1231PD

A Study of Hsp90 Inhibitor AT13387 Alone and in Combination with Crizotinib (CZT) in the Treatment of Non-Small Cell Lung Cancer (NSCLC)

B. Besse[1], E. Bertino[2], N.A. Pennell[3], A. Wozniak[4], D. Mahadevan[5], A. Spira[6], A. Oganesian[7], L. Manlapaz-Espiritu[7], H. Keer[7], J.-C. Soria[1], D.R. Camidge[8]

[1]Villejuif/FR, [2]Columbus, OH/US, [3]Cleveland, OH/US, [4]Detroit, MI/US, [5]Memphis, TN/US, [6]Fairfax, VA/US, [7]Dublin, CA/US, [8]Denver/US

BACKGROUND

- Hsp90 is required for proper ALK function
- AT13387 is a second generation Hsp90 inhibitor
- AT13387 in ALK-driven pre-clinical models:
 - Displays potent antitumor activity
 - Delays the onset of resistance[1]
- In the clinic, AT13387[2]
 - Has a safety profile consistent with the class
 - (diarrhea, mild transient visual changes)
 - PK & PD results support weekly dosing
 - Has antitumor activity (PR) at a dose of 220 mg/m^2 (D1,8,15 of 28 Day cycle)
- Crizotinib (CZT) has demonstrated clinical activity in ALK-pos NSCLC[3]
- AT13387-05 is a 3-part, Phase 1-2, randomized study

Figure 1. Depiction of HSP90 action on client proteins

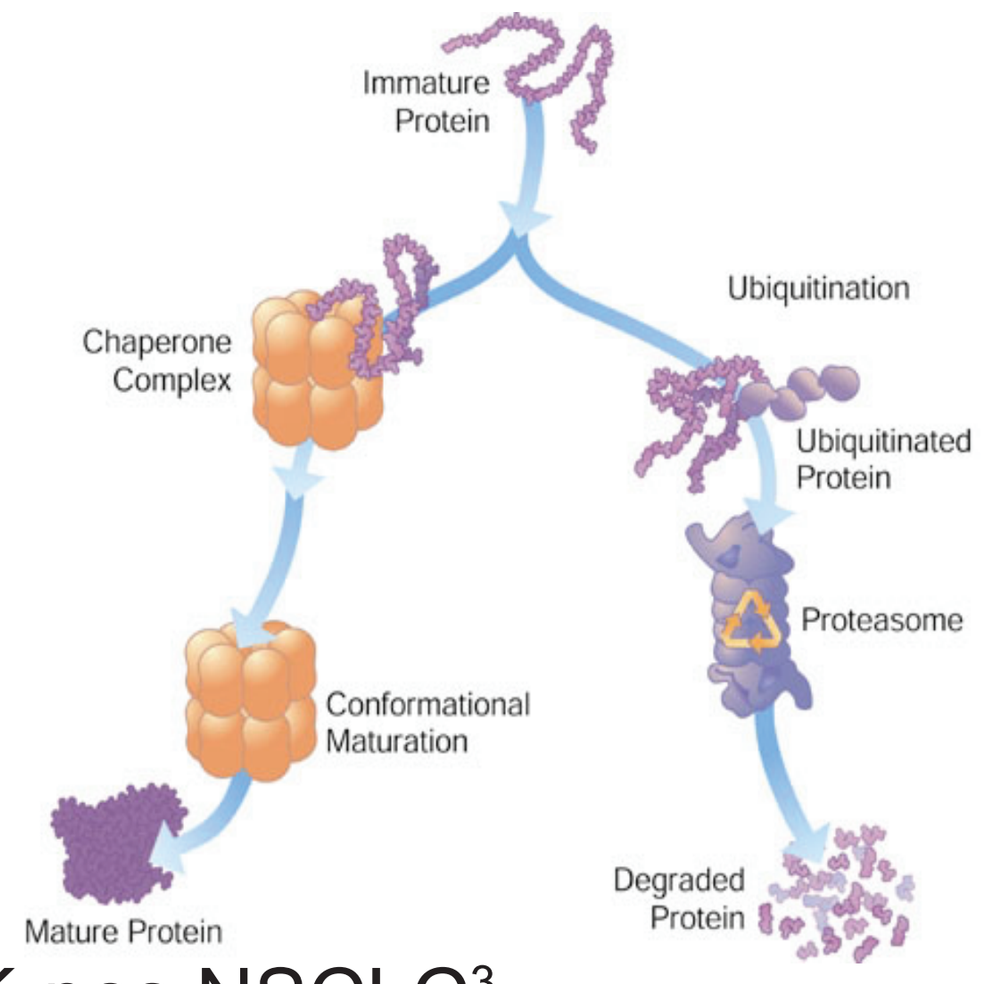
Immature Protein
Chaperone Complex
Ubiquitination
Ubiquitinated Protein
Proteasome
Conformational Maturation
Mature Protein
Degraded Protein

STUDY DESIGN

Figure 2. Study Schema. Results are presented from Part A (Phase 1) studying the safety of combination AT13387 + CZT

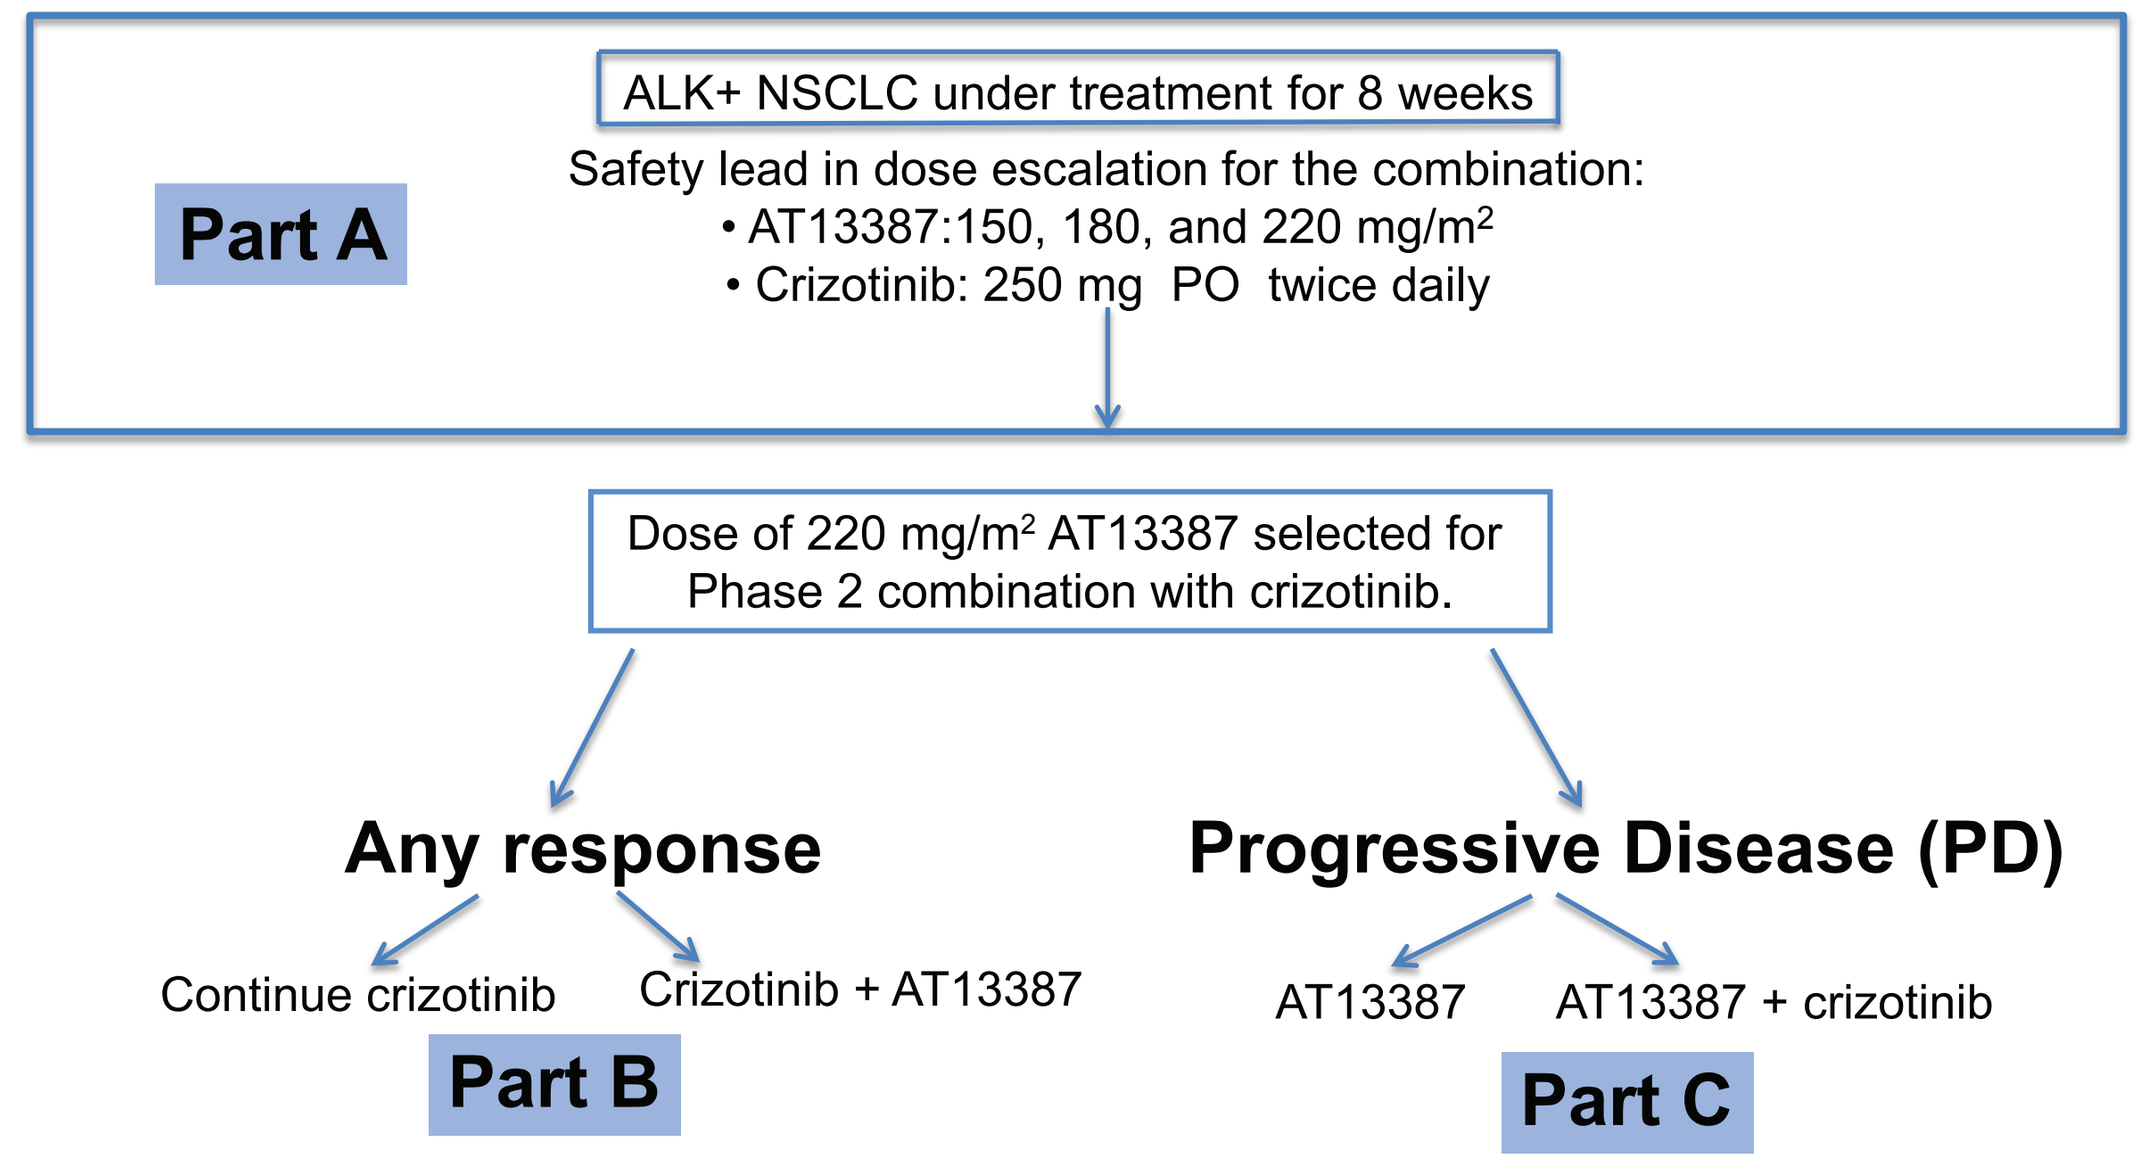
Part A
ALK+ NSCLC under treatment for 8 weeks
Safety lead in dose escalation for the combination:
• AT13387:150, 180, and 220 mg/m2
• Crizotinib: 250 mg PO twice daily
Dose of 220 mg/m2 AT13387 selected for Phase 2 combination with crizotinib.
Any response
Continue crizotinib
Crizotinib + AT13387
Part B
Progressive Disease (PD)
AT13387
AT13387 + crizotinib
Part C

Study Objectives:

- **Primary Objective (Part A):**
 - Safety and tolerability of AT13387 with CZT (250 mg BID)
- **Secondary Objectives (Part A):**
 - Pharmacokinetics in combination with CZT
 - Pharmacodynamics in combination with CZT
 - Antitumor activity of AT13387 in combination with CZT

Patient Population

- **Major inclusion criteria:**
 - Patients with NSCLC (ALK rearranged or other change potentially sensitive to CZT, eg ROS)
 - Previously had tolerated 8 weeks of CZT treatment
 - Adequate organ (cardiac, renal, hepatic) function & Performance Status (ECOG 0-1)
 - Controlled brain metastases allowed
- **Major exclusion criteria:**
 - Prior anticancer treatment with Hsp90 inhibitor
 - Known symptomatic CNS metastases
 - ≥ grade 2 bilirubin or transaminases or visual disturbances while on CZT

RESULTS

Study Methodology

- **Dosing Regimen:**
 - AT13387 is administered intravenously (IV) Days 1,8, and 15 of a 28-Day dosing cycle. AT13387 dosing is in combination with CZT 250 PO BID at escalating doses of 150, 180, and 220 mg/m2. Dose escalation is based on safety (occurrence of dose limiting toxicities[DLT])
- **Study Endpoints:**
 - Primary: The incidence of DLTs for AT13387 in combination with standard dose CZT
 - Secondary:
 - Incidence and severity of adverse events (AEs) at escalating doses of AT13387 in combination with standard dose CZT
 - Assessment of PK interaction between AT13387 + CZT
 - The ORR (CR + PR), PFS, and OS in subjects receiving AT13387 + CZT
 - Hsp70 induction and client protein knockdown in tumor biopsy
- **Study Assessments:**
 - Safety:
 - AEs assessed at D1,8,15,22 for cycle 1
 - Triplicate ECGs pre- and post-AT13387 infusion
 - Chemistry and Hematology prior to Days 1,8, and 15
 - PK: Blood samples at various timepoints Day 1 for AT13387 and CZT
 - Efficacy: Assessment of ORR and PFS by RECIST every 2 cycles

Demographics

Table 1. Patient Characteristics

Characteristic		Dosing Cohort			
		150 mg/m^2 (n=8)	180 mg/m^2 (n=9)	220 mg/m^2 (n=15)	Total (n=32)
Age (yrs) median (range)		52 (31-63)	59 (29-75)	62 (36-73)	59 (29-75)
Gender (M/F)		4/4 (50/50%)	3/6 (33/67%)	7/8 (47/53%)	14/18 (44/56%)
Histology (Adenocarcinoma/Other)		7/1	9/0	15/0	31/1
Prior ALK inhibitor					
	Crizotinib	8	9	15	32
	Ceritinib	0	0	1	1
	Alectinib	0	2	2	4

Pharmacokinetics

Pharmacokinetics of Crizotinib with and without (at screening) AT13387

Figure 3.

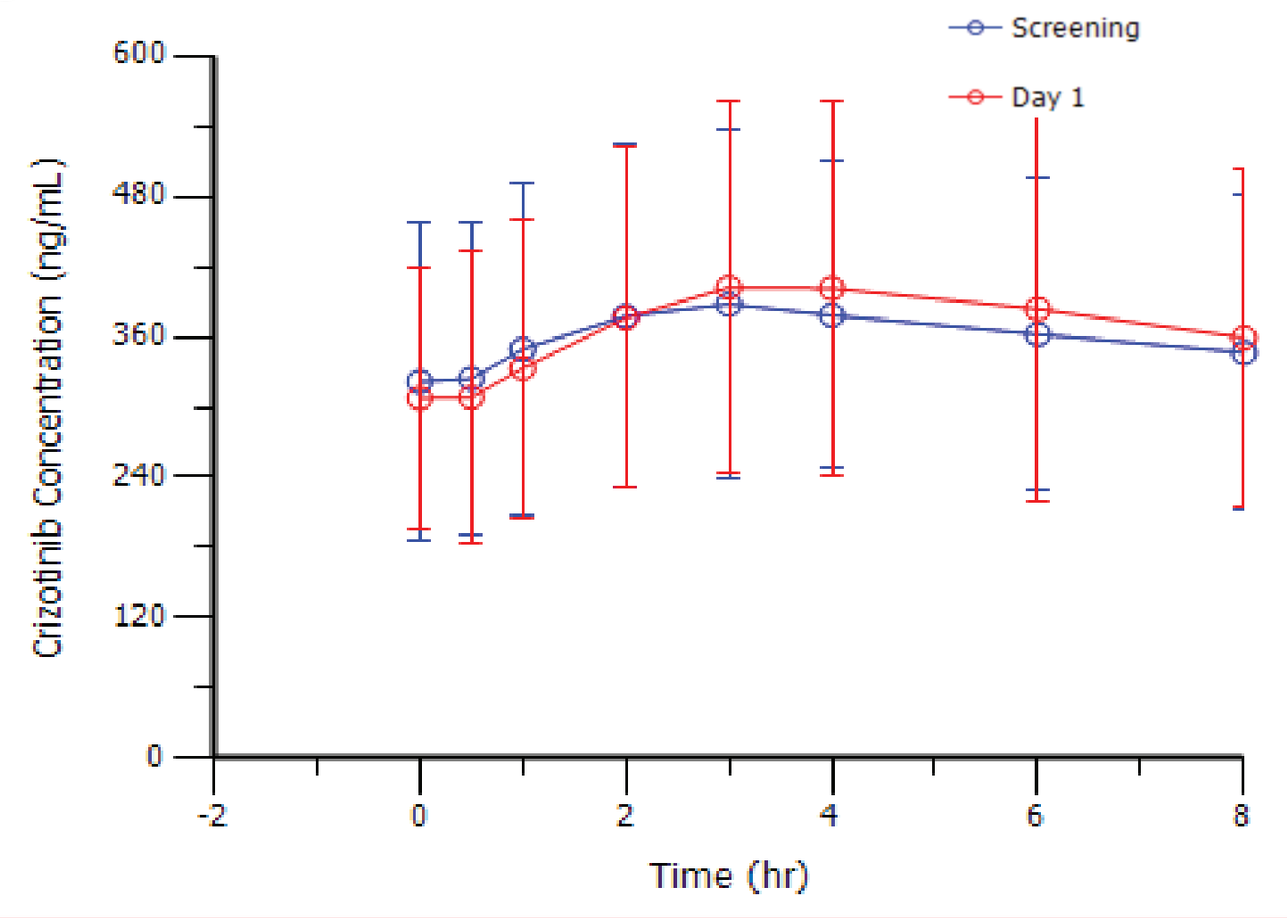
Screening
Day 1
Crizotinib Concentration (ng/mL)
600
480
360
240
120
0
-2
0
2
4
6
8
Time (hr)

Pharmacokinetics of AT13387 combined with Crizotinib

Figure 4.

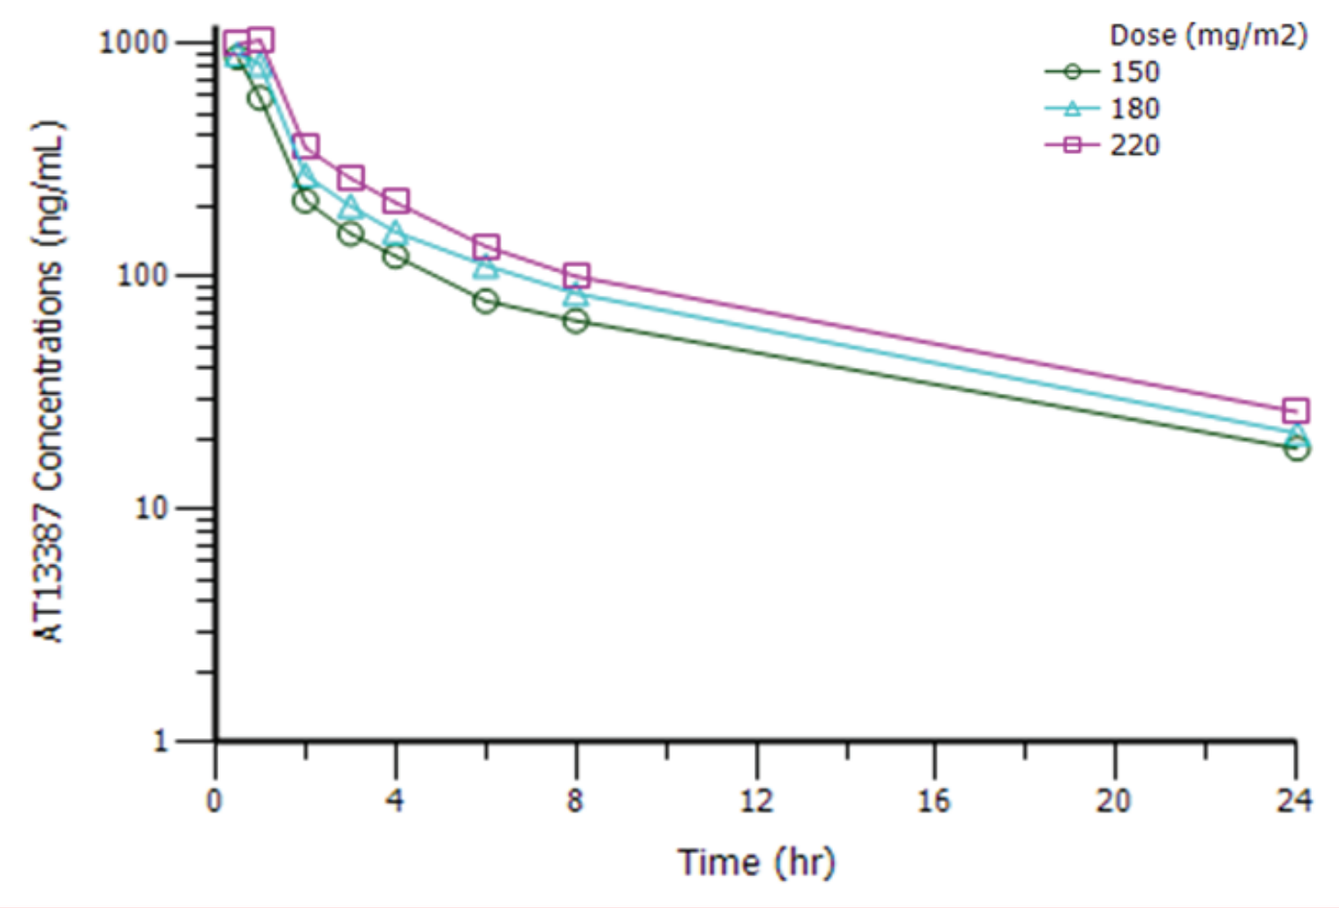
Dose (mg/m2)
150
180
220
AT13387 Concentrations (ng/mL)
1000
100
10
1
0
4
8
12
16
20
24
Time (hr)

Table 2. CZT PK Parameters

Day	$AUC_{0\text{-}8hr}$ (ng*hr/mL)	C_{max} (ng/mL)	T_{max} (hr)
Screening (n=31)	2905 (1056)	421 (143)	3.8 (2.1)
Day 1 (n=30)	2966 (1188)	427 (164)	4.0 (1.9)

Table 3. AT13387 PK Parameters

Dose (mg/m^2)	$AUC_{0\text{-}last}$ (ng*hr/mL)	C_{max} (ng/mL)	T_{max} (hr)	$T_{1/2}$ (hr)
150 (N=8)	2297 (368)	930.5 (335.8)	0.6 (0.2)	8.6 (1.3)
180 (N=8)	2923 (846)	1053 (740)	0.8 (0.3)	7.9 (1.3)
220 (N=14)	3585 (919)	1164.0 (742)	0.8 (0.2)	7.9 (1.5)

Pharmacokinetic Summary

- PK evaluation included sampling at screening and Cycle1-Day 1 for crizotinib; and Cycle1-Day1 for AT13387
- Crizotinib PK was not affected when co-administered with AT13387
- AT13387 exposures were similar to those observed in previous studies and did not appear to have been affected by crizotinib

Clinical Activity

Figure 5. Waterfall plot of subjects with measurable disease and at least 1 follow up scan

* Indicates subject still receiving treatment on study

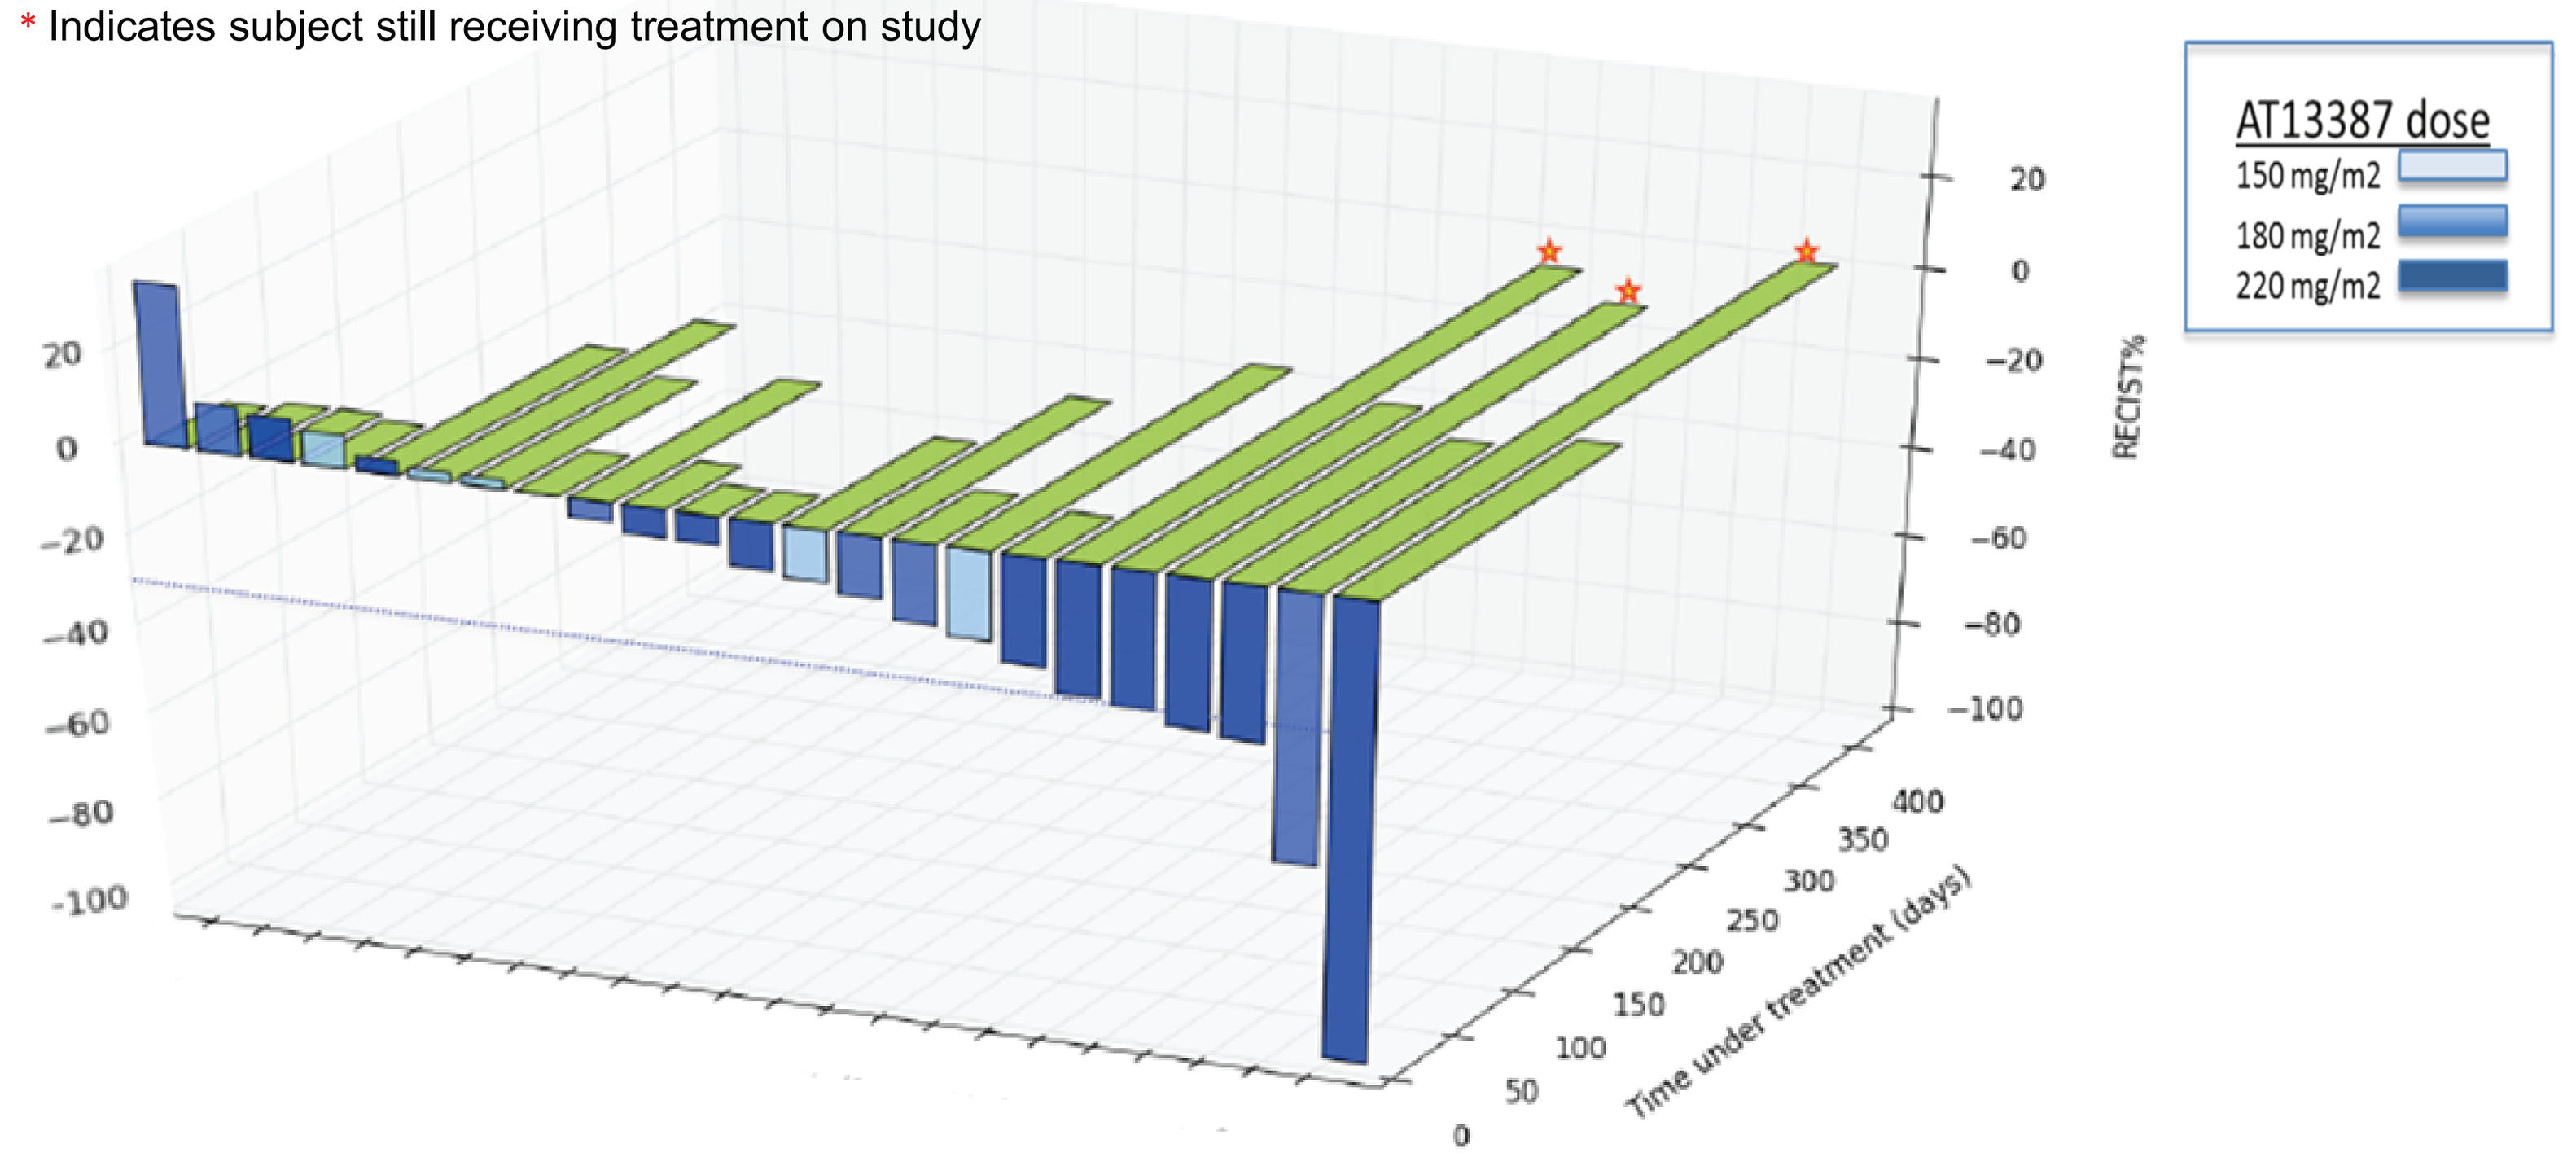
AT13387 dose
150 mg/m2
180 mg/m2
220 mg/m2
RECIST%
20
0
−20
−40
−60
−80
−100
Time under treatment (days)
0
50
100
150
200
250
300
350
400

Pt. dx'd in 2010, treated with BEV/carbo/paclitaxel, then PEM, then crizotinib, progressed on crizotinib, put on study, assigned to 180 mg/m^2 cohort

Trend toward increased activity with higher AT13387 dose:

- AT13387 at 150 mg/m^2
 - (0 PR, 2/5 with tumor reduction)
- AT13387 at 180 mg/m^2
 - (1 PR, 4/7 with tumor reduction)
- AT13387 at 220 mg/m^2
 - (3 PR, 9/11 with tumor reduction)

Figure 6. Serial CT Scans from one subject

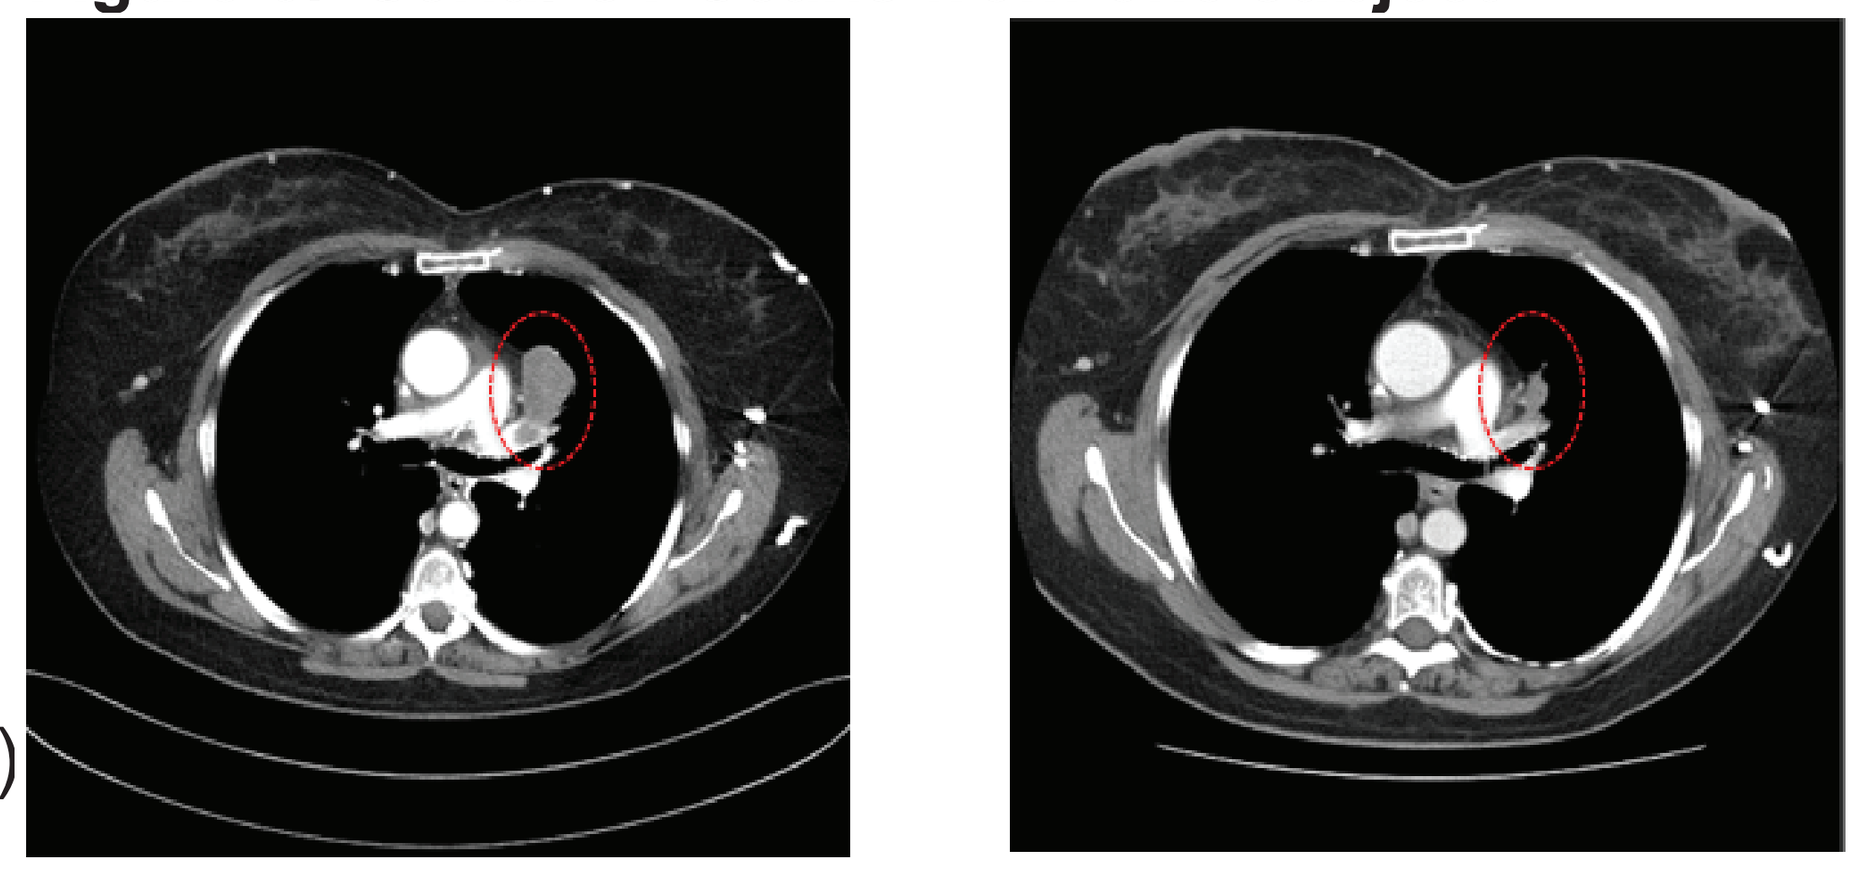

Screening (pre-cycle 1D1) After 2 cycles of treatment

RECIST - 55% decrease = PR

Table 4. Response by RECIST

Cohort (AT13387 Dose level)	150 mg/m^2 (n=8)	180 mg/m^2 (n=9)	220 mg/m^2 (n=15)	Total (n=32)
Assessable (n)	5	8	12	25
CR/PR (n, %)	0 (0%)	1 (12.5%)	3 (25%)	4 (16%)
SD (n, %)	5 (100%)	4 (50%)	5 (41.7%)	14 (56%)
PD (n, %)	0 (0%)	3 (37.5%)	4 (33.3%)	7 (28%)

SAFETY

The most common AEs related to AT13387 treatment, independent of grade, reported in >10% of subjects (cohorts combined) were as follows:
Diarrhoea (75%), Fatigue (63%), Nausea (59%), Decreased Appetite (38%), Vomiting (25%), Dizziness (19%), Weight decreased (19%), Dry mouth (16%), Dysgeusia (16%), Oedema peripheral (16%), ALT increased (13%), AST increased (13%)

Table 5. Adverse Events Grade 3 or Higher Related to AT13387 Treatment

Cohort (AT13387 dose level, CZT at 250 mg BID)	150 mg/m^2 (N=8)	180 mg/m^2 (N=9)	220 mg/m^2 (N=15)	Total (N=32)
Number of subjects who reported at least one TEAEs	1 (13%)	6 (67%)	5 (33%)	12 (38%)
Anaemia	0	1 (11%)	0	1 (3%)
Diarrhoea	0	3 (33%)	3 (20%)	6 (19%)
Fatigue	0	0	1 (7%)	1 (3%)
Pneumonia	1 (13%)	0	0	1 (3%)
Alanine aminotransferase increased	0	0	1 (7%)	1 (3%)
Neutrophil count decreased	0	0	1 (7%)	1 (3%)
Dehydration	0	1 (11%)	0	1 (3%)
Hyperkalaemia	0	0	1 (7%)	1 (3%)
Muscular weakness	0	1 (11%)	0	1 (3%)
Syncope	0	1 (11%)	0	1 (3%)
Pneumonitis	1 (13%)	0	0	1 (3%)
Pulmonary embolism	0	0	1 (7%)	1 (3%)

Note: one event at the 180 mg/m^2 dose level was uncoded and is omitted from this table

Dose Escalation & Dose Limiting Toxicities

- AT13387 dosing was initiated at 150 mg/m2 and escalated to 220 mg/m^2
- No dose limiting toxicities were observed at the doses investigated
- All CTCAE grade 3 or higher events related to AT13387 treatment are listed in Table 5

Summary/Conclusions

- AT13387 is well tolerated at a dose up to 220 mg/m^2 in combination with full dose CZT
- Administration of the agents in combination does not appear to change the pharmacokinetics of either agent
- Encouraging activity has been seen in the dose-escalation part of the study
 - 4 Objective PR by RECIST
 - More antitumor activity at higher dose
 - Over 50% patients with decrease in tumor size
- Arms B (prior to progression on CZT) and C (following progression on CZT) are open and enrolling

References

1. Smyth T, Munck J, Rodriguez-Lopez A, McMenamin R, Thompson N, Azab M, Lyons J, Wallis NG, Combining the HSP90 inhibitor, AT13387, with crizotinib improves response in an ALK-positive model of NSCLC; IASLC 15th World Conference on Lung Cancer Sydney Australia 2013: Abstract O08.06
2. Shapiro, GI, Kwak, E, Dezube BJ, Yule M, Ayrton J, Lyons J, and Mahadevan D. First-in-Human Phase 1 Dose Escalation Study of a Second-Generation Non-Ansamycin HSP90 Inhibitor, AT13387, in Patients with Metastatic Solid Tumors. Clinical Cancer Research, In Press
3. Alice T. Shaw, M.D., Ph.D., Dong-Wan Kim, M.D., Ph.D., Kazuhiko Nakagawa, M.D., Ph.D., et.al. Crizotinib versus Chemotherapy in Advanced ALK-Positive Lung Cancer. N Engl J Med 2013; 368:2385-2394 June 20, 2013

© Astex Pharmaceuticals, Inc.

Poster presented at: European Society for Medical Oncology (ESMO), Madrid, Spain September 26 - 30, 2014

Poster can be downloaded from www.astx.com