Genetic Determinants of Response to Guadecitabine (SGI-110) in AML

Patricia L. Kropf, MD[1]; Woonbok Chung, PhD[2]; Andrew D. Kelly, BA[2]; Hagop M. Kantarjian, MD[3]; Gail J. Roboz[4]; Karen W.L. Yee, MD[5]; Pietro Taverna, PhD[6]; Sue Naim[6]; Mohammad Azab, MD[6]; Jaroslav Jelinek, MD, PhD[2;] and Jean-Pierre J. Issa, MD[2]

1 Fox Chase Cancer Center, Philadelphia, PA
2 Fels Institute for Cancer Research, Lewis Katz School of Medicine at Temple University, Philadelphia, PA
3 Department of Leukemia, The University of Texas MD Anderson Cancer Center, Houston, TX
4 Weill Cornell/NY Presbyterian Medical Center, New York, NY
5 Princess Margaret Cancer Centre, University Health Network, Toronto, ON, Canada
6 Astex Pharmaceuticals, Inc., Pleasanton, CA

BACKGROUND

Guadecitabine is a next–generation hypomethylating drug with improved pharmacokinetics and pharmacodynamics compared to decitabine and demonstrated clinical activity in both treatment naïve (tn) and relapsed-refractory (rr) AML. Previous studies reported similar response rates to guadecitabine in different cytogenetic subsets but it remains unknown whether this extends to genetic changes.

METHODS

Pre-treatment blood or bone marrow derived DNA was available for analysis from a total of 220 patients with AML enrolled on guadecitabine phase I-II trials (122 rrAML and 98 tnAML). We included only patients treated at therapeutic doses. Mutations in a panel of 54 genes was studied by the TruSight Myeloid Sequencing Panel (Illumina) and deep sequencing on the Illumina HiSeq platform. FLT3 mutations were separately determined by PCR analysis. The sequence data was analyzed for mutations using the TruSeq Amplicon Application in the Illumina BaseSpace Suite. Putative mutation calls were further filtered by sequencing read quality, minimum variant allelic fraction, and presence in the dbSNP and COSMIC databases.

RESULTS

In aggregate, responses to guadecitabine in rrAML were 15 CR (12%), 12 PR/CRi/CRp and 95 non responders (NR) and in tnAML, responses were 34 CR (34%), 21 PR/CRi/CRp and 43 NR. Overall, a median of 1 (range 0 – 5) mutation was present in each patient, with no significant differences between tn and rr AML. The most frequently mutated genes were ASXL1 (16.8%), TET2 (14.1%), IDH2 (10.9%), NPM1 (10%), RUNX1 (9.5%), DNMT3A (9.1%), NRAS (9.1%), FLT3-ITD (8.6%), U2AF1 (8.2%), IDH1 (6.8%), TP53 (5%), and KRAS (4.5%). The distribution of mutations was as expected for a group of patients with rrAML and elderly tnAML

TABLES & FIGURES

RESPONSE RATE

	rrAML	tnAML
CR	12% (15/122)	35% (34/98)
PR/CRi/CRp	10% (12/122)	21% (21/98)
NR	78% (95/122)	44% (43/98)

FREQUENCIES OF MUTATIONS

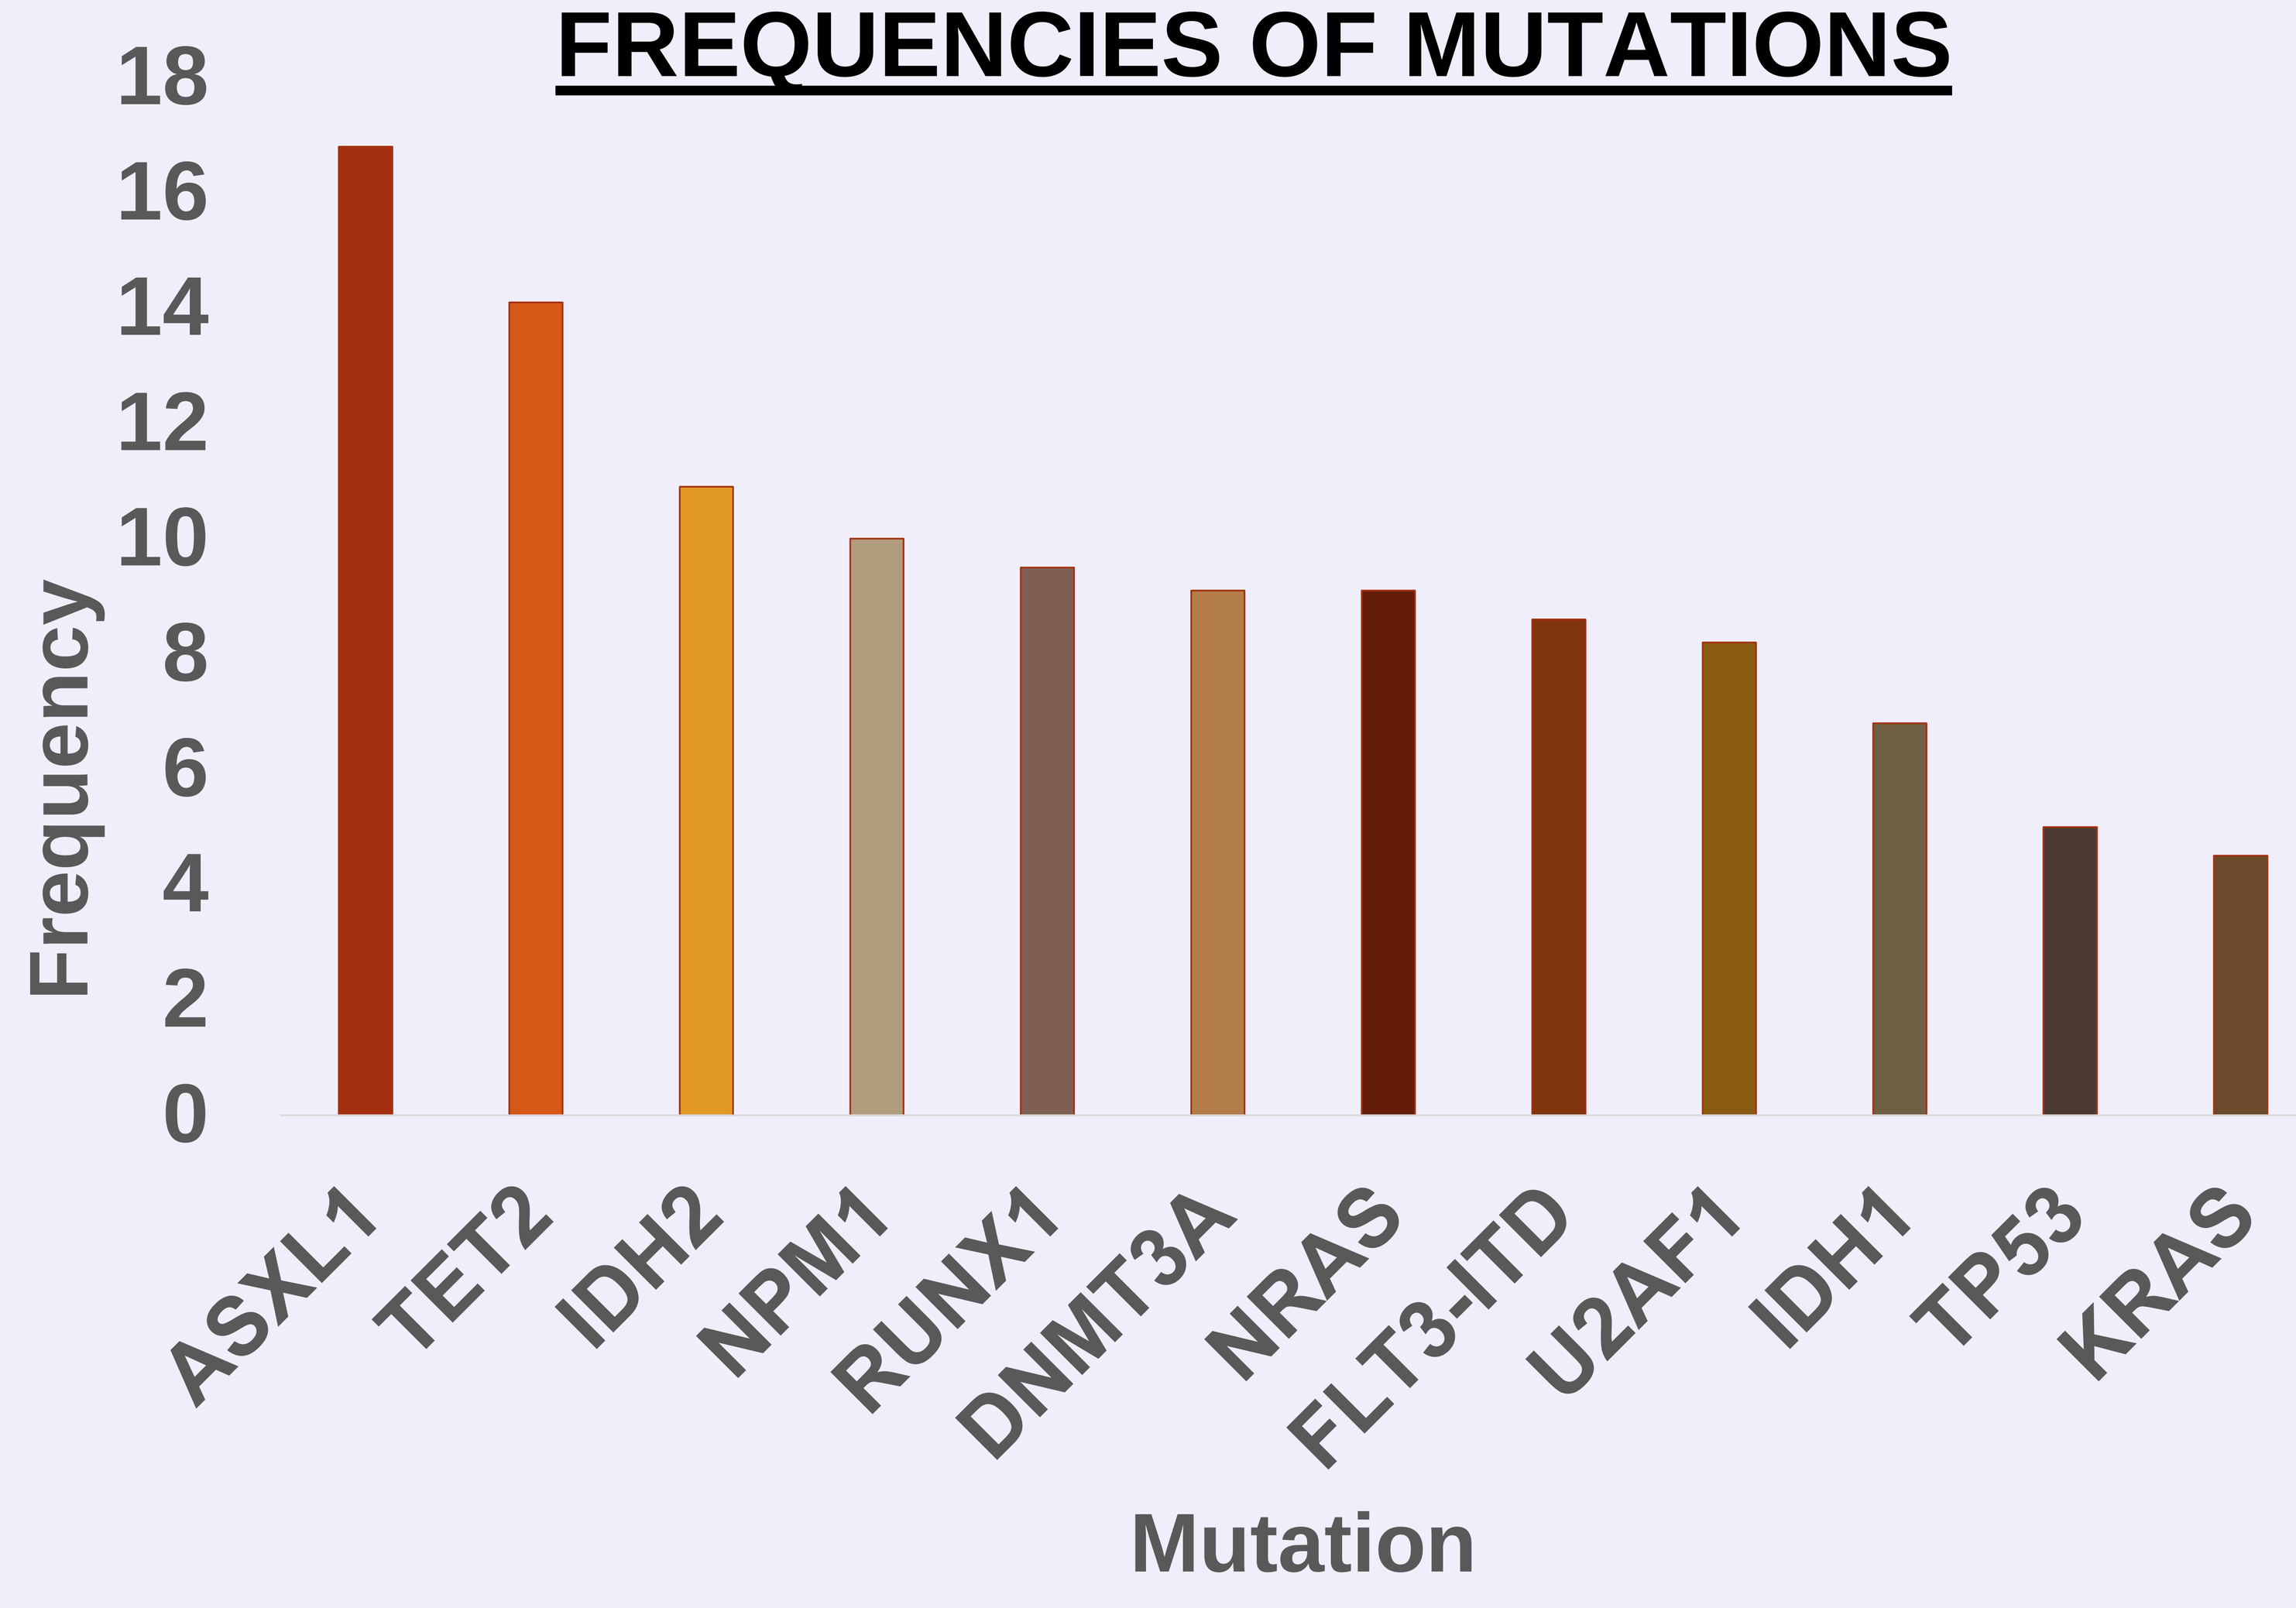
18
16
14
12
10
8
6
4
2
0
Frequency
ASXL1
TET2
IDH2
NPM1
RUNX1
DNMT3A
NRAS
FLT3-ITD
U2AF1
IDH1
TP53
KRAS
Mutation

RESPONSE RATE BASED ON MUTATION

Mutation Type	CR	Non-CR	P-value
NRAS	0/49	20/171	0.009
NRAS & KRAS	1/49	28/171	0.007
IDH2	1/49	23/171	0.02
Other Epigenetic Regulators*	27/49	88/171	0.26

* DNMT3A, ASXL1, EZH2, TET2, U2AF1 or WT1

RESULTS (Cont.)

We used Fisher's exact tests to compare mutation frequencies between patients who achieved CR and those who did not achieve CR. When we evaluated rrAML and tnAML separately, none of the genes showed a significantly different mutation rate between response subgroups. We then examined the population as a whole (N=220) and found that mutations in NRAS were significantly lower in patients who achieved CR (0/49) compared to those who did not (20/171, p=0.009). NRAS and KRAS mutations were inversely correlated, and when we considered the two genes together, mutations were present in 1/49 CR patients compared to 28/172 non-CR patients (p=0.007). Overall, CR rate was 3.4% in patients with RAS mutations compared to 25.1% in patients without such mutations. There was a similar significant trend for IDH2 mutations to be lower in CR patients (1/49) compared to non-CR patients (23/172, p=0.02) but this was not seen for IDH1. None of the mutations in other epigenetic regulators (DNMT3A, ASXL1, EZH2, TET2, U2AF1 or WT1) were significantly different between CR and non-CR patients individually or when we considered mutations in any of 8 epigenetic regulators (mutated in 27/49 CR patients vs. 88/172 non-CR patients, p=0.26). RAS mutations were higher in rrAML (22/122, 18.2%) than in tnAML (7/98, 7.1%, p=0.017) which may explain the lower CR rate in this group. Patients with PR/CRi/CRp were genetically similar to NR.

CONCLUSION

In patients with AML treated with guadecitabine, RAS pathway mutations and IDH2 mutations are associated with a lower likelihood of achieving a CR.

Disclosure: Agios, Amgen, Amphivena, Astex, AstraZeneca, Boehringer Ingelheim, Celator , Celgene, Cellectis, Genoptix, Janssen, Juno, , MEI Pharma, MedImmune, Novartis, Onconova, Pfizer, Roche/Genentech, Sunesis, Takeda, Teva Pharmaceutical Industries