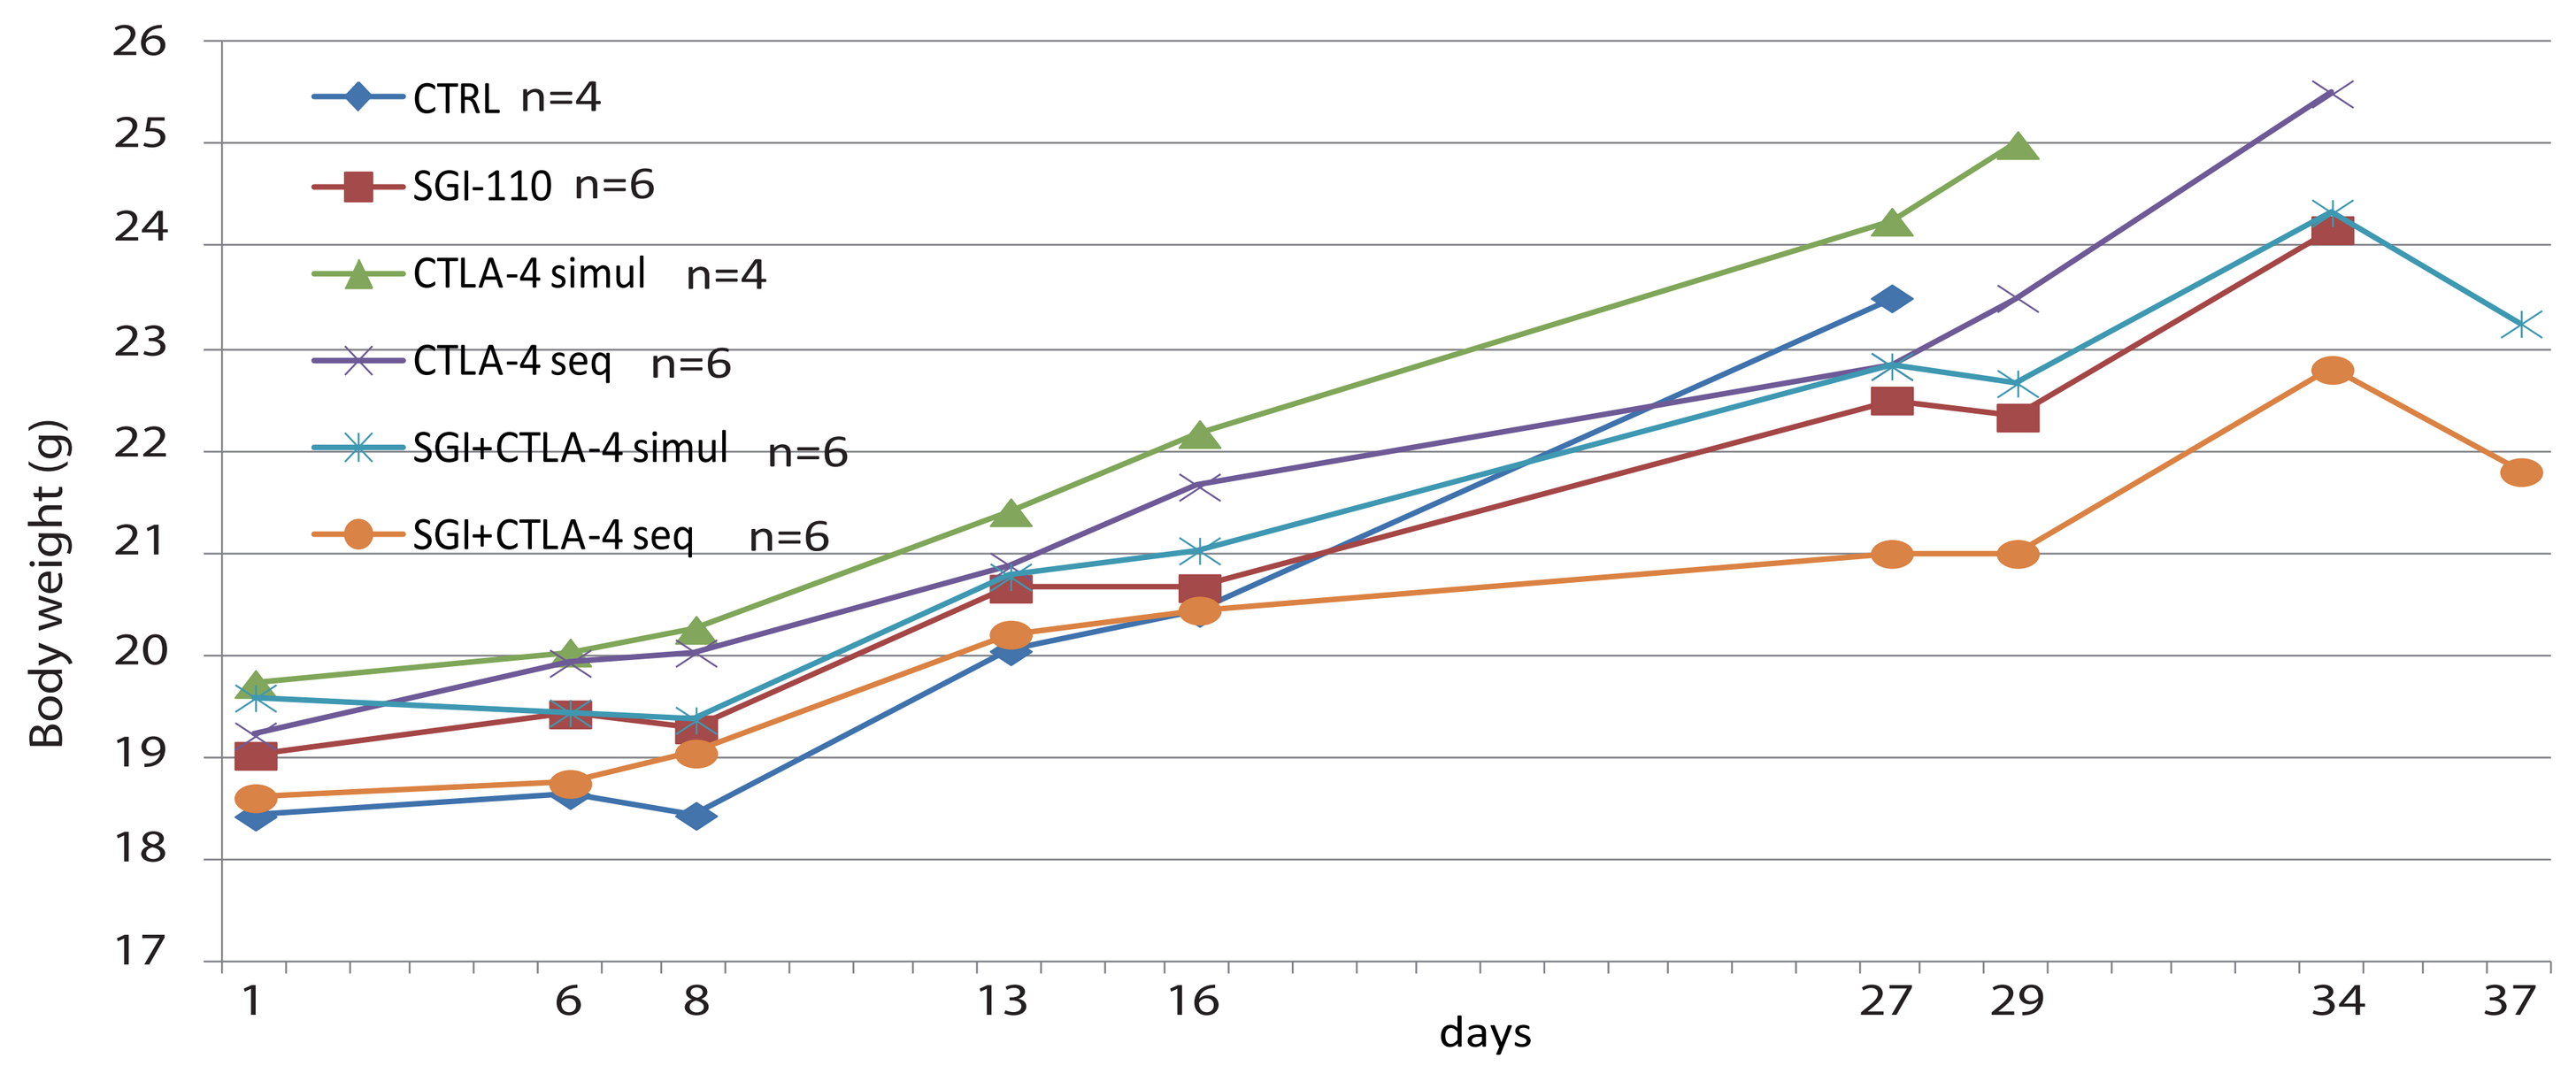
Body weight (g)
CTRL n=4
SGI-110 n=6
CTLA-4 simul n=4
CTLA-4 seq n=6
SGI+CTLA-4 simul n=6
SGI+CTLA-4 seq n=6
days

BALB/c mice were SQ grafted, in the flank region, with the poorly immunogenic murine mammary carcinoma TS/A cells (2×10^5). Then, groups of mice were injected with diluent solution for control, SGI-110, anti-CTLA-4 mAb, or with combined administration of SGI-110 and anti-CTLA-4 mAb, according to the reported qdx5 schedules of treatment (see Table 1). Tolerability of treatments was evaluated by measurements of body weight in mice, at different time points of the investigated treatment schedules.

Table 2. qdx5 days x2 schedules of in vivo administration of SGI-110 and anti-CTLA-4 mAb

Group	Treatment[a]	Dose	Route[b]	Schedule
CTRL	Vehicle		SQ	Days 1-5, 21-25
SGI-110 x2	SGI-110	3 mg/kg day	SQ	Days 1-5, 21-25
CTLA-4 seq x2[c]	Anti-CTLA-4 mAb	100 μg day	IP	Days 8, 11, 14
SGI + CTLA-4 seq x2	SGI-110 + Anti-CTLA-4 mAb	3 mg/kg day 100 μg day	SQ IP	Days 1-5, 21-25 Days 8, 11, 14; 28, 31, 33 (sequentially)

[a] Mice were SQ injected with 0.1 ml of diluent for reconstitution (Vehicle) or reconstituted SGI-110 (SGI-110);
[b] SQ, subcutaneous; IP, intraperitoneum;
[c] seq, sequentially;

Fig.3 Effectiveness of repeated administration of SGI-110 in combination with immunostimulatory anti-CTLA-4 mAb, in mouse cancer

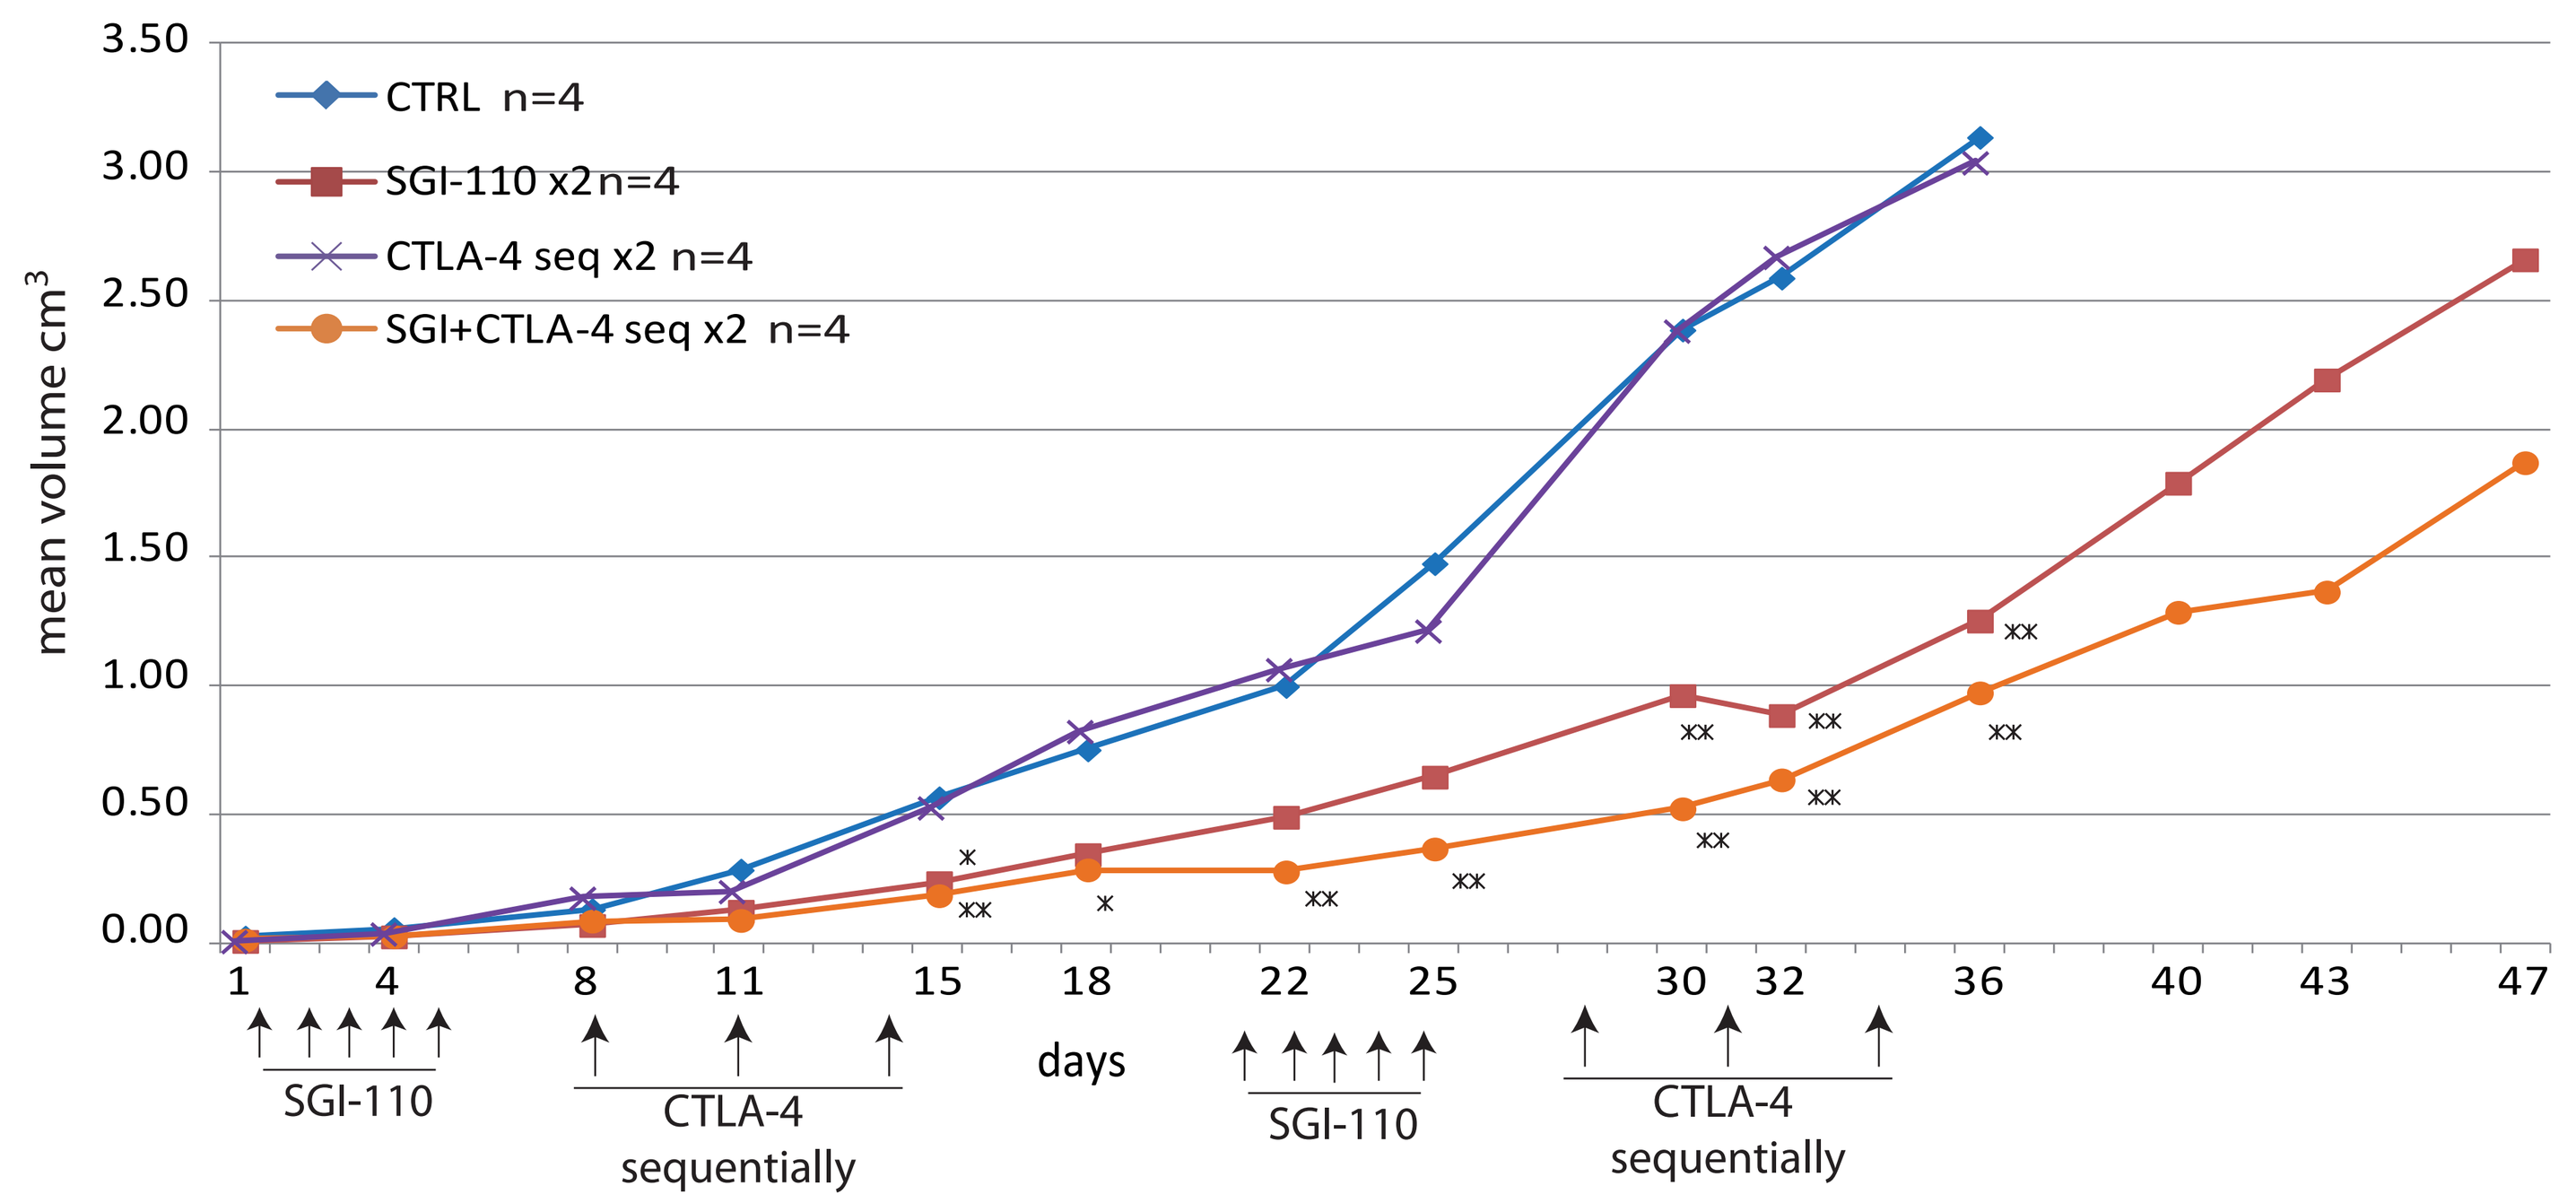
mean volume cm³
CTRL n=4
SGI-110 x2n=4
CTLA-4 seq x2 n=4
SGI+CTLA-4 seq x2 n=4
days
SGI-110
CTLA-4
sequentially

BALB/c mice were SQ grafted, in the flank region, with the poorly immunogenic murine mammary carcinoma TS/A cells (2×10^5). Then, groups of mice were injected with diluent solution for control, SGI-110, anti-CTLA-4 mAb, or with combined administration of SGI-110 and anti-CTLA-4 mAb, according to the reported qdx5 schedules of treatment (see Table 2), repeated for a 2nd cycle of treatment at day 21. To evaluate the effectiveness of therapies, tumor volumes (TV) from mice were measured periodically, all along the treatment, by using a caliper and calculated as follows: TV=LD2/2 (in which L is the longest diameter and D the shortest one). Tumor mean values for each group are reported. Vertical arrows indicate days of different treatments. *, p≤0,05 ; **, p≤0,01; vs. tumor volumes of control group.

Fig.4 Tolerability of repeated administration of SGI-110 in combination with immunostimulatory anti-CTLA-4 mAb, in mouse cancer

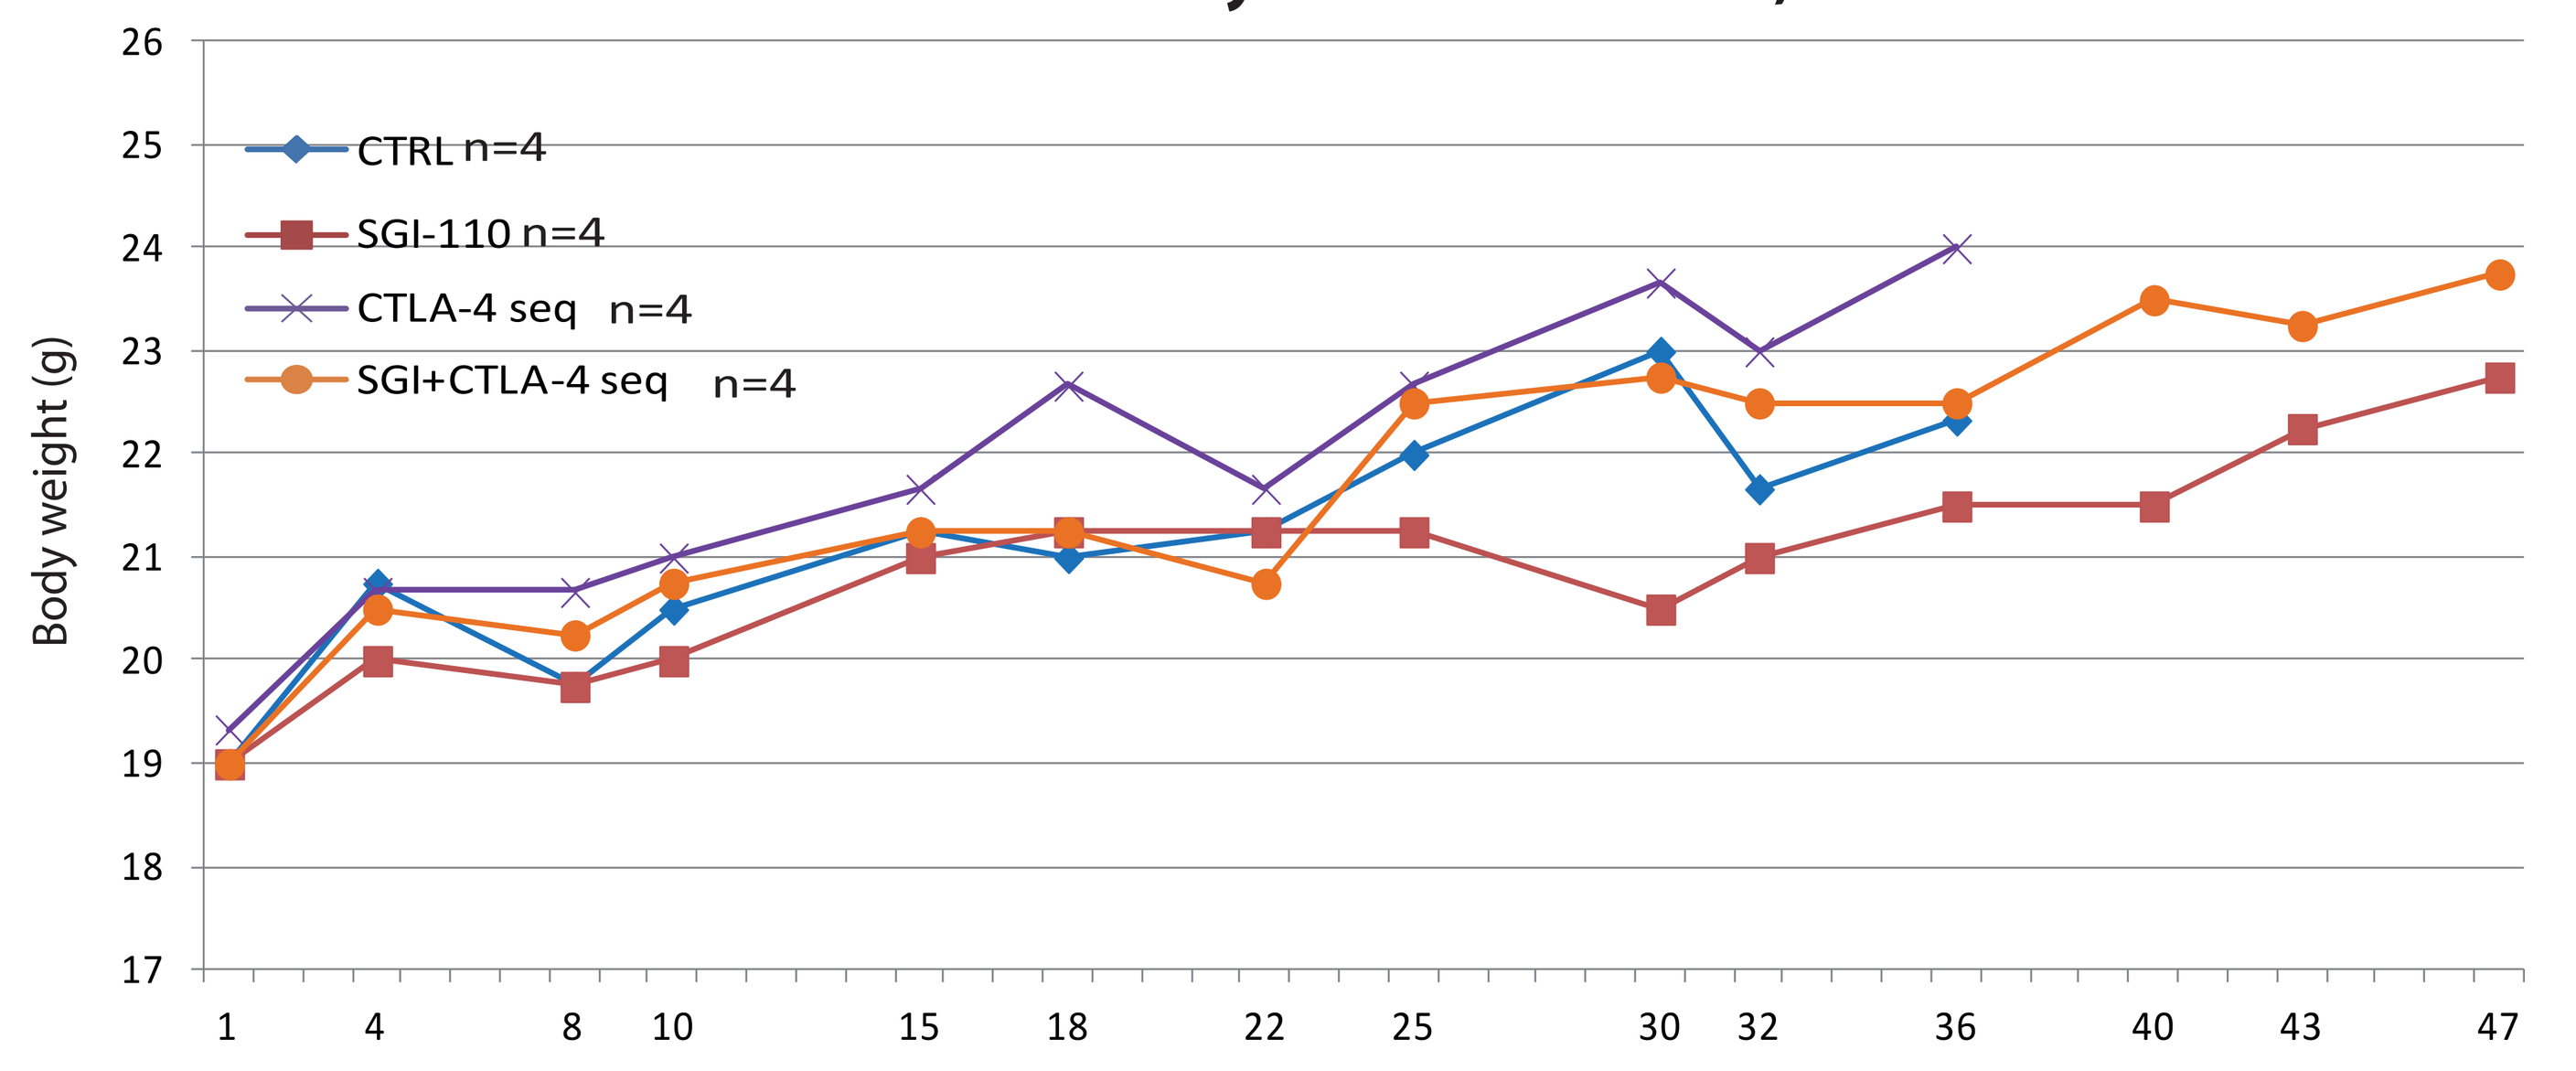
Body weight (g)
CTRL n=4
SGI-110 n=4
CTLA-4 seq n=4
SGI+CTLA-4 seq n=4

BALB/c mice were SQ grafted, in the flank region, with the poorly immunogenic murine mammary carcinoma TS/A cells (2×10^5). Then, groups of mice were injected with diluent solution for control, SGI-110, anti-CTLA-4 mAb, or with combined administration of SGI-110 and anti-CTLA-4 mAb, according to the reported qdx5 schedules of treatment (see Table 2), repeated for a 2nd cycle of treatment at day 21. Tolerability of treatments was evaluated by measurements of body weight in mice, at different time points of the investigated treatment schedules

Fig.5 Time-dependency of CTA expression induced in solid malignancies by SGI-110

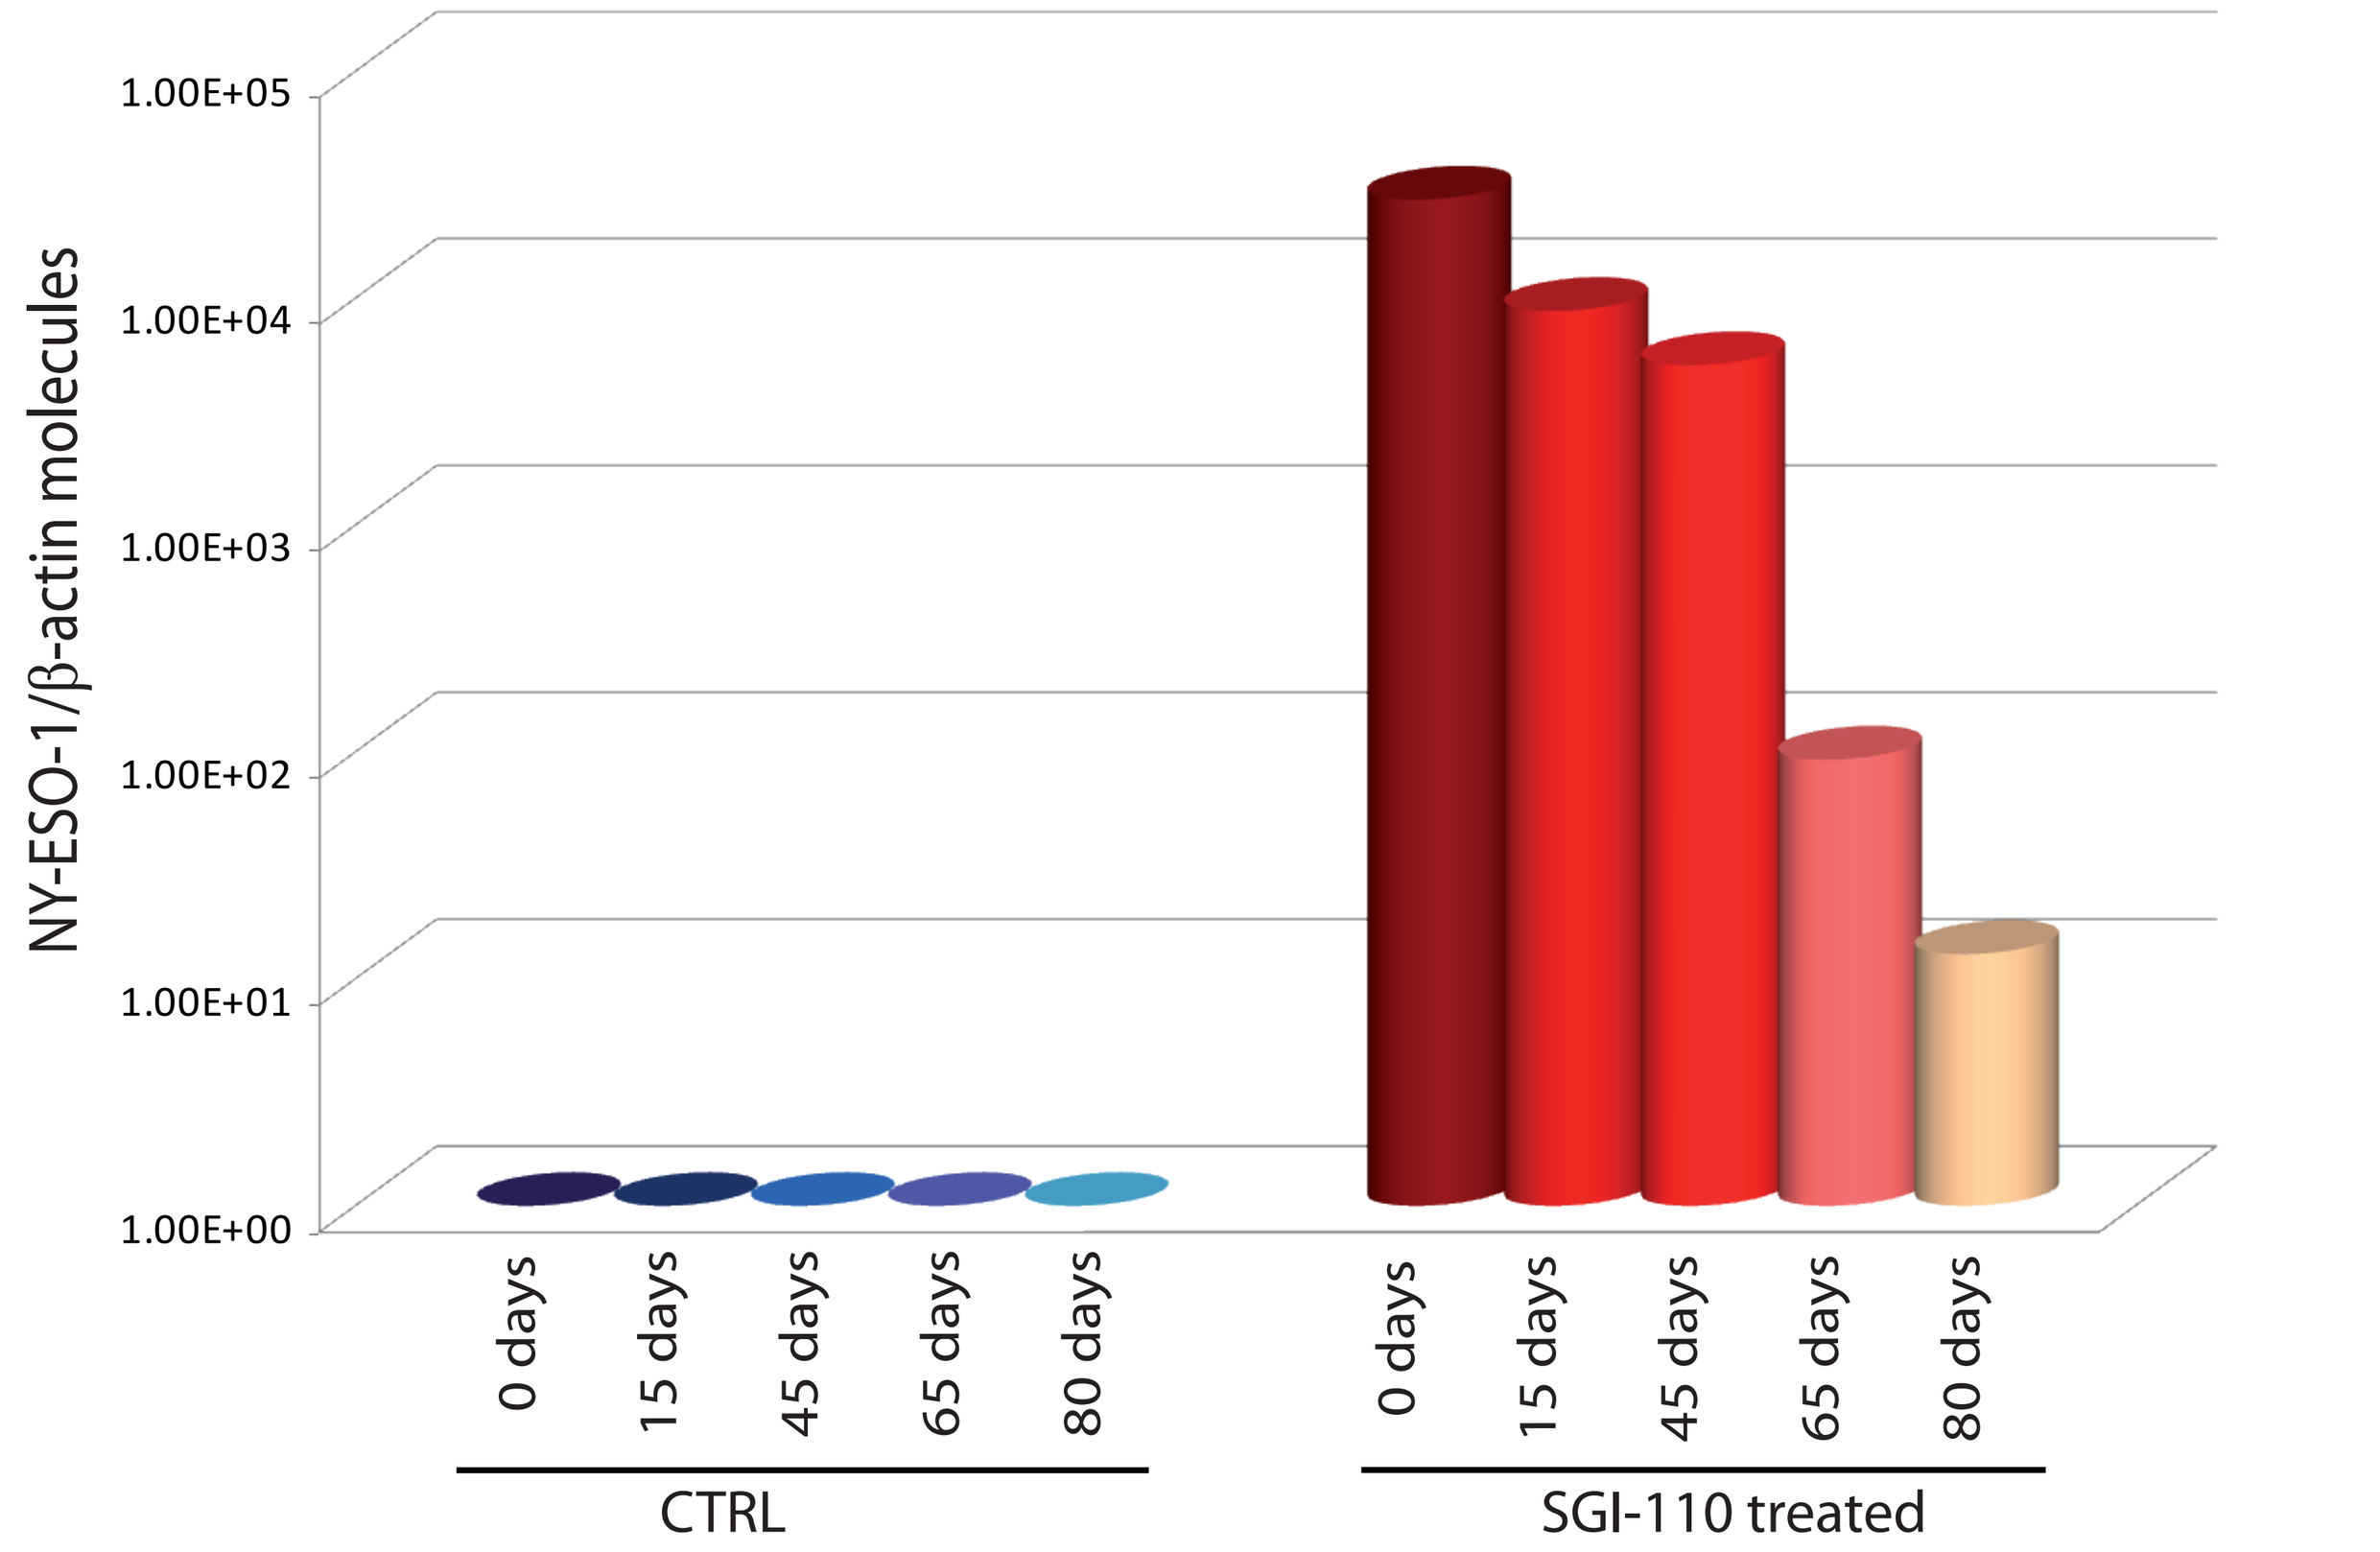
NY-ESO-1/β-actin molecules
1.00E+05
1.00E+04
1.00E+03
1.00E+02
1.00E+01
1.00E+00
0 days
15 days
45 days
65 days
80 days
CTRL
SGI-110 treated

Total RNA was extracted from Mel 275 human melanoma cells untreated (blue) or treated (red) with 1μM SGI-110 every 12 hours for 2 days (4 pulses) and in vitro cultured for the indicated days after the end of treatment. SYBR Green quantitative RT-PCR reactions were performed on retrotranscribed total RNA, utilizing NY-ESO-1- and β-actin-specific primers. CTA expression was normalized to the expression of the housekeeping gene β-actin. Values are reported as NY-ESO-1 molecules/β-actin molecules multiplied by 10^6 and visualized on logarithmic scale.
(Coral S et al., Cancer Immunol Immunother. 2013 Mar;62(3):605-14)

Conclusions

-SGI-110 was able to modify CTA-profile of poorly immunogenic cancer cells.

-The combination of SGI-110 plus anti-CTLA-4 mAb resulted into the best antitumor effect.

-The most effective tumor growth control at tolerated doses and schedules was achieved with the qdx5 schedule of SGI-110 SQ administration followed by anti-CTLA-4 mAb.

- The anti-tumor effect of SGI-110/anti-CTLA4 mAb was evident also after a second cycle of treatment suggesting the maintenance of the SGI-110-mediated immunological effects.

These findings demonstrate that SGI-110 is an attractive therapeutic agent to improve the anti-tumor activity of anti-CTLA-4 mAb and to expand the partial therapeutic efficacy of immunostimulatory mAb to poorly immunogenic tumors.

SGI-110 is currently being investigated in 3 clinical trials for the treatment of patients with myelodysplastic syndromes and acute myeloid leukemia (ClinicalTrials.gov identifier: NCT01261312), ovarian cancer in combination with carboplatin (ClinicalTrials.gov identifier: NCT01696032) and advanced hepatocellular carcinoma (ClinicalTrials.gov identifier: NCT01752933).

Altogether these evidences provide a strong scientific rationale to develop novel and more effective epigenetic-immunotherapeutic combination strategies for cancer patients. An exploratory phase I clinical trial to evaluate safety and immunobiologic activities of SGI-110 epigenetic priming followed by CTLA-4 blockade is being planned.

Study supported by Astex Pharmaceuticals

AACR-NCI-EORTC Molecular targets and cancer therapeutics, October 18-23, 2013, Boston MA

"poster can be downloaded from www.astx.com"