Abstract
2025

Immune checkpoint(s) expression in AML patients enrolled in a phase 1-2 study with guadecitabine

Carolina Fazio[1], Alessia Covre[1], Maria Lofiego[1], Pietro Taverna[2], Mohammad Azab[2], James N. Lowder[2], Sandra Coral[1,3] and Michele Maio[1]

[1]Medical Oncology and Immunotherapy, Department of Oncology, University Hospital of Siena, Istituto Toscano Tumori, Siena, Italy; [2]ASTEX Pharmaceuticals Inc. Pleasanton, CA, USA; [3]Epigen Therapeutics, Pordenone, Italy.

Abstract

Immune checkpoint(s) blockade is becoming the therapeutic mainstay in melanoma and lung cancer. Based on these important clinical achievements, immunotherapy with immunomodulating monoclonal antibodies (mAb) is being explored as a new therapeutic modality in most human malignancies, either alone or in combination with other immunomodulating agents. We have provided extensive evidence that the second generation DNA hypomethylating agent (DHA), guadecitabine (SGI-110), plays a promising role in potentiating the immunogenicity and the immune recognition of human malignancies through the up-regulation of the expression of different immune molecules on cancer cells. These findings led us to hypothesize that DHA could represent ideal partners for immune checkpoint(s) blocking agents to improve their therapeutic efficacy. Here, we studied the expression of programmed death 1 (PD-1), programmed death-ligand 1 (PD-L1), and 2 (PD-L2), and cytotoxic T lymphocyte-associated antigen 4 (CTLA-4) in peripheral blood mononuclear cells of acute myeloid leukemia (AML) patients (N=23), enrolled in a phase 1-2 study with guadecitabine. Patients included in this preliminary analysis were selected based on their responsiveness to therapy. Quantitative RT-PCR analyses showed a constitutive expression (gene/β-actin molecules >7.5E-05) of PD1, PD-L1, PD-L2 and CTLA-4 in 82.6%, 100%, 47.8% and 8.7% patients, respectively. Guadecitabine therapy up-regulated (≥2 fold) the expression of PD1, PD-L1, PD-L2 and CTLA-4 in 57.9%, 56.5%, 18.2% and 0% patients, respectively, at any of the time-points investigated during treatment. *De novo* expression of PD1, PD-L2 and CTLA-4 was observed in 100%, 41.6% and 38.1% of the immune checkpoint(s)-negative patients, respectively.

Though preliminary, these results further support the strong link between epigenetics/DNA methylation and immune response, in cancer patients, and strengthen the therapeutic potential of epigenetic immunomodulation, with “consolidated” and emerging immune checkpoint(s) blocking mAb.

Background

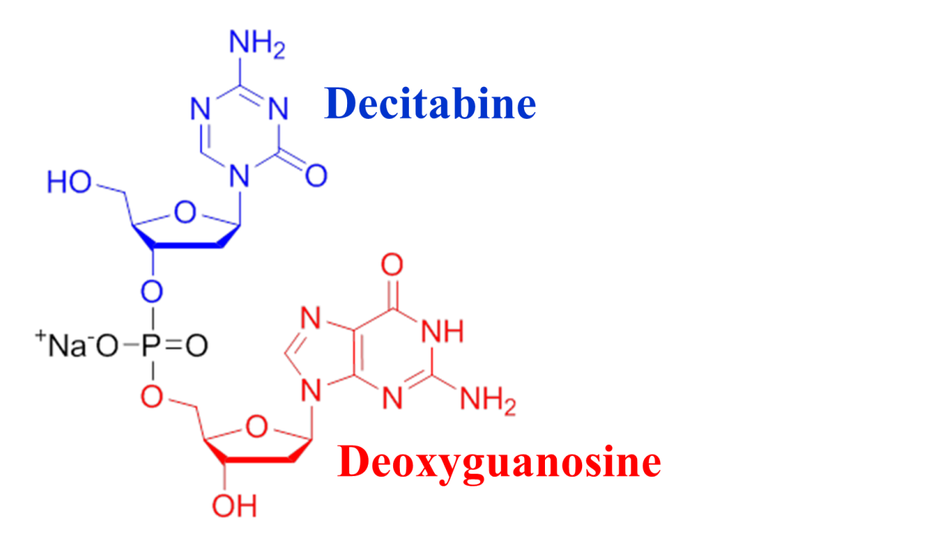
Decitabine
Deoxyguanosine

Figure 1: Guadecitabine: next generation HMA

Guadecitabine (SGI-110) is a novel hypomethylating dinucleotide of decitabine (DAC) and deoxyguanosine that is resistant to degradation by cytidine deaminase and results in prolonged *in vivo* exposure to its active metabolite DAC. The differentiated pharmacokinetic profile offers the potential of improved biological and clinical activity and safety over currently available hypomethylating agents (HMA).

Immunomodulatory activity of guadecitabine

Pre-clinical *In vitro* Data:

- Guadecitabine demethylates the promoters and induces the expression of CTAs MAGE-A3, NY-ESO-1 in cancer cell lines[1];
- Guadecitabine induces tumor recognition by cytotoxic T lymphocytes.

Pre-clinical *In vivo* Data:

- A synergistic antitumor activity was observed when guadecitabine is combined with anti-CTLA-4 mAb[2].

Clinical Data

We reported previously results from the Phase 1 dose-escalation study in AML and MDS[3] and the Phase 2 randomized dose-response study in RR AML patients of SGI-110 given SC at 2 doses (60 and 90 mg/m^2) in a 5-day regimen[4] or at 60 mg/m^2 in a 10-day regimen[5].

- Guadecitabine achieves dose-dependent demethylation and induction of CTAs (NY-ESO-1; MAGE-A1) in AML and MDS patients[2].

Results

Figure 1: Regulation of Immune checkpoints expression in PBMC from AML patients by guadecitabine

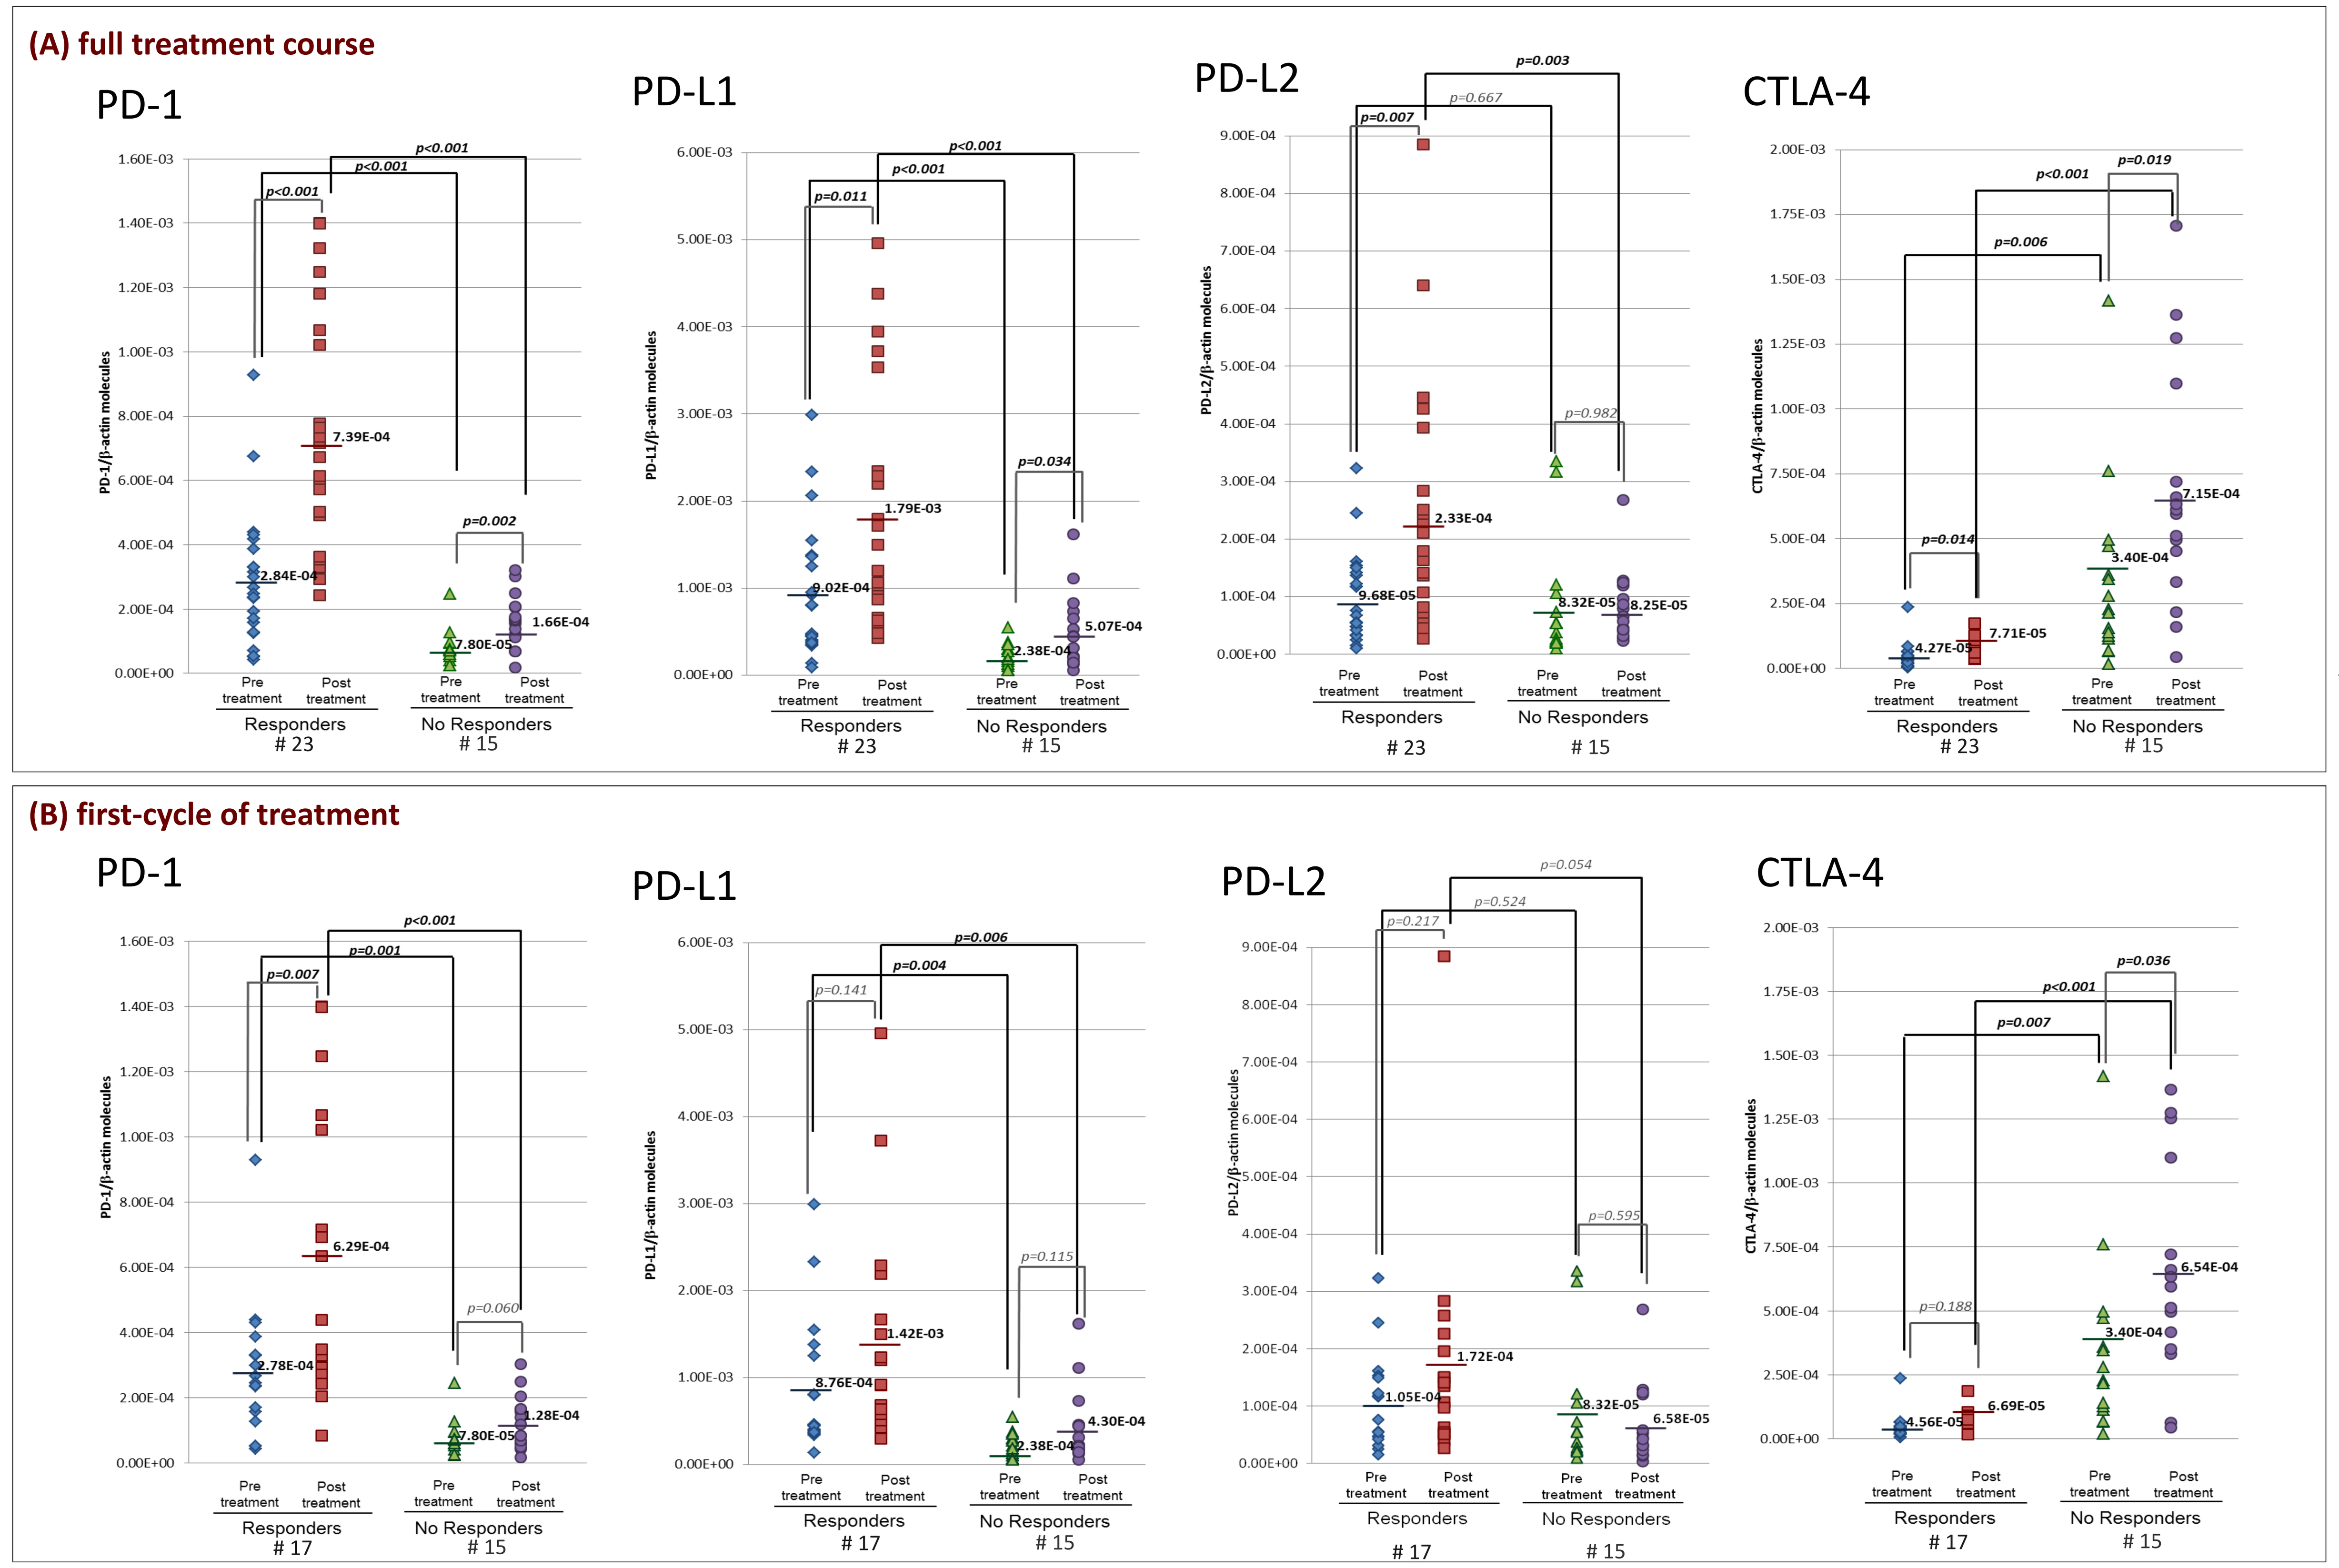
(A) full treatment course
PD-1
p<0.001
p<0.001
p<0.001
p=0.002
7.39E-04
2.84E-04
7.80E-05
1.66E-04
PD-L1
p<0.001
p<0.001
p=0.011
p=0.034
1.79E-03
9.02E-04
2.38E-04
5.07E-04
PD-L2
p=0.003
p=0.007
p=0.667
p=0.982
2.33E-04
9.68E-05
8.32E-05
8.25E-05
CTLA-4
p<0.001
p=0.019
p=0.006
p=0.014
4.27E-05
7.71E-05
3.40E-04
7.15E-04
Pre treatment
Post treatment
Responders # 23
No Responders # 15
(B) first-cycle of treatment
PD-1
p<0.001
p=0.001
p=0.007
p=0.060
6.29E-04
2.78E-04
7.80E-05
1.28E-04
PD-L1
p=0.006
p=0.004
p=0.141
p=0.115
1.42E-03
8.76E-04
2.38E-04
4.30E-04
PD-L2
p=0.054
p=0.524
p=0.217
p=0.595
1.72E-04
1.05E-04
8.32E-05
6.58E-05
CTLA-4
p=0.036
p<0.001
p=0.007
p=0.188
4.56E-05
6.69E-05
3.40E-04
6.54E-04
Responders # 17
No Responders # 15

Real-time RT-PCR analyses for PD-1, PD-L1, PD-L2 and CTLA-4 were performed on total RNA extracted from PBMC of responders and non responders AML patients, enrolled in a Phase 1-2 study with guadecitabine. The scatter plots show the comparison between constitutive and the highest expression values reached, at any time points (A) or within the first cycle (B), after treatment.

Table 1. Summary of Immune chekpoints expression in PBMC from responder and NON responders AML patients by guadecitabine

N° patients responders = 23	PD-1	PD-L1	PD-L2	CTLA-4
Constitutive positive expression (gene/β-actin molecules ≥1E-04)	82.6% (19/23)	95.6% (22/23)	43.4% (10/23)	4.30% (1/23)
De novo expression (gene/β-actin molecules ≥1E-04 with FC≥2)	100% (4/4)	100% (1/1)	46.1% (6/13)	13.6% (3/22)

N° patients NON responders=15	PD-1	PD-L1	PD-L2	CTLA-4
Constitutive positive expression (gene/β-actin molecules ≥1E-04)	13,3% (2/15)	86,6% (13/15)	26,6% (4/15)	80% (12/15)
De novo expression (gene/β-actin molecules ≥1E-04 with FC≥2)	61.5% (8/13)	100% (2/2)	0% (0/11)	100% (3/3)

Table 2. Clinical characteristics of selected AML patients enrolled in a phase 1-2 study with guadecitabine

Patient	Diagnosis	Dose mg/m^2	Age	Sex	Response
RR-10D	AML	60	55	F	CRp
RR-10D	AML	60	51	F	CR
RR-10D	AML	60	61	F	CRp
TN-5D	AML	90	80	F	CRi
TN-5D	AML	90	77	F	CRp
TN-5D	AML	60	75	F	CR
TN-5D	AML	60	82	F	PR
TN-5D	AML	60	77	F	CRi
RR-10D	AML	60	33	M	CR
RR-10D	AML	60	53	M	CR
RR-10D	AML	60	67	M	CR
RR-5D	AML	90	71	M	CR
RR-5D	AML	90	71	M	CRi
RR-5D	AML	60	77	M	CR
TN-5D	AML	90	75	M	CR
TN-5D	AML	90	81	M	CR
TN-5D	AML	90	77	M	CR
TN-5D	AML	90	76	M	CR
TN-5D	AML	90	80	M	CRi

Patient	Diagnosis	Dose mg/m^2	Age	Sex	Response
TN-5D	AML	90	76	M	CRi
TN-5D	AML	60	74	M	CR
TN-5D	AML	60	83	M	CR
TN-5D	AML	60	75	M	CRi
TN-5D	AML	90	80	M	NR
TN-5D	AML	90	69	F	NR
TN-5D	AML	60	65	M	NR
TN-5D	AML	90	77	F	NR
TN-5D	AML	60	81	F	NR
TN-5D	AML	90	69	M	NR
TN-5D	AML	90	83	M	NR
TN-5D	AML	60	92	M	NR
TN-5D	AML	60	72	M	NR
TN-5D	AML	90	70	M	NR
TN-5D	AML	60	85	F	NR
RR-5D	AML	60	46	F	NR
RR-5D	AML	90	78	M	NR
RR-5D	AML	60	58	M	NR
RR-5D	AML	90	78	F	NR

AML = acute myeloid leukemia; RR = relapsed/refractory; TN = treatment naive; 5D = 5-days treatment (days 1-5); 10D = 10-days treatment (days 1-10); CR = complete response; CRp = complete response with incomplete platelet recovery; CRi = complete response with incomplete blood county recovery; NR = No Response

Conclusions

- The mean baseline expression levels of PD-1 and PD-L1 was significantly (p<0.001) higher in AML patients responders *vs* non responders to guadecitabine treatement.
- The mean baseline expression levels of CTLA-4 was significantly (p=0.006) lower in AML patients responders *vs* non responders to guadecitabine treatement.
- Guadecitabine induced/up-regulated the expression of immune checkpoints molecules in PBMC from AML patients.
- A statistically significant (p<0.05) higher expression of PD-1, PD-L1 and PD-L2 was observed in responders *vs* non responders AML patients treated with guadecitabine.
- A statistically significant (p<0.001) lower expression of CTLA-4 was observed in responders *vs* non responders AML patients treated with guadecitabine.
- In detail, the statistically significant differences in the modulation of PD1, PD-L1 and CTLA-4 expression between responders and non responders was observed within the first cycle of therapy.
- Guadecitabine induced PD1, PD-L1, PD-L2 and CTLA-4 expression in 100% vs 61.5%, 100% vs 100%, 41.6% vs 0% and 13.6% vs 100% of responders *vs* NON responders AML patients, respectively.
- A phase I-II clinical study is ongoing to test guadecitabine epigenetic priming followed by CTLA-4 blockade in metastatic cutaneous melanoma patients. The study will enroll from 6 to 19 metastatic melanoma patients. Four pts have been enrolled to date[6].

www.clinicaltrials.gov identifier: NCT02608437

References

1.Coral et al, Cancer Immunol. Immunother., 2013; Srivastava et al, Leuk. Res., 2014; Srivastava et al, Epigenetics 2015
2.Covre et al, Semin. Oncol., 2015
3.Issa JP et al, Lancet Oncol. 2015
4.KantarjianH et al, ASH 2013
5.Griffiths E et al, ASCO 2014
6.Maio et al, Ann. Oncol. 2015

AACR annual meeting 16-20 April 2016, New Orleans

Study supported by Astex Pharmaceuticals